Donors are Universal in the Fight Against *Clostridium difficile*: Results from Two Trials Investigating the Safety and Efficacy of RBX2660, a Microbiota-based Drug

Arnab Ray, MD[a], Courtney Jones, BS[b], Bill Shannon, PhD, MBA[c], Sharina Carter, BSr[c]

[a]Ochsner Clinic Foundation, New Orleans, LA; [b]Rebiotix Inc., Roseville, MN; [c]BioRankings, LLC, St. Louis, MO

Background

- Live microbial therapies are becoming an increasingly accepted treatment for recurrent *Clostridium difficile* infection (CDI).
- Questions remain regarding factors contributing to therapeutic success, including donor influence on patient outcome.
- RBX2660 is a microbiota-based drug derived from healthy human donors.
- The impact of donor on clinical outcome following RBX2660 treatment was evaluated in two clinical trials: PUNCH CD, an open label clinical trial, and the blinded phase of PUNCH CD 2, a randomized, double-blind, placebo-controlled trial.

Methods

Donor Sourcing and Traceability

- Donors underwent pre-enrollment blood and stool pathogen screening as well as extensive health history reviews prior to acceptance into the program.
- All drug product associated with that donor during a cycle is destroyed if a positive result is detected for any test.
- Four donors were used to prepare the RBX2660 used in the PUNCH CD study. Seventeen donors were used to prepare the RBX2660 used in the PUNCH CD 2 study.
- Donations were made on site at Rebiotix Inc., Roseville, MN.
- Donations were not pooled allowing for direct donor to patient traceability.
- Donors were randomized to patients for both the first and second doses.

RBX2660

- Each dose of RBX2660 is 150 mL of microbial suspension is packaged in a ready-to-use enema format.
- Manufactured using standardized, quality-controlled processes with guaranteed minimum quantity of microbes.
- In both studies, donor batches were randomized to the patient, and randomized again for the second dose. Patients could receive product from the same or different donors.

Clinical Studies

- Patients in the PUNCH CD (Phase 2) – an open label study where patients received 1 dose of RBX2660, with opportunity to receive a second dose.
- Patients in the PUNCH CD 2 (Phase 2B) trial were randomized to receive either: 2 doses of RBX2660; 2 doses of placebo; or 1 dose of RBX2660 followed by 1 dose of placebo. Therapies were administered via enema with doses 7 days apart.
- Success was defined as the absence of *C. difficile*-associated diarrhea at 8 weeks following completion of the last treatment.

Mathematical Model

- For each clinical study, a mixed model was used to predict treatment success taking into account donors and dose number.
- Repeated measures data was included to account for patients who received 2 doses of RBX2660. A patient was classified as either a success or failure for each dose.
- $P<0.05$ indicates statistical significance; all analyses were done using the R statistical package.

Results

Results from PUNCH CD[1]

- A total of 34 patients enrolled in the PUNCH CD Phase 2 trial (mean age 68.8 years, 67.6% female) were treated with at least one dose of RBX2660. Fifteen patients received a second dose.
- Success was not impacted by the donor or the dose order.

	ESTIMATE	P VALUE
FIXED EFFECT	Coefficient	
Intercept	0.4521	0.333
Dose (Dose 2)	1.38477	0.151
RANDOM EFFECT	Variance	
Donor	0.00	0.3921
Donor: Dose	0.53	0.397

Results from PUNCH CD 2[2]

- A total of 83 patients enrolled in the PUNCH CD 2 Phase 2B trial (mean age 62 years; 59% female) received at least one dose of RBX2660 in the blinded arm of the study. Forty-one patients received 2 doses of RBX2660.
- Success was not impacted by the donor or the dose order.

	ESTIMATE	P VALUE
FIXED EFFECT	Coefficient	
Intercept	10.9265	0.00
Dose (Dose 2)	0.4084	0.831
RANDOM EFFECT	Variance	
Donor	0.00	0.9999
Donor: Dose	0.00	0.9996

- The donor was not significant ($P>0.99$).
- The variance estimate refers to variation in outcome between donors. Variance = 0 indicates no difference in outcome based on donor.

References

1. Ray A, Jones C. Does the donor matter? Donor vs patient effects in the outcome of ta next-generation microbiota-based drug trial for recurrent *Clostridium difficile* infection. *Future Microbiol.* 2016; 11:611-6.

2. Ray A, Jones C, Shannon B, Carter S. Does the donor matter? Results from PUNCH CD 2, a Randomized Controlled Trial of a Microbiota-based Drug for Recurrent *Clostridium difficile* Infection, ACG 2016 Annual Scientific Meeting October 15-19, 2016, Las Vegas, NV

Figure 1: Proportion of Successes and Failures with RBX2660 by Donor in PUNCH CD and PUNCH CD 2 Clinical Trials

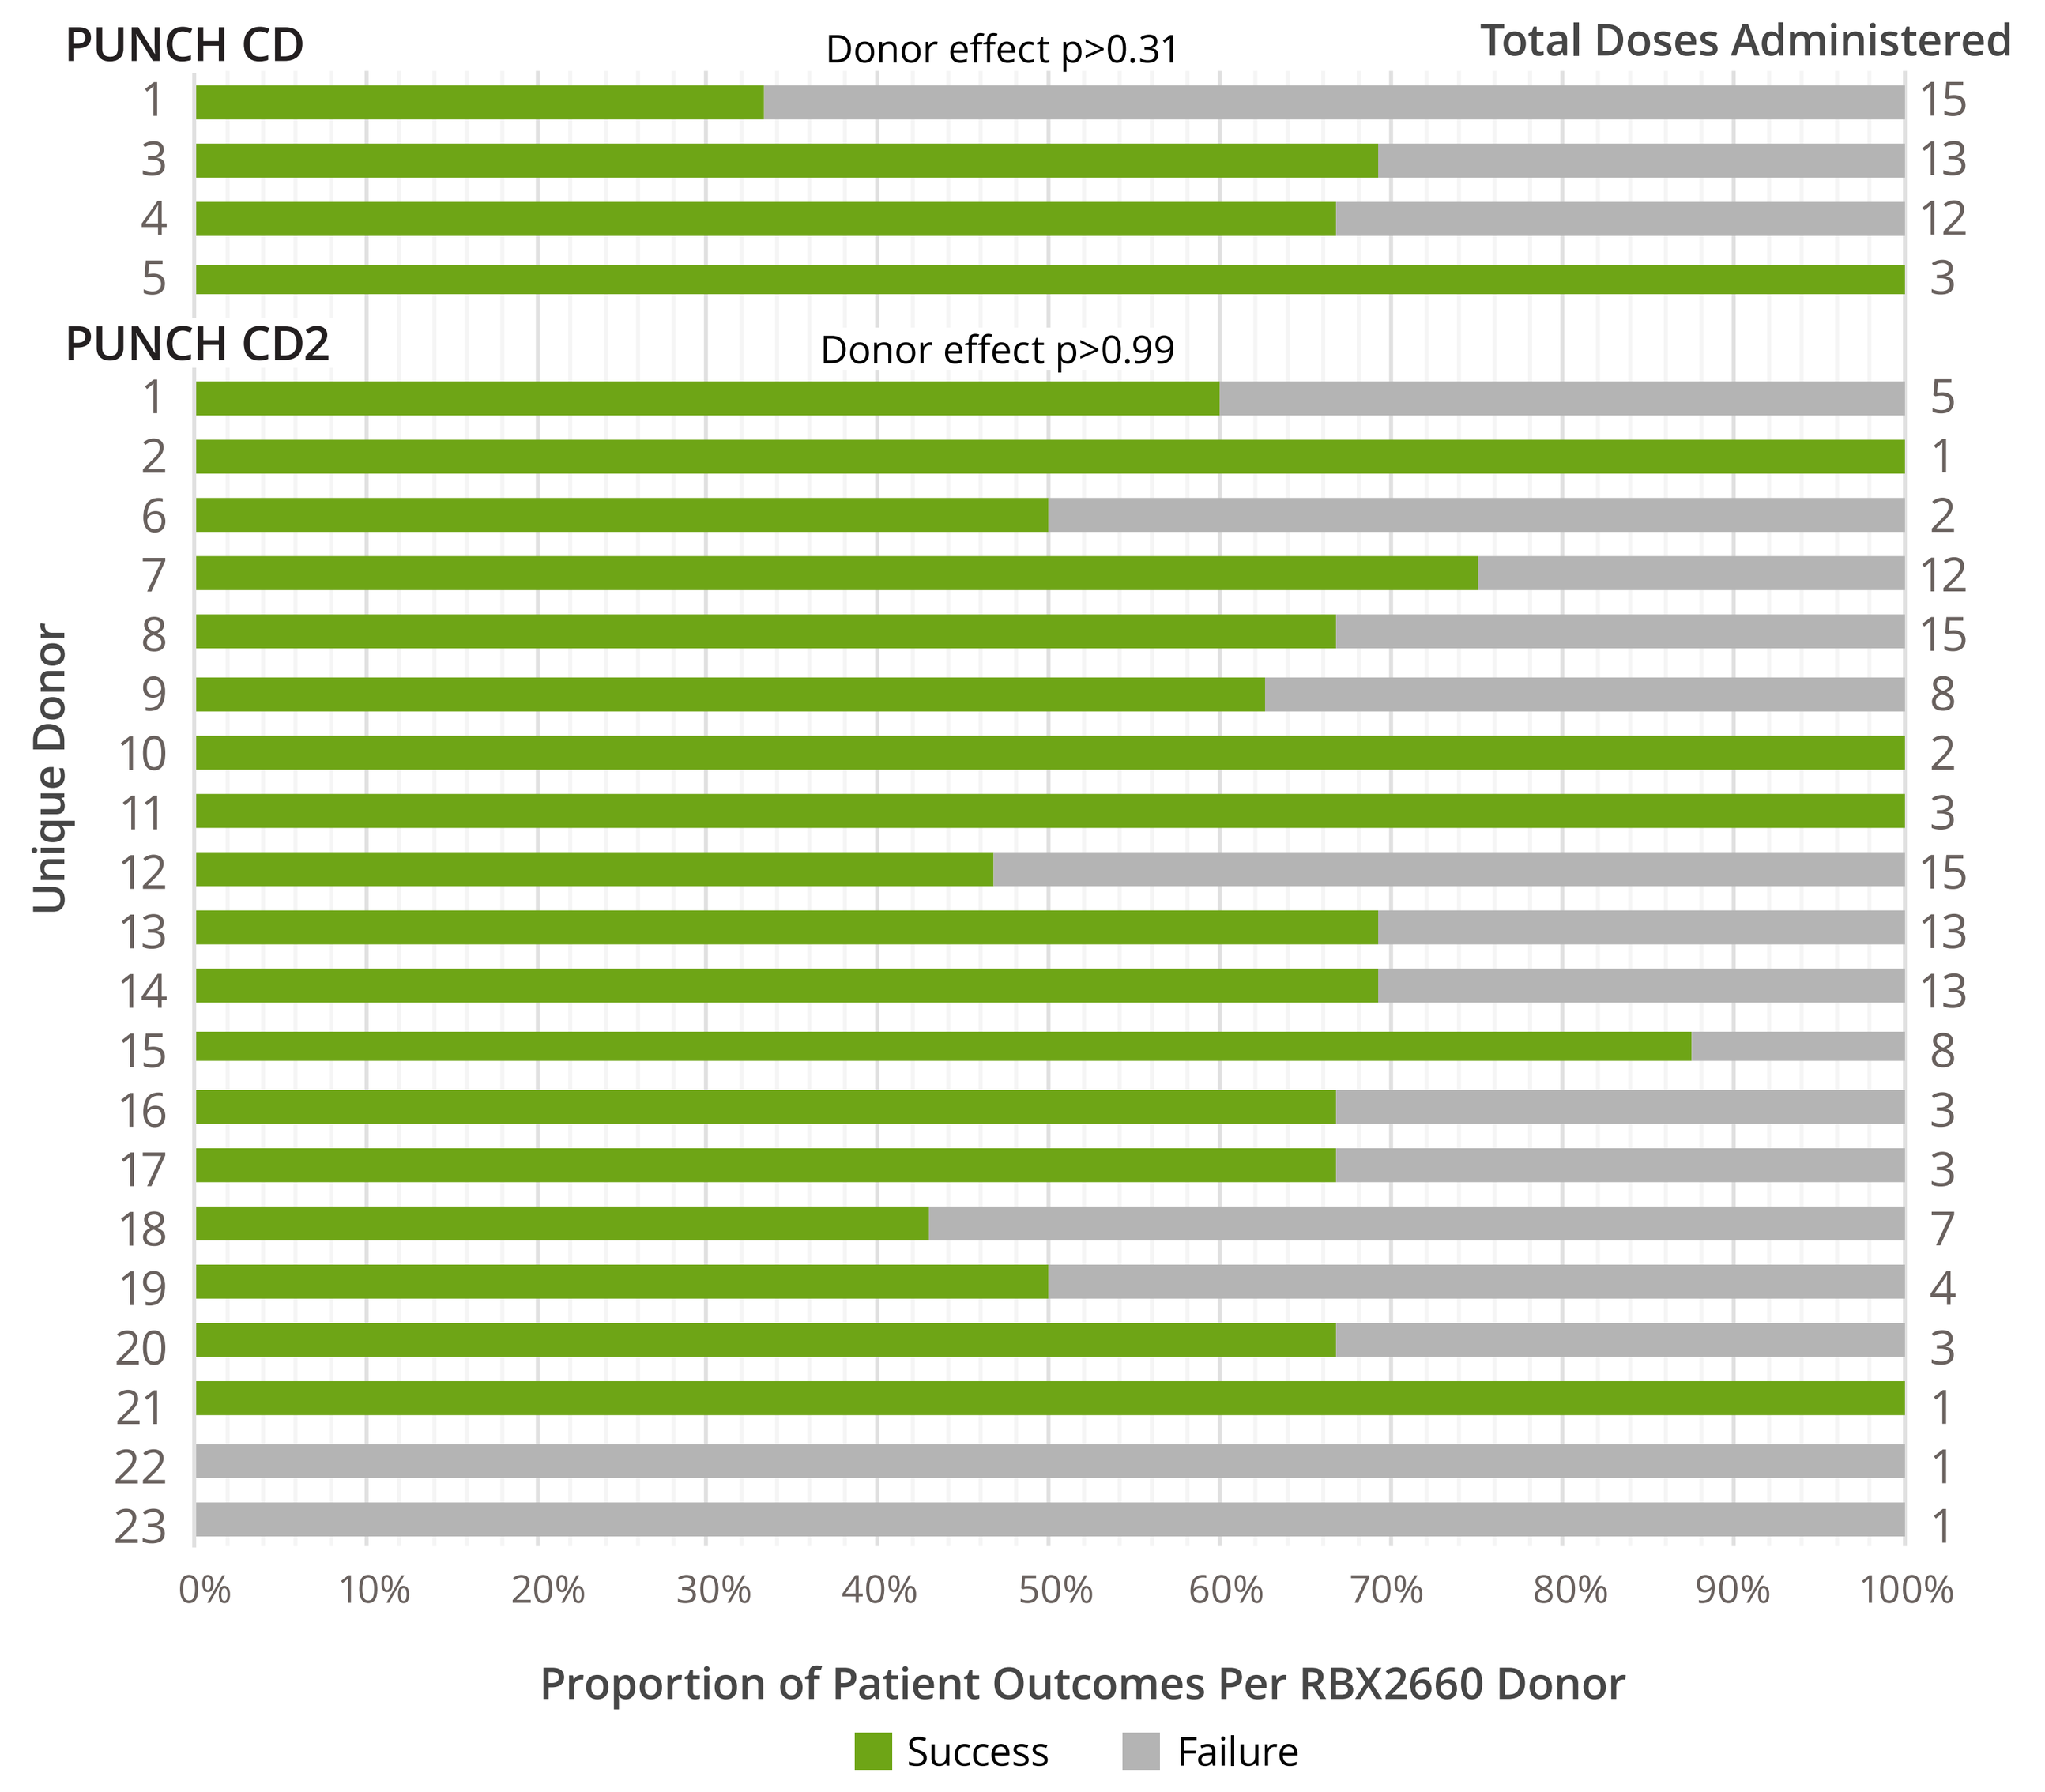
PUNCH CD
Donor effect p>0.31
Total Doses Administered
PUNCH CD2
Donor effect p>0.99
Unique Donor
0%
10%
20%
30%
40%
50%
60%
70%
80%
90%
100%
Proportion of Patient Outcomes Per RBX2660 Donor
Success
Failure

- One outcome (patient) may be associated with up to two different donors. Therefore, patients that received two doses of RBX2660 are represented twice.
- RBX2660 generated from Donor 1 was administered in both clinical trials.
- Successes and failures per Donor 1 were consistent across donors.

Conclusions

- RBX2660 success in preventing recurrent CDI is not determined by specific donor.
- Data from two separate clinical studies demonstrate that preparation of RBX2660 from a universal group of screened donors does not impact patient outcomes.
- A super donor was not identified.
- The role of donor in RBX2660 success for treatment of other clinical indications requires further study.