Assessing Infectious Threats in "Healthy" FMT Donors: Results of an Assessment Program to Support a Next-Generation Drug for Recurrent *Clostridium difficile* Infection

Courtney Jones, BS; Mary Kay Sobcinski, RN, MHA; Matt Adams MS; Rebiotix Inc, Roseville, MN

Corresponding Author
Courtney Jones
Rebiotix Inc.
Roseville, MN 551113
Tel: 651-705-8770
E-mail: cjones@rebiotix.com

Background

- Microbiota Restoration Therapy is the Rebiotix platform for delivering human-derived microbes into a sick patient's intestinal tract to treat disease.
- RBX2660 (microbiota suspension) is currently under investigation for recurrent *Clostridium difficile* infection (CDI).
- RBX2660 is sourced from human-derived microbes and manufactured using standardized, quality controlled processes.
- A donor program was implemented to provide a reliable source of raw material to supply Phase 2 studies.

Donor Recruitment and Screening

- Potential donors were recruited by word-of-mouth.
- Nearby universities were targeted as a source of young, healthy donors.
- A one-time recruitment bonus and nominal payments for accepted donations were offered.
- Potential donors underwent an initial screening process (Figure 1) for an extensive list of blood and stool pathogens prior to enrollment.
- Failures were permanently excluded.

Ongoing Donations

- If accepted into the program, donors agreed to make a minimum of 3 donations per week. A maximum of 10 donations per week was allowed (2 times daily, Monday through Friday). Each donation was tested for blood and stool pathogens.
- Daily health and diet questionnaires were completed with each donation to ensure there were no major changes in travel, medication, diet and health status (Figure 2).
- All donations were made on site and stored under controlled conditions.
- Donations were not pooled; aliquots from every donated stool were tested for pathogens using an extensive screen.
- At the end of each approximately 45-day donation cycle (Figure 3), donors underwent blood testing and donated aliquots underwent testing for pathogens.
- A positive result on any of the screens triggered destruction of all drug product associated with the donor during a cycle and exclusion of the donor from the program.

Figure 1. Initial Screening Process for Potential Donors

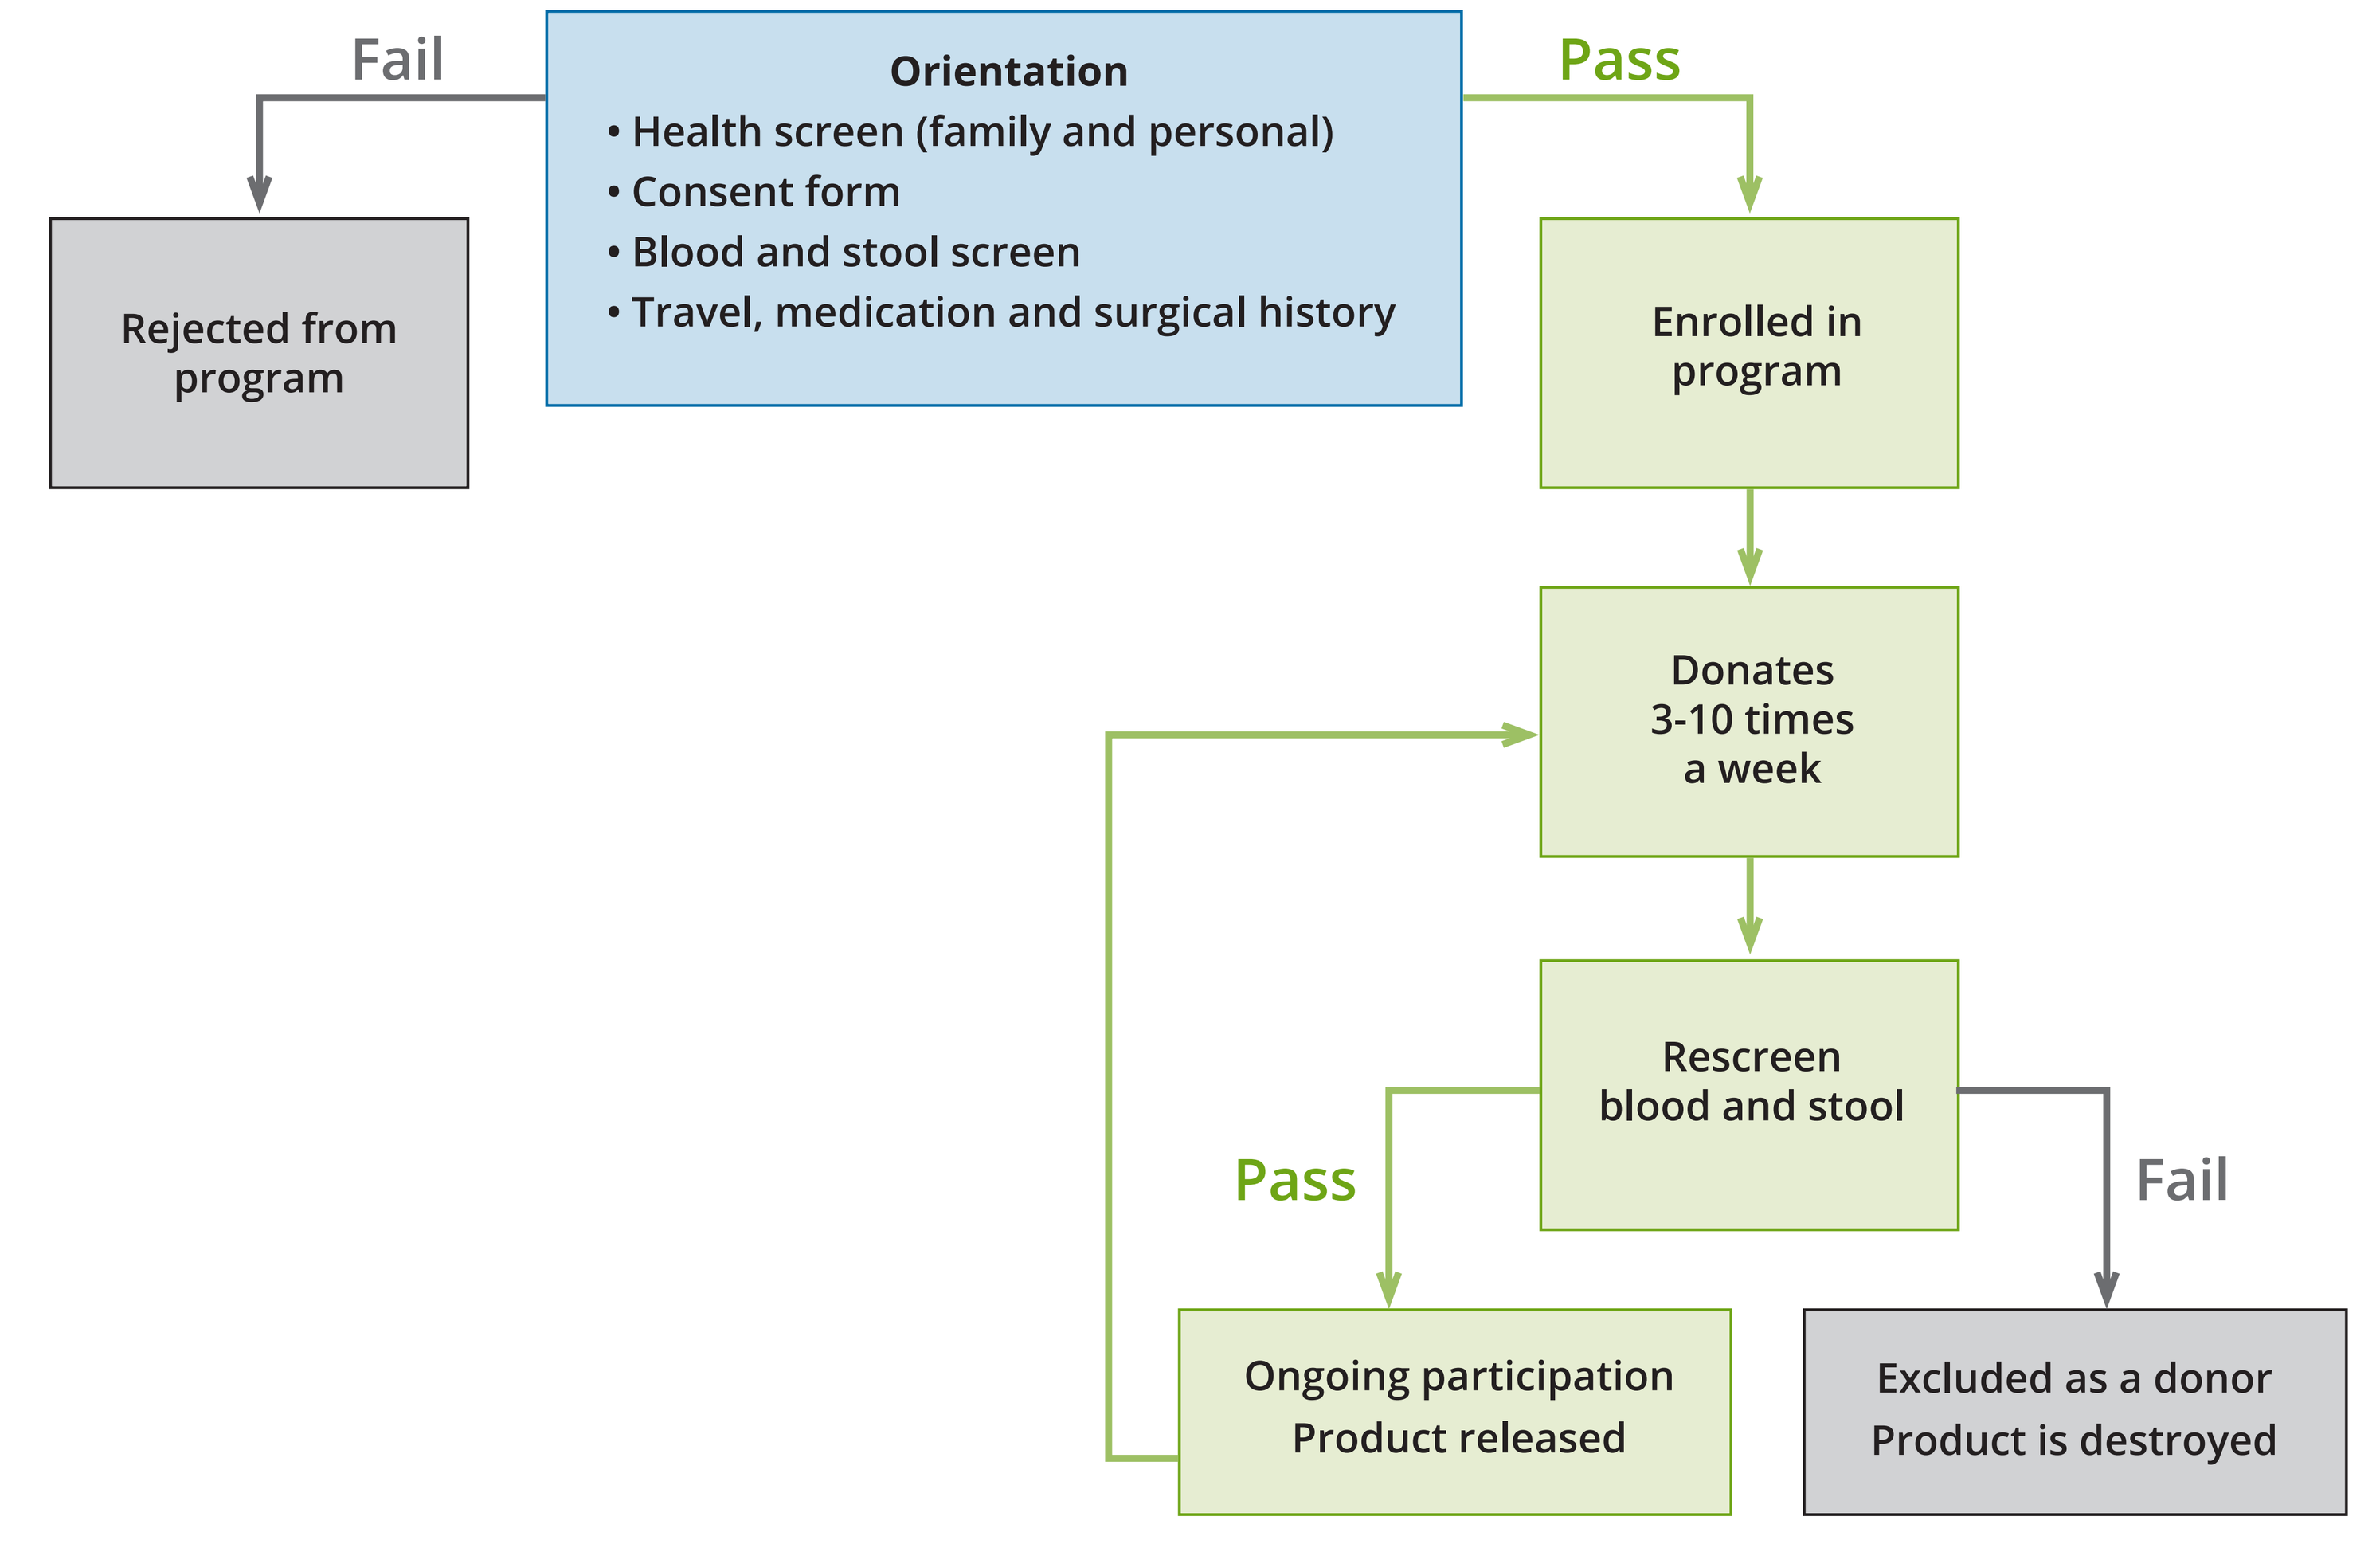
Fail
Pass
Orientation
• Health screen (family and personal)
• Consent form
• Blood and stool screen
• Travel, medication and surgical history
Rejected from program
Enrolled in program
Donates 3-10 times a week
Rescreen blood and stool
Pass
Fail
Ongoing participation
Product released
Excluded as a donor
Product is destroyed

Figure 2. Screening Process for Enrolled Donors: Per Donation

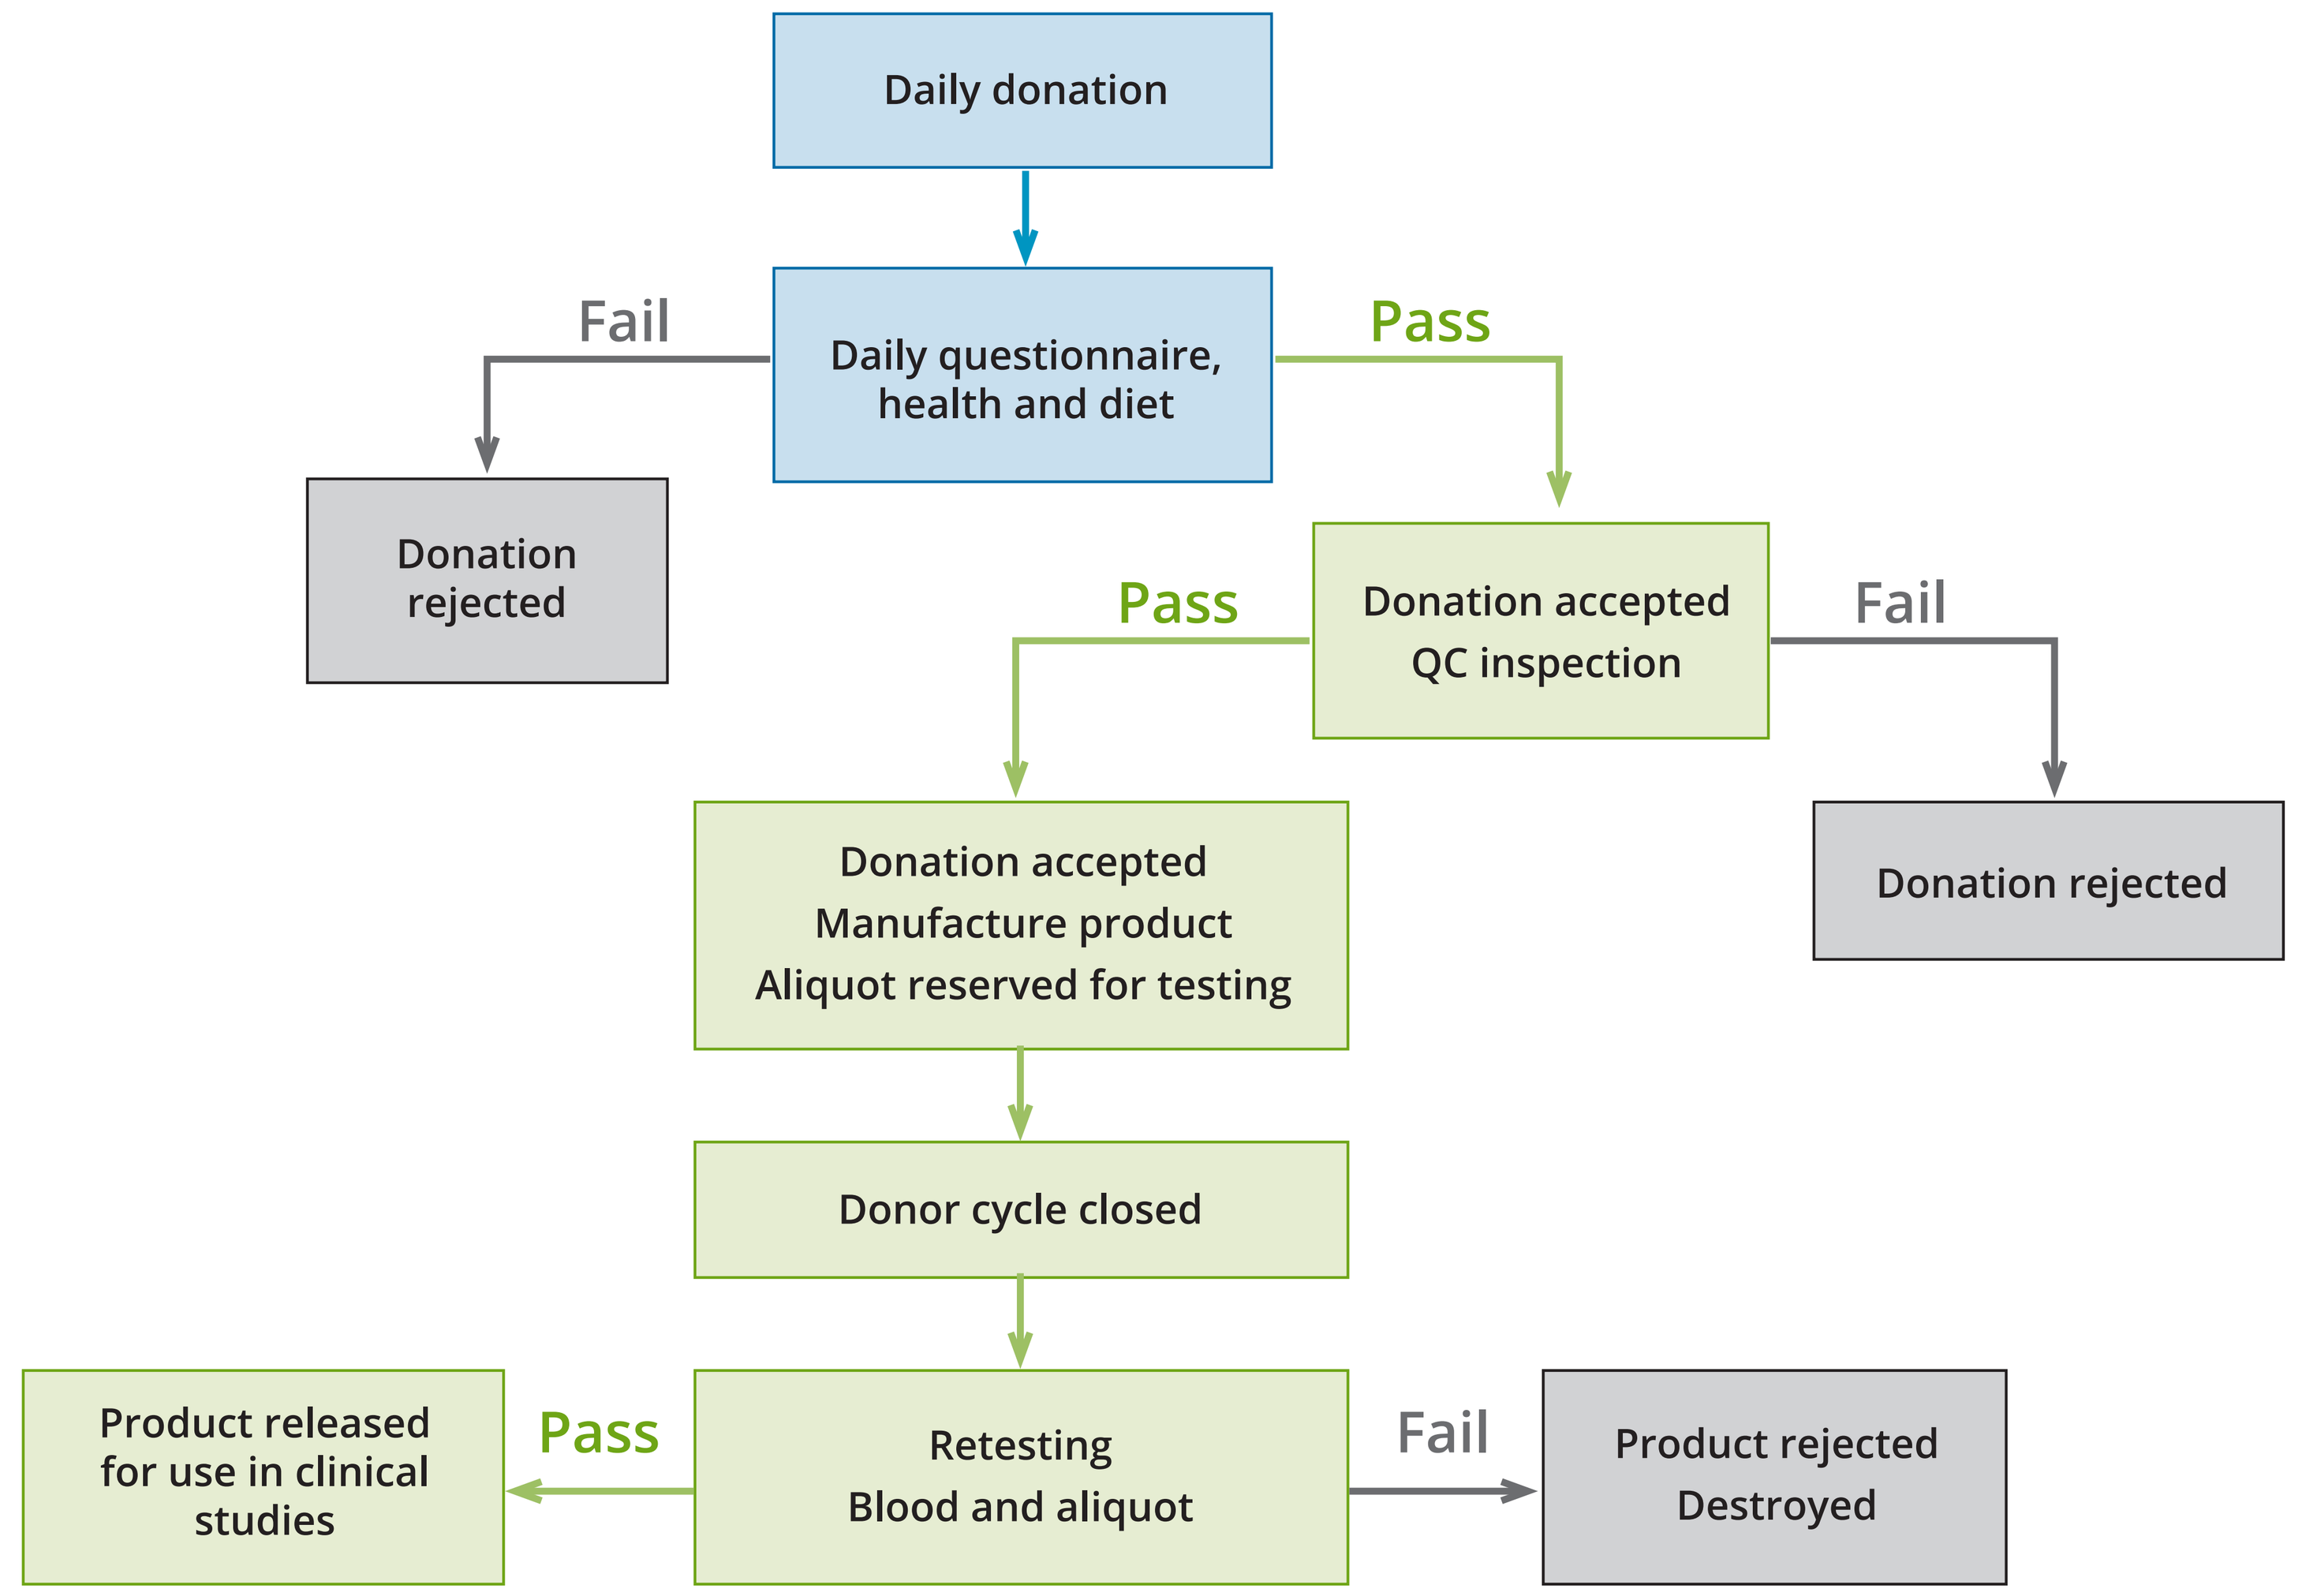
Daily donation
Fail
Daily questionnaire, health and diet
Pass
Donation rejected
Pass
Donation accepted
QC inspection
Fail
Donation accepted
Manufacture product
Aliquot reserved for testing
Donation rejected
Donor cycle closed
Product released for use in clinical studies
Pass
Retesting
Blood and aliquot
Fail
Product rejected
Destroyed

Figure 3. Donation Cycle

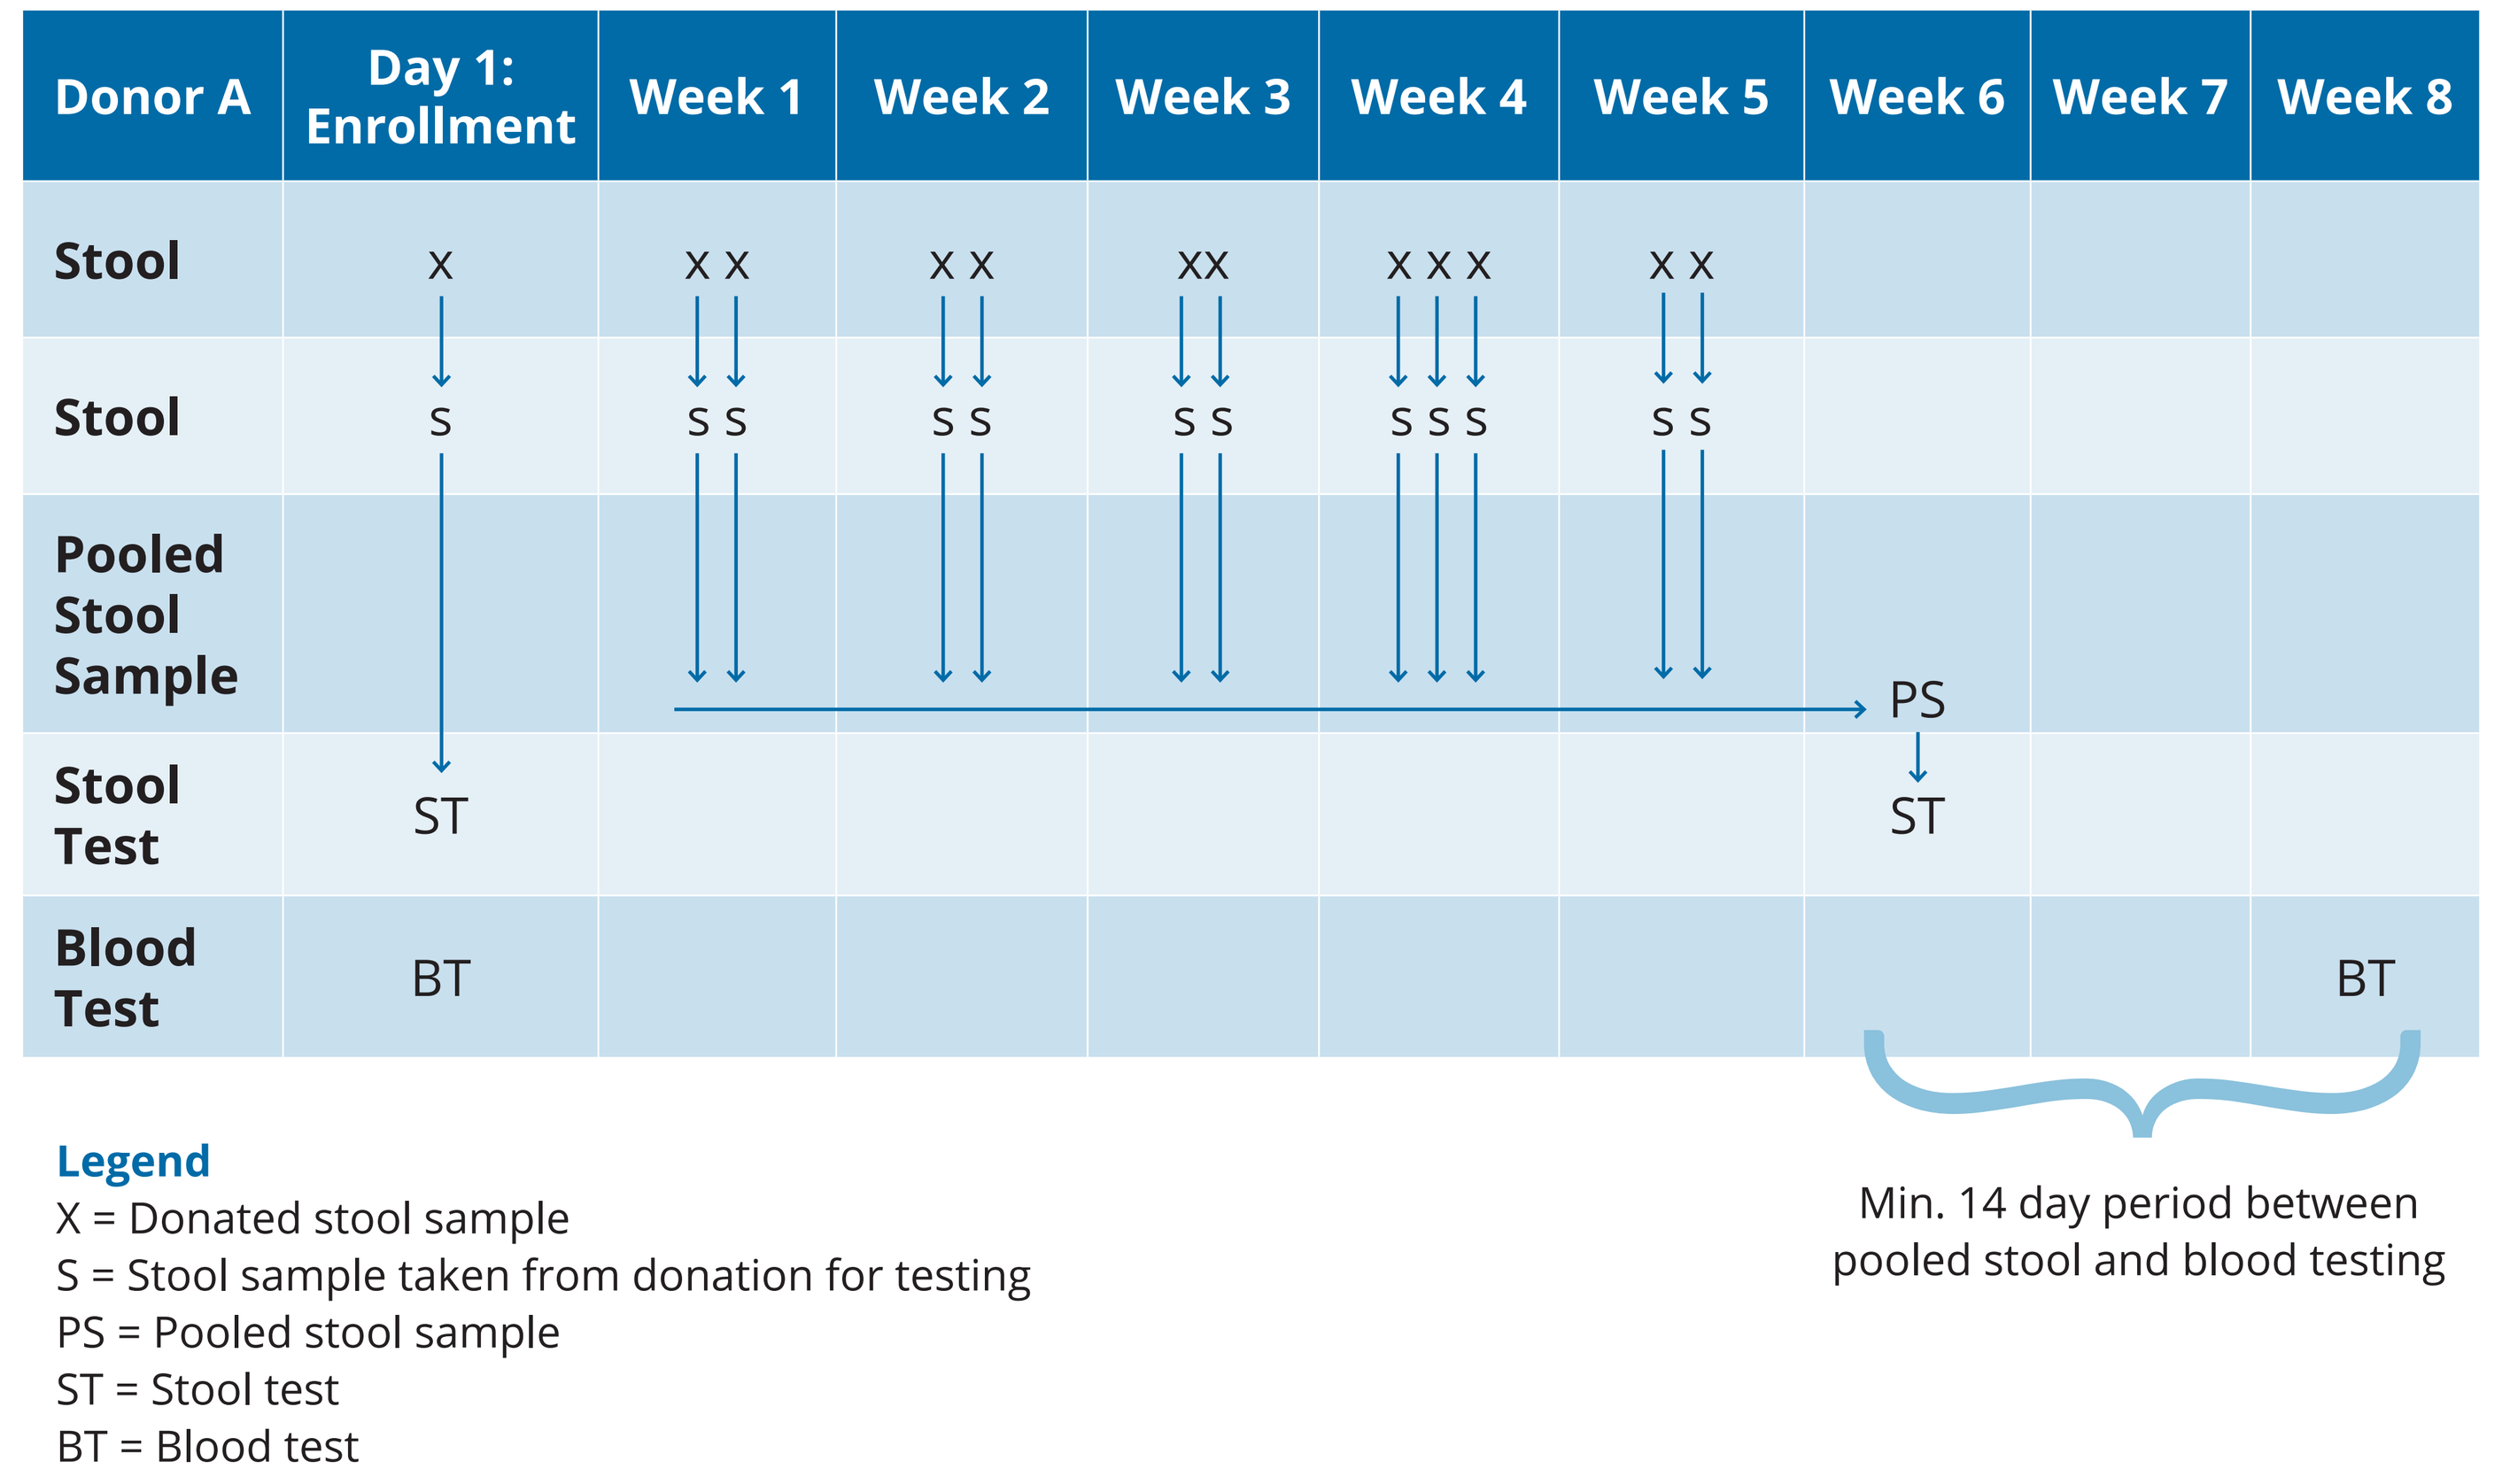
Donor A
Day 1: Enrollment
Week 1
Week 2
Week 3
Week 4
Week 5
Week 6
Week 7
Week 8
Stool
Stool
Pooled Stool Sample
Stool Test
Blood Test
X
S
ST
BT
X X
S S
X X
S S
XX
S S
X X X
S S S
X X
S S
PS
ST
BT
Min. 14 day period between pooled stool and blood testing
Legend
X = Donated stool sample
S = Stool sample taken from donation for testing
PS = Pooled stool sample
ST = Stool test
BT = Blood test

Results

- A total of 62 potential donors were recruited from July 15, 2013 through August 25, 2015.
- Of these, 11 (17.8%) failed the initial screening protocols.
- A total of 51 (82.2%) passed the screening protocols and made at least one donation.
- On subsequent retesting, a further 22 people (43% of the remaining donor pool) were excluded: 12 donors failed one or more of the blood and pathogen screens; 10 donors discontinued participation.

Donor Characteristics (N=62)

Parameter	Value
Mean, age, years	28 (19-59)
Gender, male	66%
Race, white	93.5%
Full or part-time student	53.2%

Initial Screen Failures (n=11)

Parameter	Value
Asymptomatic *E. Coli* strain #0157	1
Rheumatoid arthritis	1
Hepatitis C	1
Malaria (past infection)	1
Grave's disease	1
Antibiotic use within past 90 days	2
Incomplete screening	1
Psoriasis	1

Rescreen or Intra-Cycle Failures (n= 12)*

Parameter	Value
Asymptomatic norovirus	2
Asymptomatic rotavirus	2
Ova & parasites	3
Medications	1
Antibiotic use	3
Asymptomatic *Salmonella*	1
Vancomycin-resistant *Enteroccocus* (VRE)	1
Staphylococcus aureus	3

* A donor could be excluded for multiple reasons

Conclusions

- A substantial number of outwardly healthy donors used to source intestinal microbes for a drug under study for recurrent *Clostridium difficile* infection were found unsuitable due to underlying conditions.
- Reasons for rejection included asymptomatic carriage of an *E. coli* strain associated with hemorrhagic diarrhea and asymptomatic norovirus and rotavirus, which typically cause symptoms.
- Stringent screening of donors is required when sourcing live human-microbes used for treatment of disease.