RBX2660 (microbiota suspension) for Recurrent *C. difficile* Infection: 60-Day Interim Analysis of the PUNCH CD Phase 2 Safety Study

Erik Dubberke, MD, MSPH[a]; Robert Orenstein, DO[b]; Paul Mariani, MD[c]; Kathleen Mullane, DO, FIDSA[d]; Mary Kay Sobcinski, RN, MHA[e]

[a]Washington University School of Medicine, St. Louis, MO; [b]Mayo Clinic Arizona, Phoenix, AZ; [c]Sanford Health, Fargo, ND; [d]University of Chicago Medicine, Chicago, IL; [e]Rebiotix Inc, Roseville, MN

FOR CORRESPONDENCE:
Erik Dubberke, MD, MSPH
Infectious Disease
Washington University School of Medicine
660 S. Euclid Ave., Box 8051
St. Louis, MO 63110
Tel: (314) 454-8293
E-mail:edubberk@dom.wustl.edu

Background

There is increasing recognition that fecal transplant (FT) is an effective treatment for recurrent *C. difficile* infection (CDI). However, donor screening and product preparation can be burdensome. Despite promising efficacy results, safety data are limited. We report on a 60-day interim analysis of the first prospective open-label multi-center safety study of a next generation microbiota restoration therapy that has been standardized and manufactured under controlled conditions.

Methods

Patients with recurrent CDI, defined as at least 3 CDI episodes or at least 2 severe episodes resulting in hospitalization, were enrolled. All patients received treatment with RBX2660 (microbiota suspension) administered via enema. A second treatment was permitted if CDI recurred in <8 weeks after the first treatment. Follow-up was at 7, 30 and 60 days and 3 and 6 months after the last treatment. The primary objective was the product-related adverse events (AEs). A secondary objective was CDI resolution.

Results

Of the 40 patients enrolled at 11 centers in the US, 34 patients (mean age 66.8 years, 67.6% female) received at least one treatment. Thirty-one patients were included in a 60-day interim analysis. A total of 158 AEs were elicited in 29 patients. AEs were predominantly mild to moderate and included flatulence, belching, constipation, and occasional bouts of diarrhea. There were 9 serious AEs reported in 6 patients (3 recurrent CDI $\leq$ 8 weeks post-treatment, all of which required hospitalization; 1 case of pneumonia; 1 pelvic fracture; 1 stab wound; 1 chronic obstructive pulmonary disease; 1 pulmonary edema and 1 respiratory failure). None of the serious AEs was related to RBX2660 or its administration. Efficacy of RBX2660 defined, as the absence of CDI at 8 weeks, after the last dose was 87.1%.

Figure 1: Patient Flow Through 60-day Follow-up

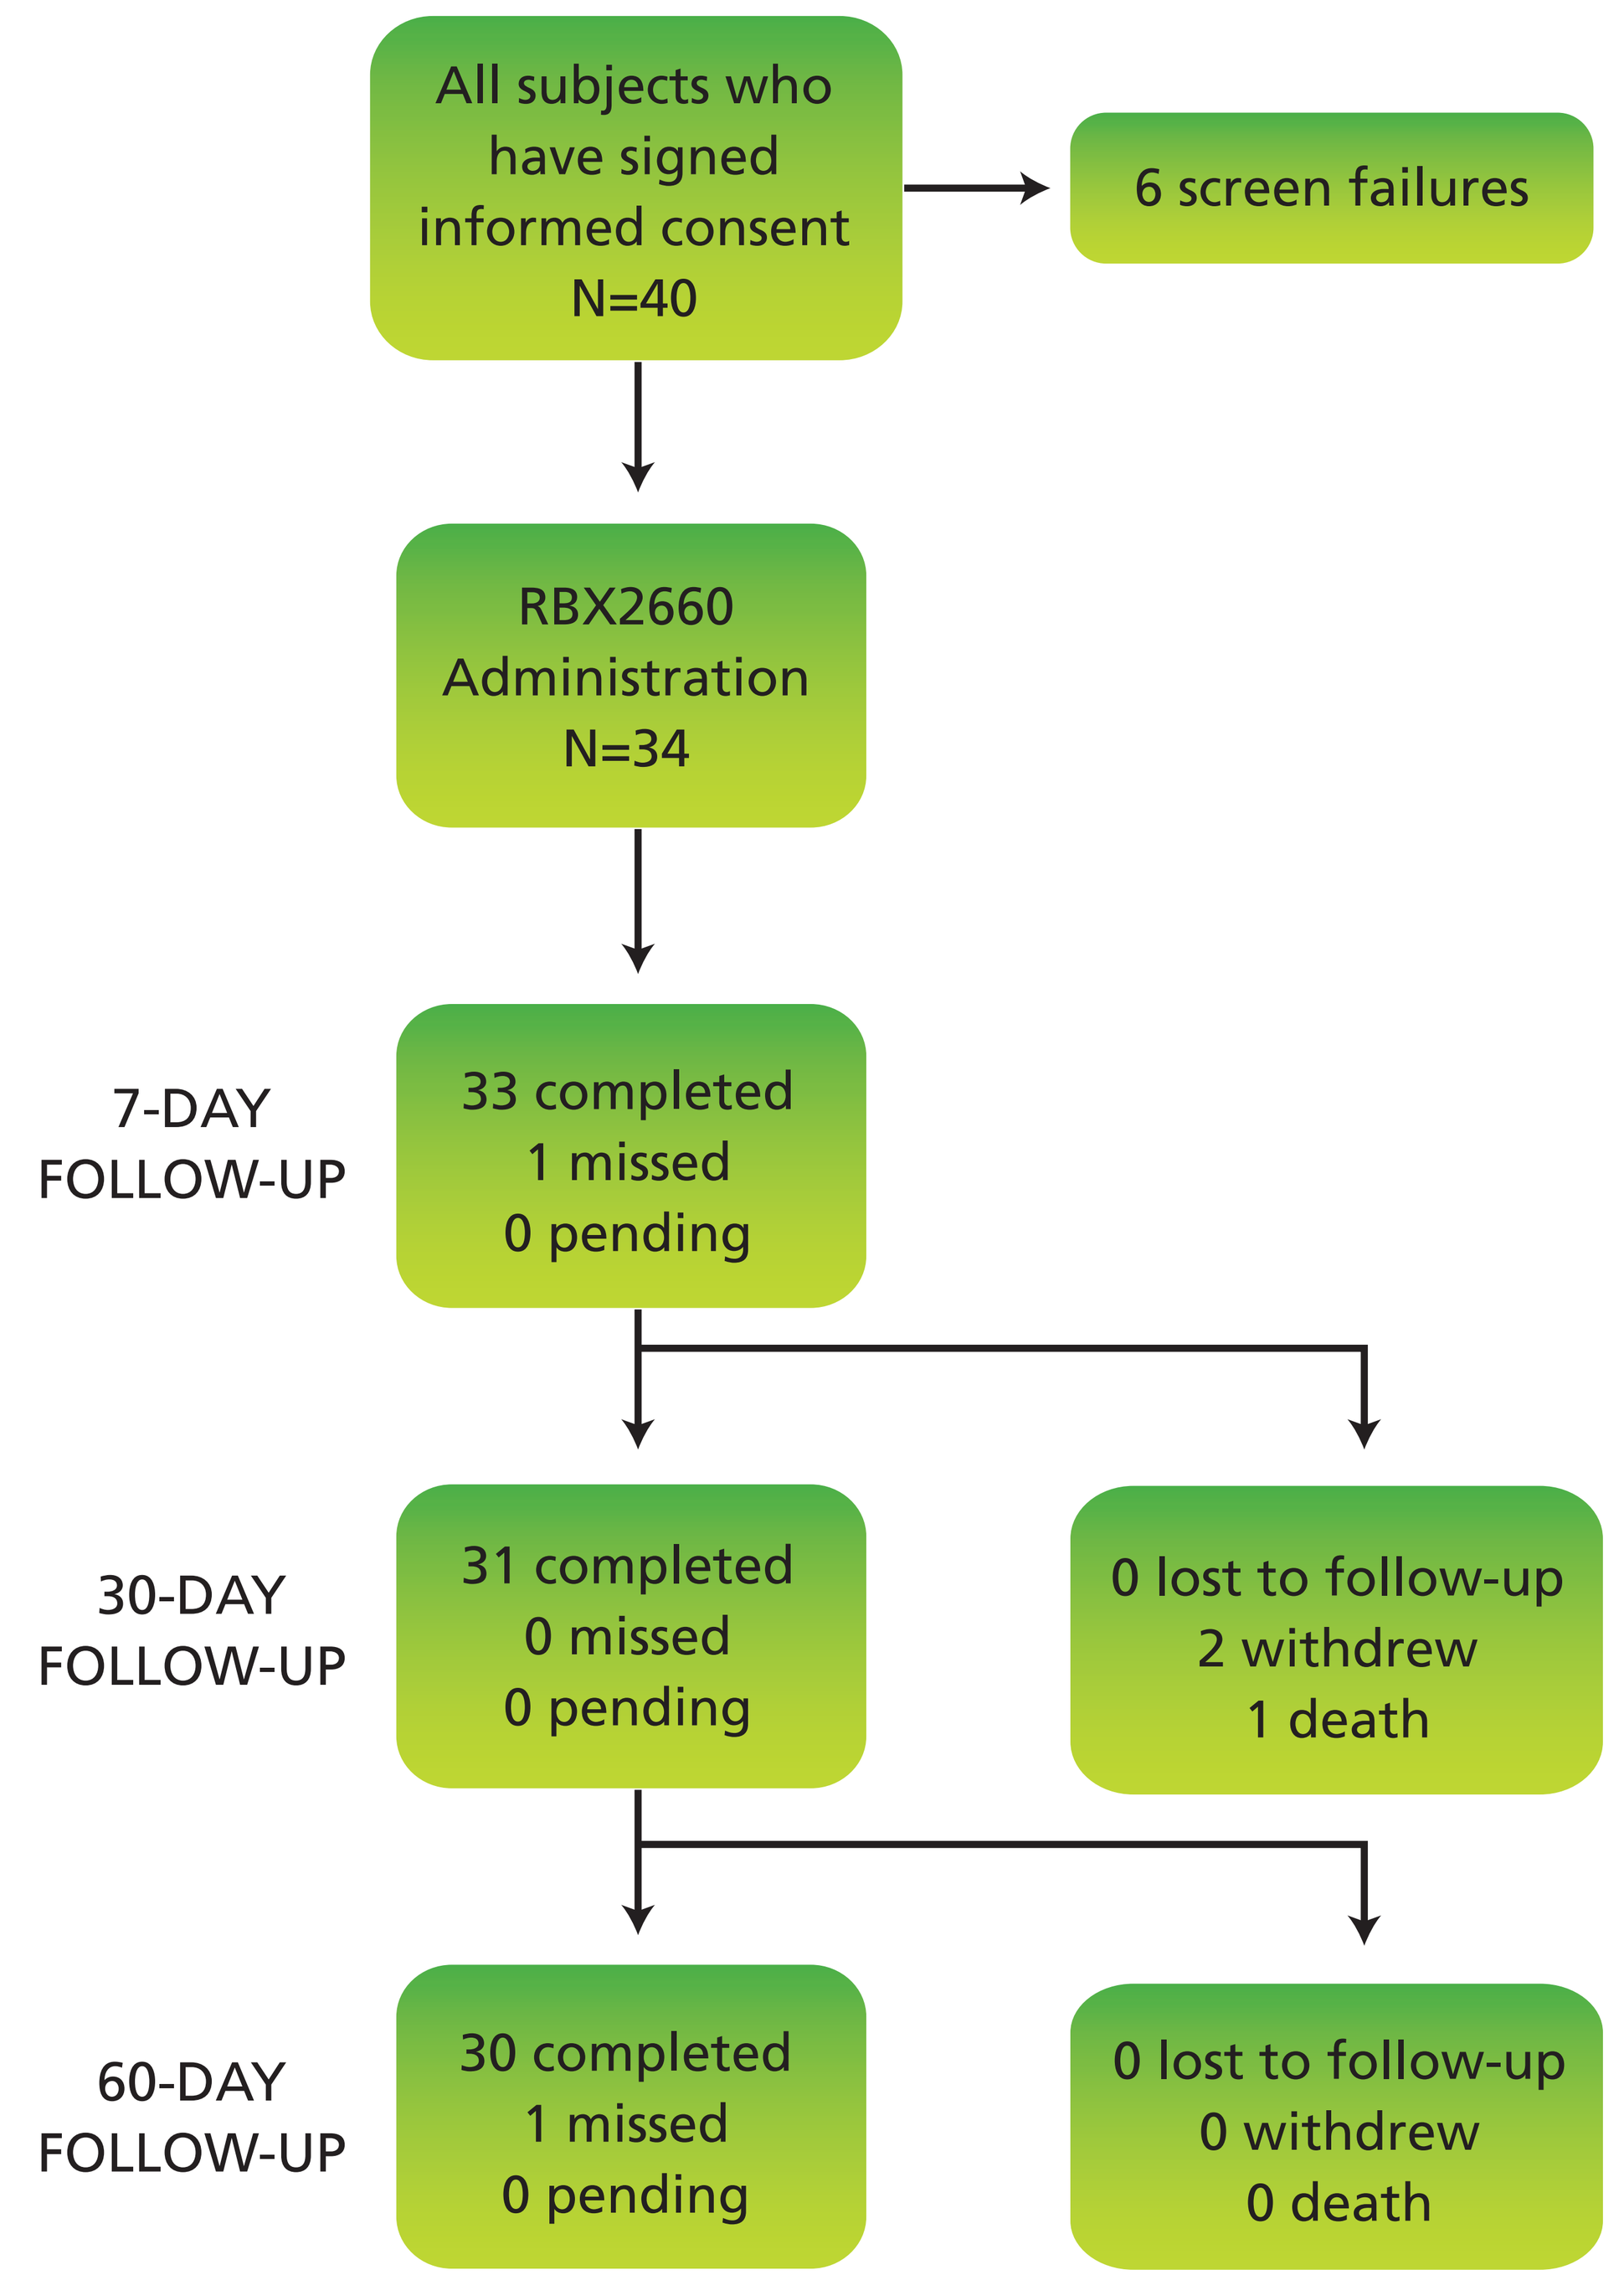
All subjects who have signed informed consent N=40
6 screen failures
RBX2660 Administration N=34
7-DAY FOLLOW-UP
33 completed 1 missed 0 pending
30-DAY FOLLOW-UP
31 completed 0 missed 0 pending
0 lost to follow-up 2 withdrew 1 death
60-DAY FOLLOW-UP
30 completed 1 missed 0 pending
0 lost to follow-up 0 withdrew 0 death

Table 1. Baseline Characteristics

Age	
Mean (SD)	66.8 (15.1)
Range	26.7 - 89.6
Gender	
Female	23/34 (67.6%)
Race	
American Indian or Alaska Native	1/34 (2.9%)
Asian	0/34 (0.0%)
Black or African American	1/34 (2.9%)
Native Hawaiian or Other Pacific Islander	0/34 (0.0%)
White	32 (94.2%)
BMI (kg/m²)	
Mean (SD), Range	24.4 (5.2), 15.0 - 37.0
Prior Medical History	
Head, eyes, ears, nose and throat	15/34 (44.1%)
Cardiovascular	19/34 (55.9%)
Gastrointestinal	21/34 (61.8%)
Musculoskeletal	13/33 (39.4%)
Neurological	5/34 (14.7%)
Genitourinary	18/26 (69.2%)
Psychiatric	9/32 (28.1%)

Table 2. Most Common Adverse Events through 60-day Follow-up

Type	No. Events	Patients n/N%
Diarrhea	24	10/34 (29.4)
Flatulence	15	13/34 (38.2)
Abdominal pain	14	10/34 (29.4)
Constipation	12	12/34 (35.3)
Abdominal distension	9	7/34 (20.6)

Figure 2: Overall Efficacy

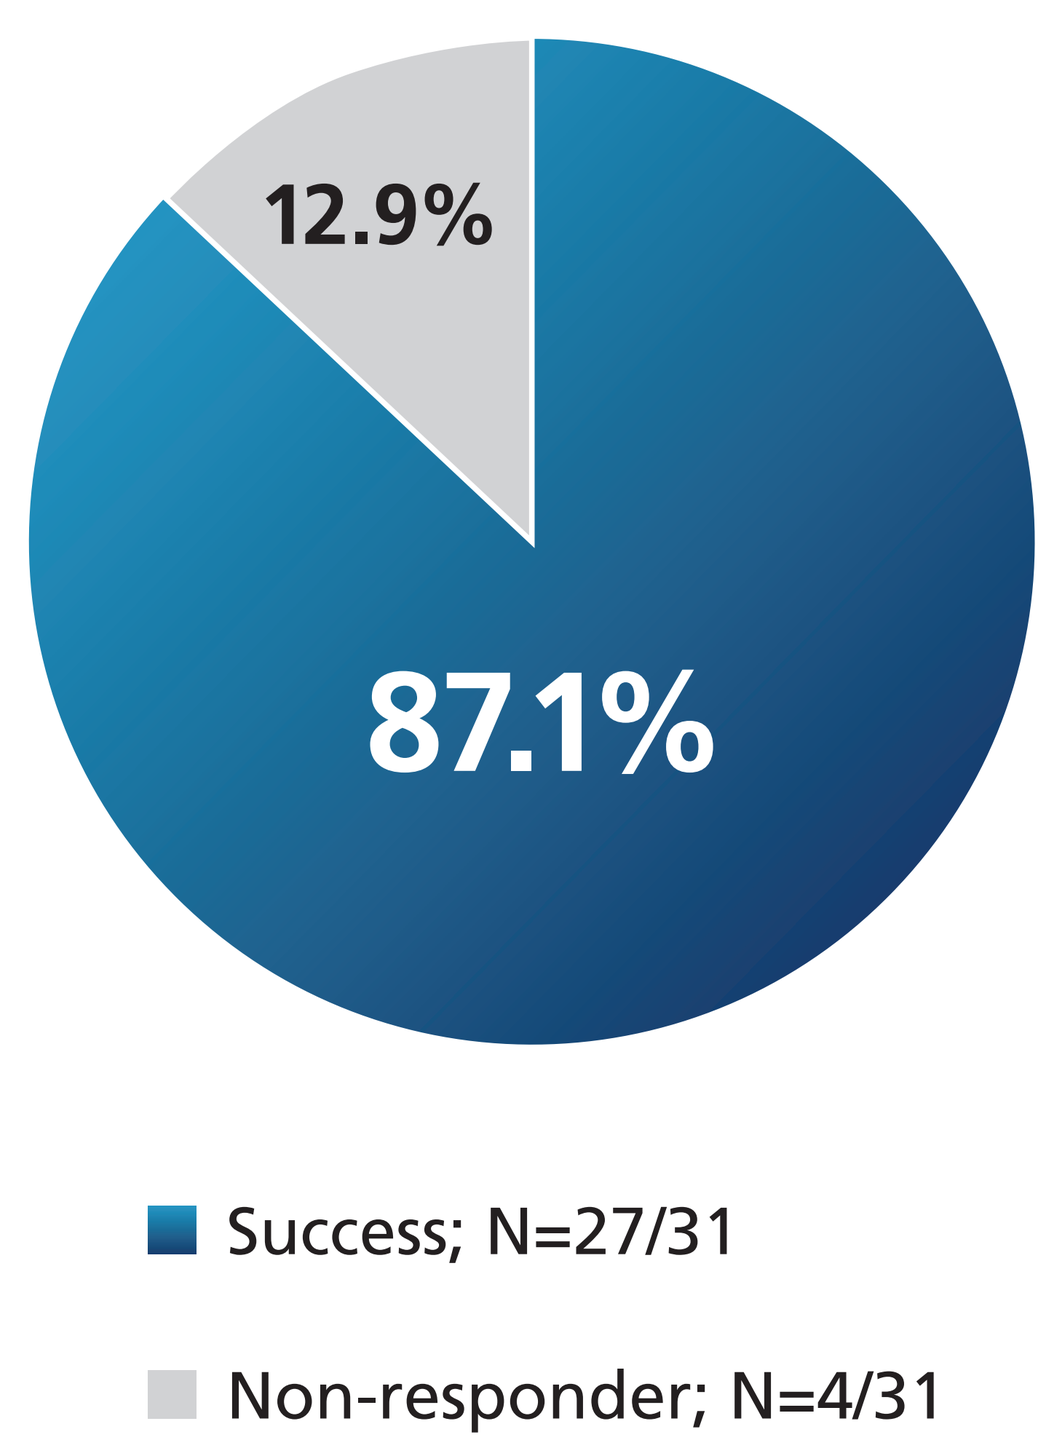
12.9%
87.1%
Success; N=27/31
Non-responder; N=4/31

Conclusion

RBX2660 was well-tolerated and demonstrated satisfactory safety in a 60-day interim analysis of the first prospective multi-center study of a next generation standardized, commercially prepared microbiota restoration therapy for recurrent CDI.