Session 193
POSTER #212
June 3rd,, 2017
12:15 - 2:15 PM

Changing the Microbiome: Patients with a Successful Outcome Following Microbiota-Based RBX2660 Treatment Trend Toward Human Microbiome Project Healthy Subjects' Profile

Ken Blount, PhD[1], Courtney Jones, BS[1], Bill Shannon, PhD MBA[2], Sharina Carter, PhD[2]

[1]Rebiotix Inc, Roseville, MN, [2]BioRankings LLC, St. Louis, MO

Corresponding Author

Ken Blount, PhD
2660 Patton Road
Roseville, MN 55113
Phone: 651-705-8770
Email: kblount@rebiotix.com

Background

- Recurrent *Clostridium difficile* infections (rCDI) are associated with decreased taxonomic diversity in patients' intestinal microbiome compared to healthy microbiomes.
- RBX2660 is a standardized microbiota-based drug designed to rehabilitate a patient's intestinal microbiome.
- The effect of RBX2660 on rCDI patient microbiomes was evaluated by comparing pre- and post-treatment samples collected from PUNCH CD 2—a multicenter, randomized, double-blind, placebo-controlled study—to the healthy intestinal microbiome defined by the Human Microbiome Project (HMP).
-

Methods

- Patients with rCDI enrolled in the PUNCH CD 2 trial were randomized to receive blinded treatment by enema of 2 doses of RBX2660 (Group A), 2 doses of placebo (Group B), or 1 dose of RBX2660 and 1 dose of placebo (Group C), with doses 7 days apart (Figure 1).
- Success was defined as the absence of *Clostridium difficile*-associated diarrhea at 8 weeks following completion of the last treatment. Patients were classified as a treatment failure if all four(4) of the following criteria were met: recurrence of diarrhea less than 8 weeks after administration of the last assigned study enema, a positive laboratory diagnosis of *C. difficile* as conducted and reported by the study investigator, a need for retreatment for CDI, and no other cause for diarrhea.
- Voluntary stool samples were collected from subjects at baseline and at 7, 30, and 60 days after treatment.
- Longitudinal 16s rRNA analysis using the Illumina MiSeq platform was performed on stool samples collected from subjects in Groups A and C with successful outcomes. The variable region V4 was targeted to identify the operational taxonomic units (OTUs) in each sample.
- Simpson and Shannon diversity values were calculated for all sample time point groups, and post-treatment diversity indices for each time point were compared to baseline using a paired Wilcoxon test.
- Relative abundance data from subject samples were grouped longitudinally and compared to HMP data using a Bray-Curtis dissimilarity calculation with non-metric multi-dimensional scaling. Additional analyses were performed based on the Dirichlet-Multinomial distribution to compare group mean relative taxonomic abundances, pi, and within group over dispersion, theta.
- Differentiation between sample communities were visualized using a Kullback-Leibler (KL) divergence analysis model (BioRankings, St. Louis, MO) KL divergence is a measure of the difference between microbial diversity at different time points or between different samples.

Figure 1: PUNCH CD 2 Trial Design

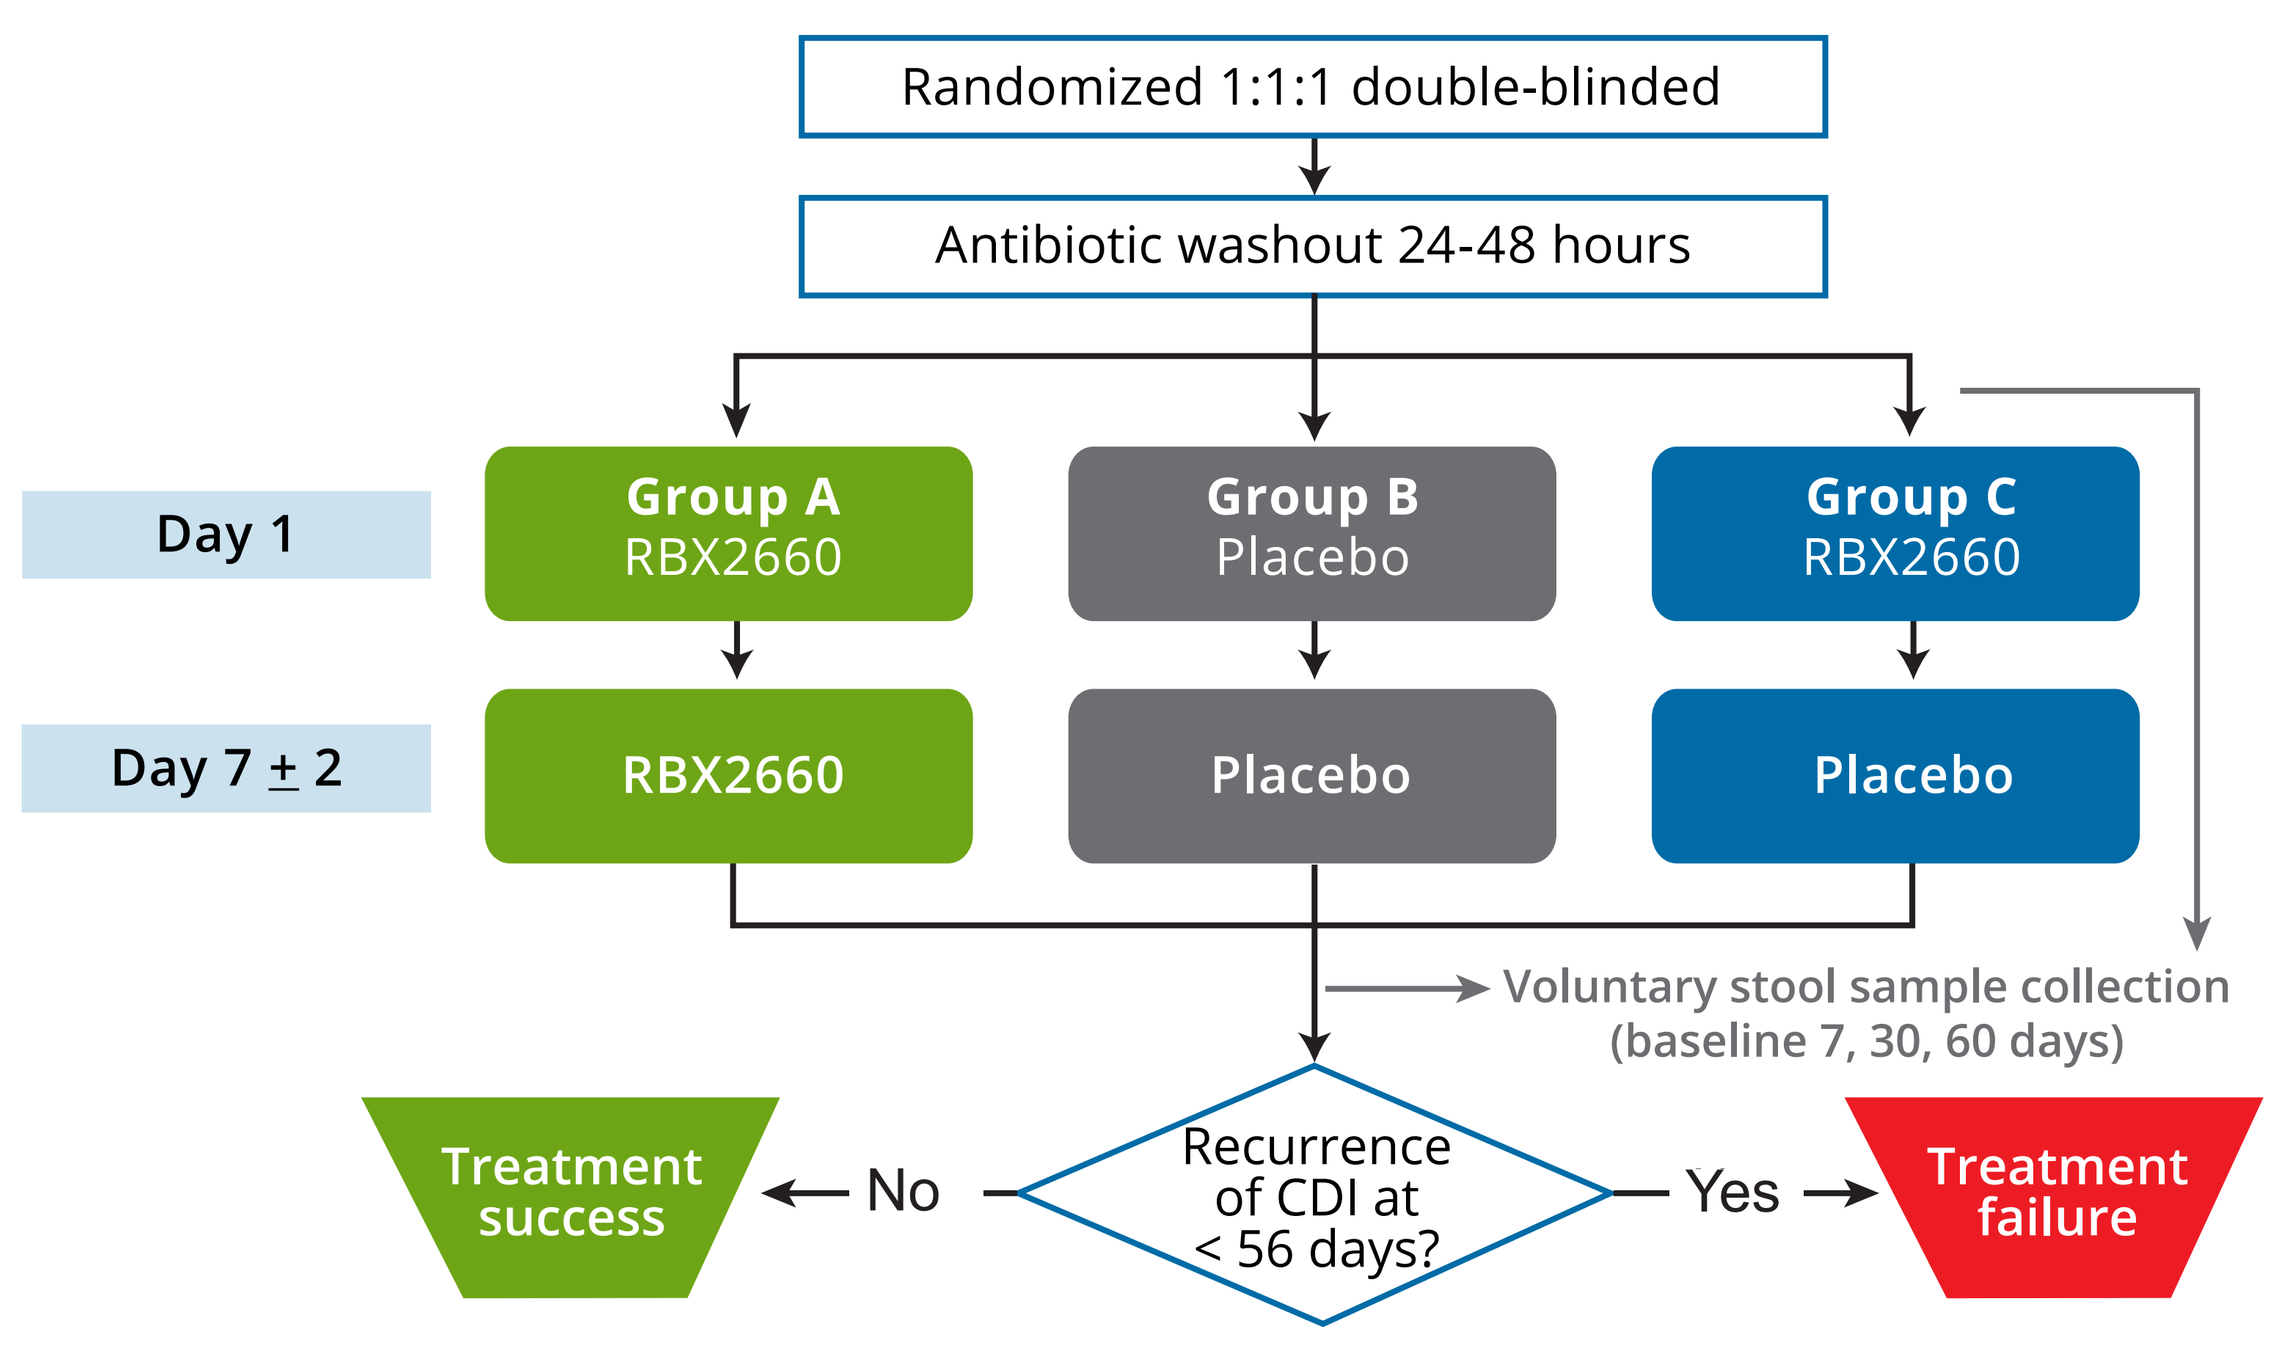
Randomized 1:1:1 double-blinded
Antibiotic washout 24-48 hours
Day 1
Day 7 ± 2
Group A
RBX2660
Group B
Placebo
Group C
RBX2660
RBX2660
Placebo
Placebo
Voluntary stool sample collection
(baseline 7, 30, 60 days)
Treatment success
No
Recurrence of CDI at < 56 days?
Yes
Treatment failure

PUNCH CD 2 Clinical Trial Results[1]:

- A single dose of RBX2660 was significantly better than placebo (67%, Group C vs 46% Group B, P=0.48) (Figure 2).
- A second dose of RBX2660 one week later (Group A) did not provide additional clinical benefit.
- Groups A and C combined were superior to placebo (P=0.046)

Figure 2: Patients who received at least one dose of microbiota-based drug RBX2660 showed greater efficacy than placebo-treated patients following blinded treatment.

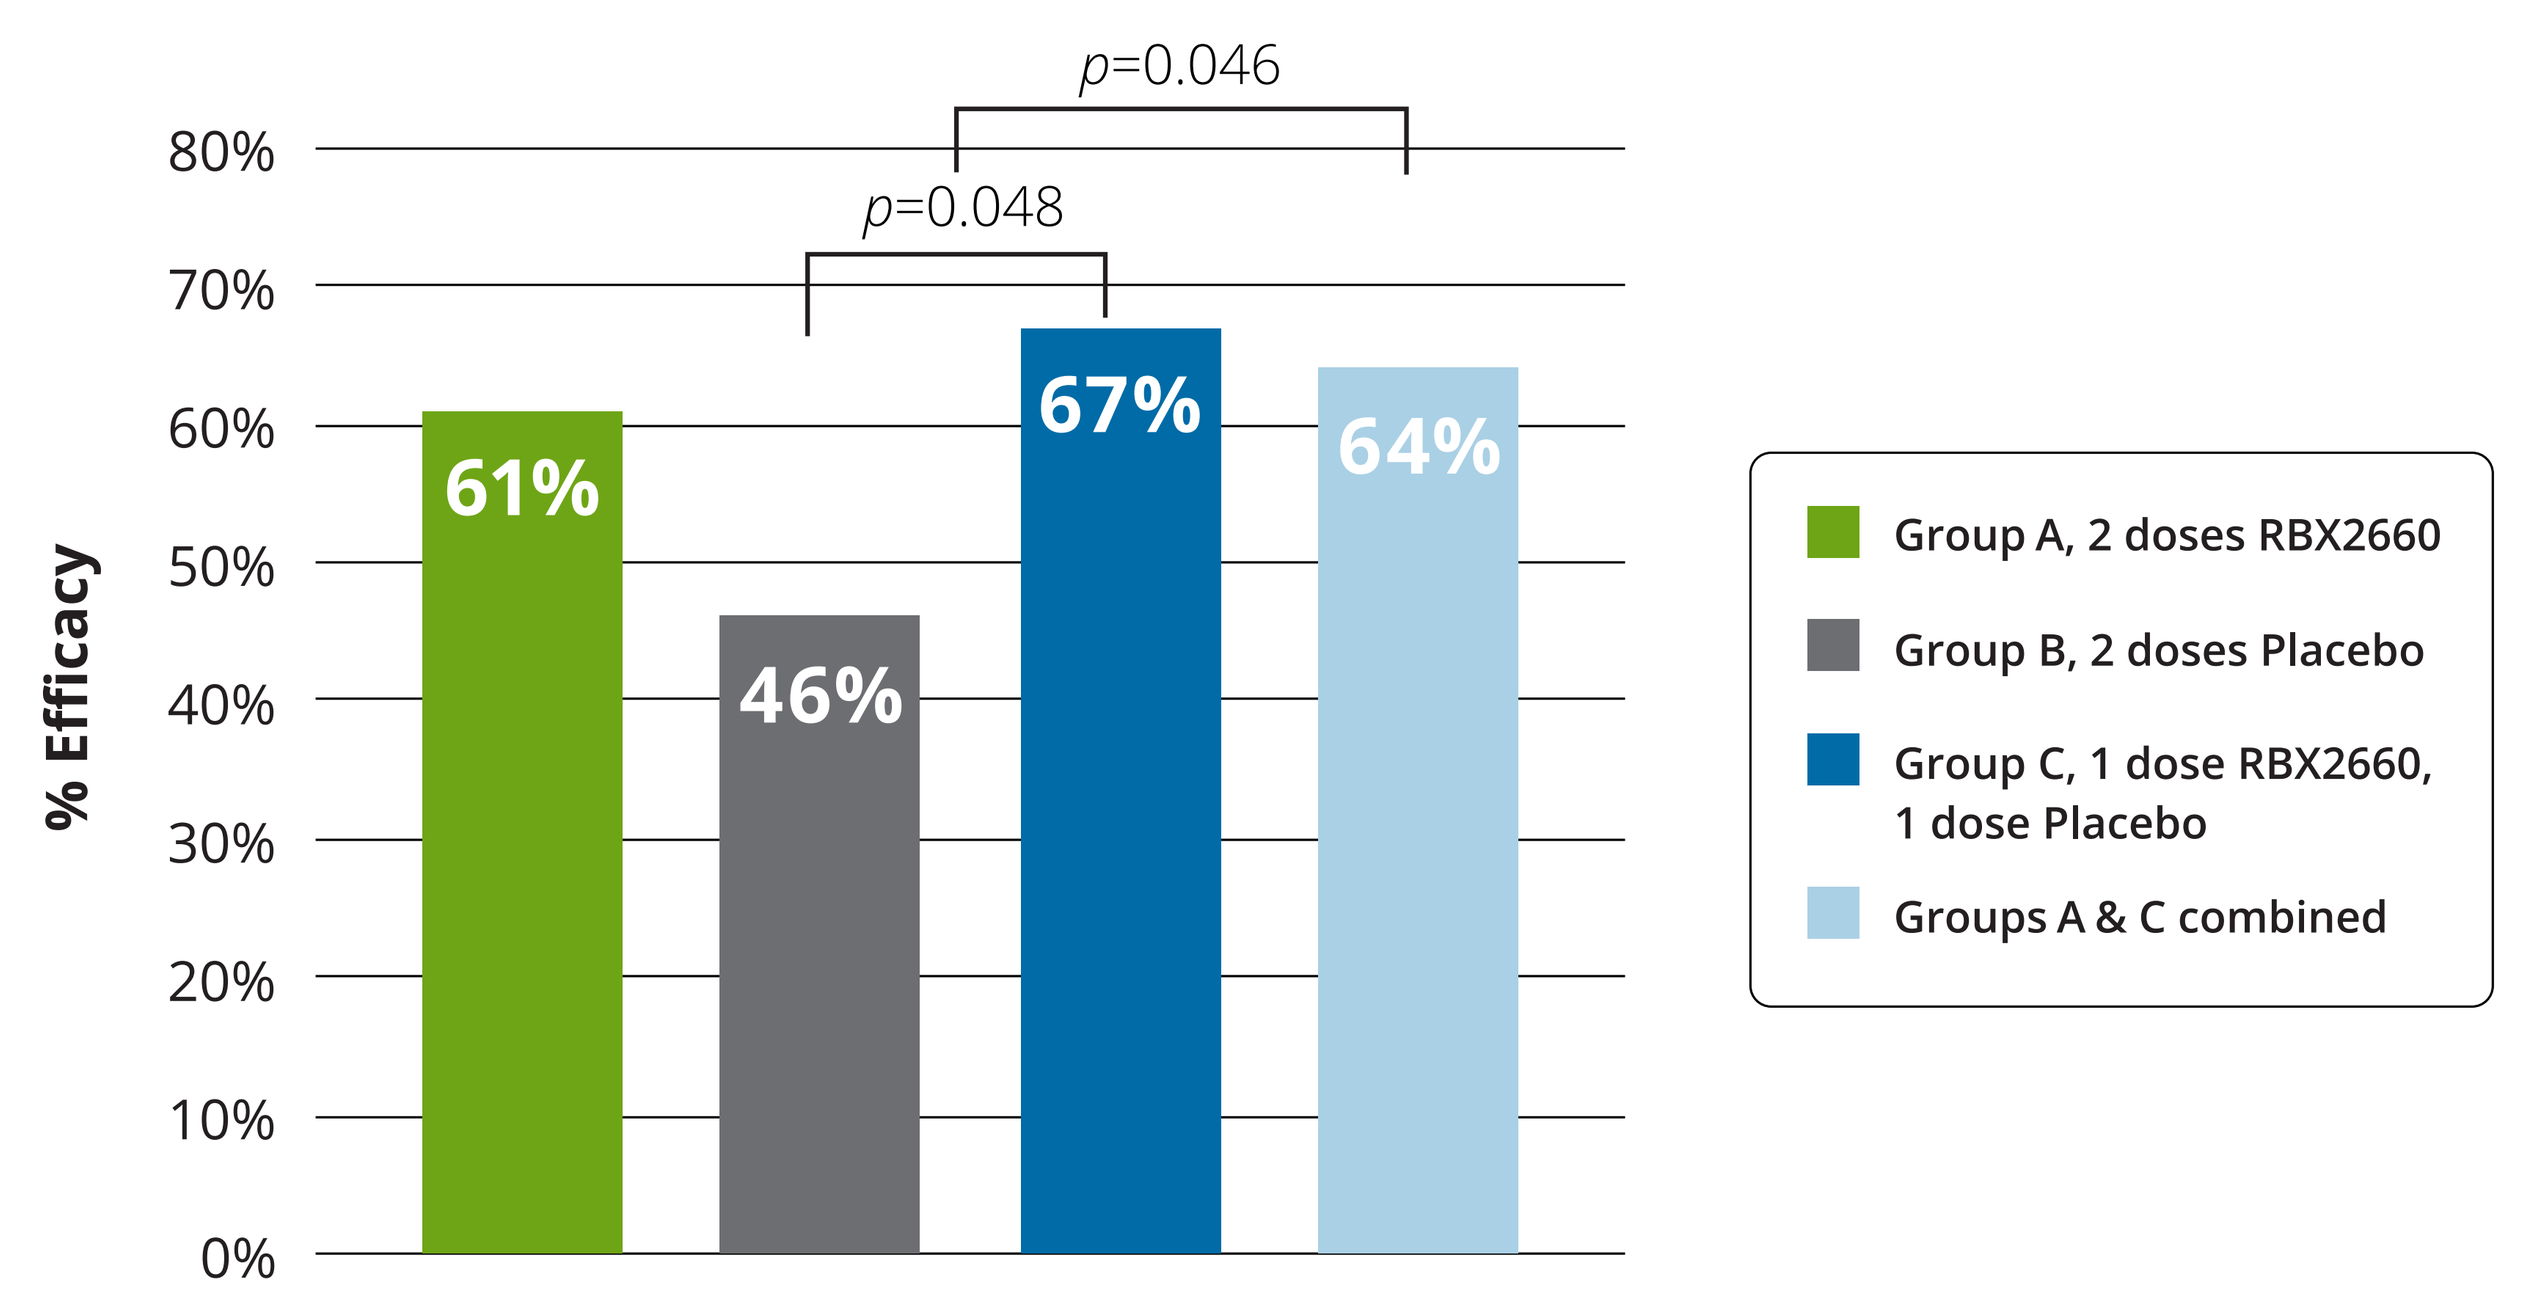
p=0.046
p=0.048
80%
70%
60%
50%
40%
30%
20%
10%
0%
% Efficacy
61%
46%
67%
64%
Group A, 2 doses RBX2660
Group B, 2 doses Placebo
Group C, 1 dose RBX2660, 1 dose Placebo
Groups A & C combined

Reference: [1]Dubberke ER et al. Efficacy and Safety of RBX2660 for the Prevention of Recurrent Clostridium difficile Infection: Results of the PUNCH CD 2 Trial Infectious Diseases Week (ID Week) October 26-30, 2016, New Orleans, LA

Results

- Ninety-four (94) stool samples collected from 45 patients who received at least one dose of RBX2660 (Groups A and C) were analyzed (Table 1).

Table 1

	# Subjects	Product Samples	Baseline	7 days	30 days	60 days
Group A responders	21		12	17	13	11
Group C responders	21	136	12	18	14	16
Groups A/C failures	23		23	14	7	--

- RBX2660 significantly increased microbiome diversity compared to baseline (Table 2)
- Patient baseline microbiomes were less diverse and more divergent from the "healthy" HMP microbiome. After treatment, patient microbiomes became more closely related to the HMP over time, with the largest shift observed after 7 days (Figures 3, 4, 5).
- Specific taxonomic distributions were consistent with pres microbiome analyses of CDI patients. Taxonomic classes that diverged the most at baseline showed the largest longitudinal post-treatment change. (Figure 4).

Table 2

	Simpson Diversity		Shannon Diversity	
Time	Mean	*p* value Compared to baseline	Mean	*p* value Compared to baseline
Baseline	0.422	---	0.806	---
7 days, Success	0.554	0.002	1.118	<0.001
30 days, Success	0.561	0.004	1.158	<0.001
60 days, Success	0.532	0.003	1.059	0.002

Figure 3: Microbiomes from patients with recurrent CDI shift towards donor biomes following RBX2660 treatment.

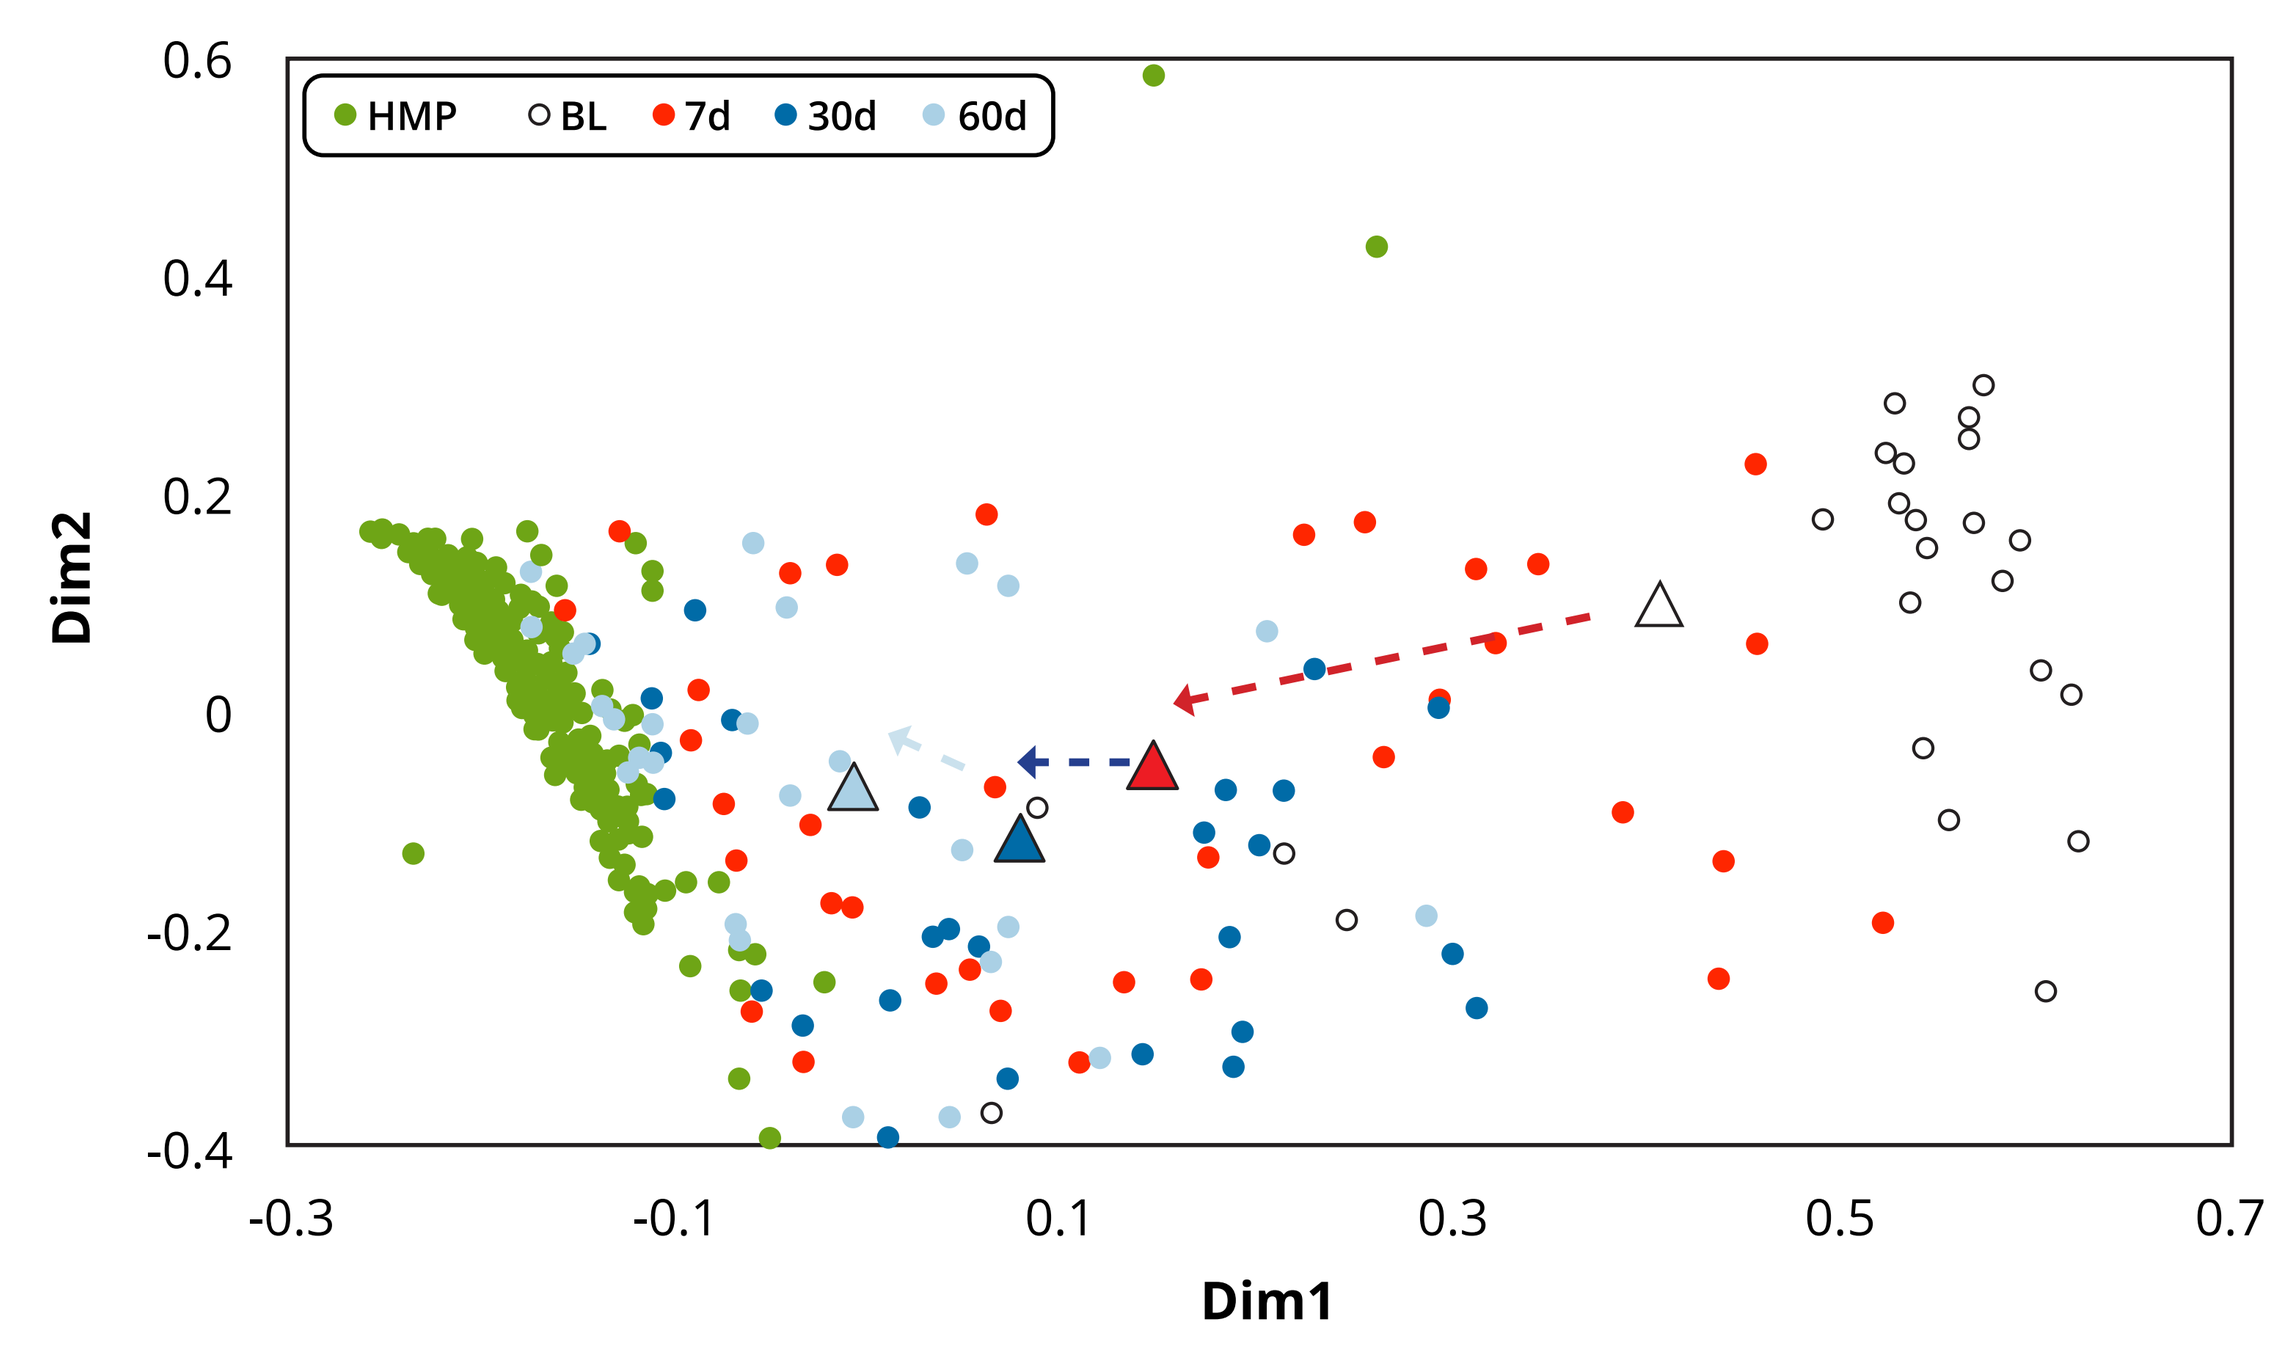
HMP
BL
7d
30d
60d
0.6
0.4
0.2
0
-0.2
-0.4
Dim2
-0.3
-0.1
0.1
0.3
0.5
0.7
Dim1

Comparisons were made at the sample level using non-metric multidimensional scaling based on the Bray Curtis measure. In the plot above, mean microbiomes for individual patients (dots) and group mean microbiomes (triangles) are shown. Arrows highlight the shift of group means from baseline toward cluster of HMP samples (right side of the plot; green).

Figure 4: Patient taxa levels approach HMP levels over time after RBX2660 treatment.

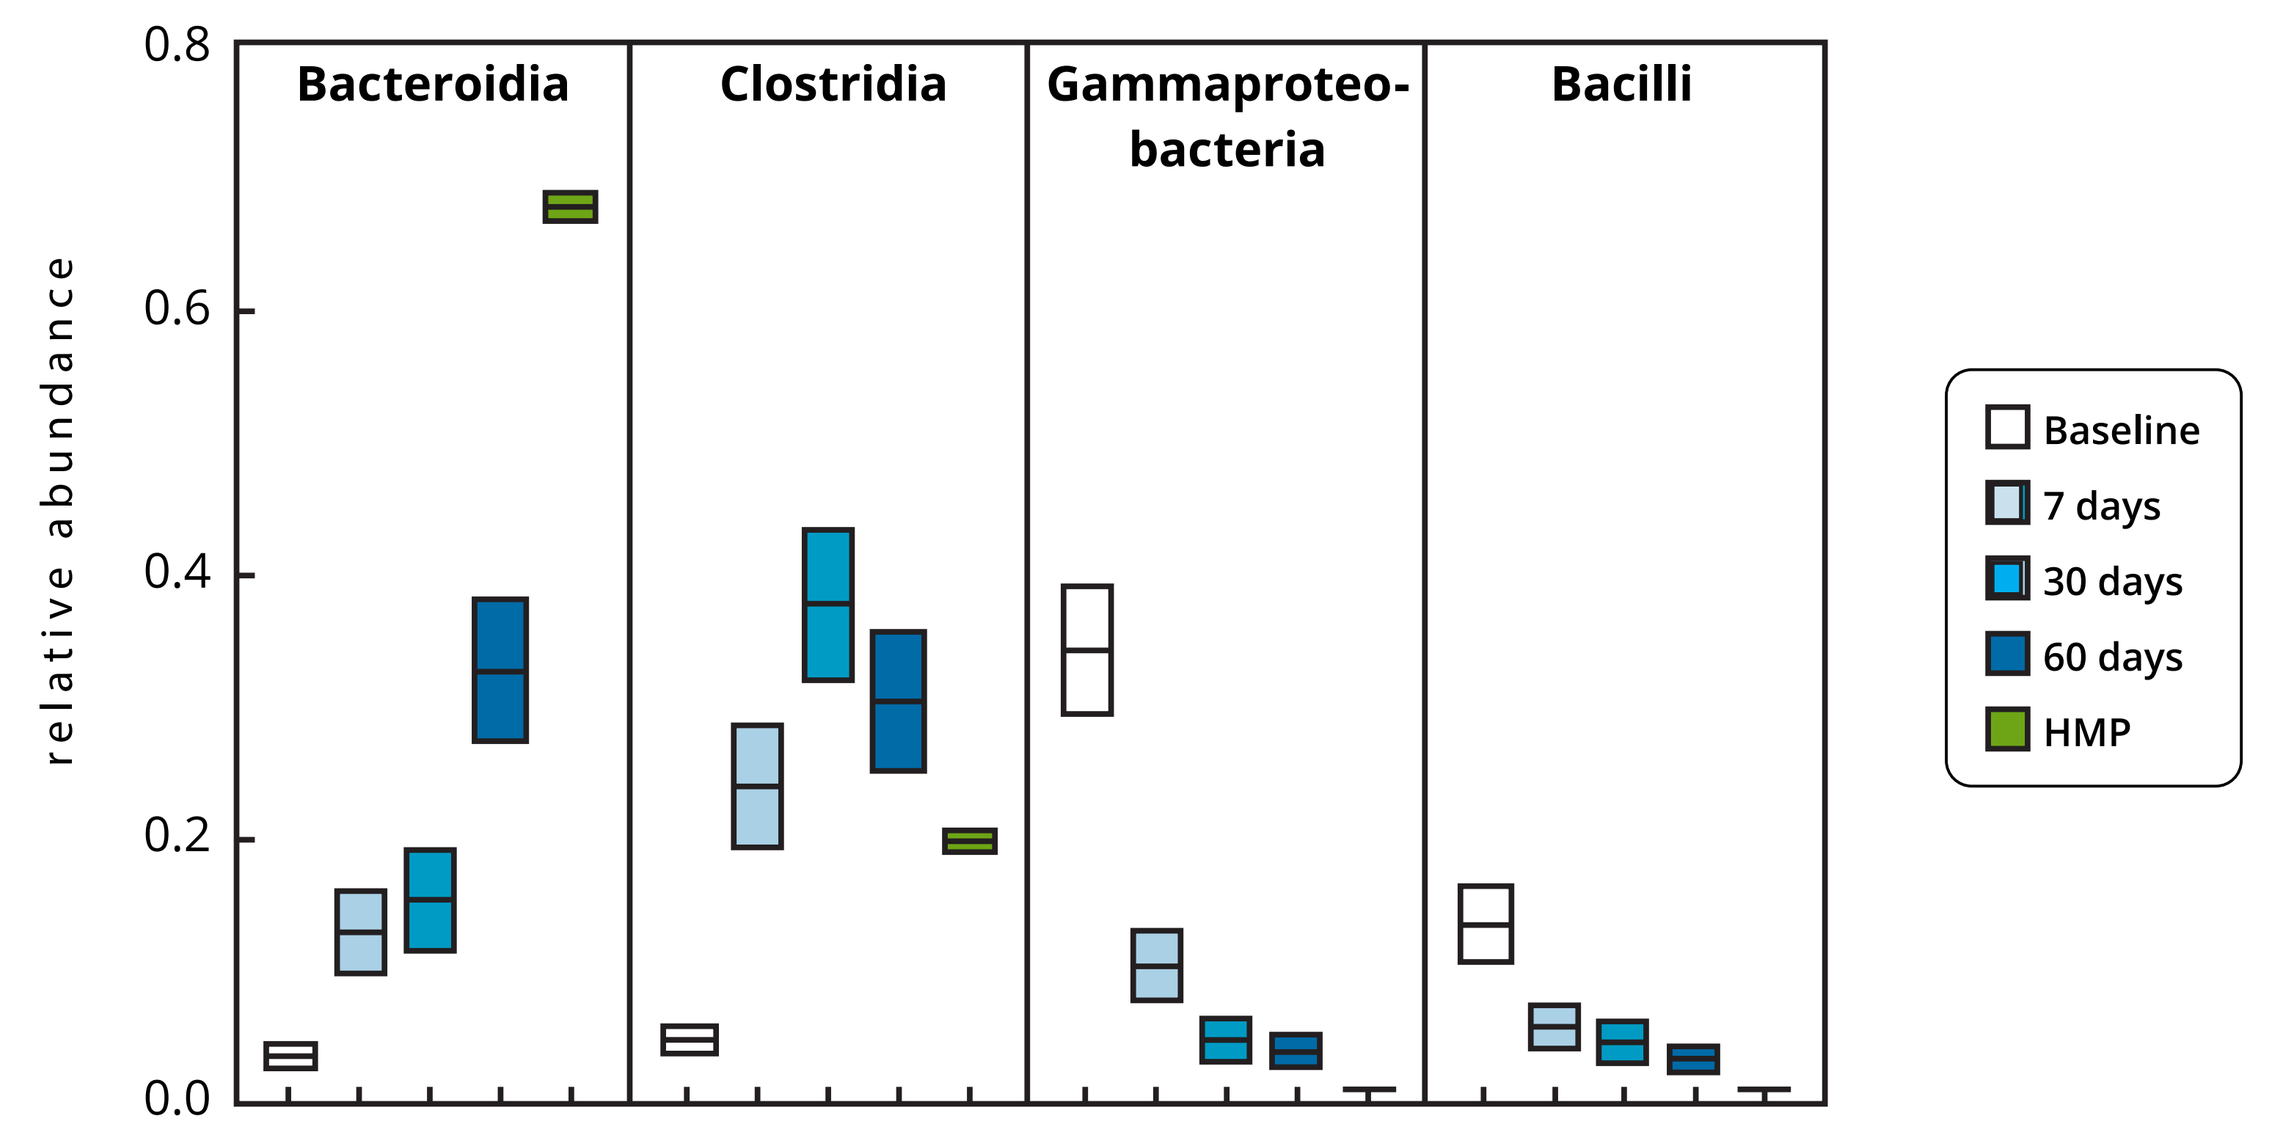
0.8
0.6
0.4
0.2
0.0
relative abundance
Bacteroidia
Clostridia
Gammaproteo-
bacteria
Bacilli
Baseline
7 days
30 days
60 days
HMP

Dirichlet Comparisons were made at the taxa level using Dirichlet Multinomial parameter pi, representing the group's mean relative taxa abundance, at each time point. A) The graph shows that patients' mean taxa levels approach the HMP levels over time. B) Plots of individual taxa demonstrating the greatest change over time, all samples and group means are shown. The plot shows patients shifting from baseline (right side of the plot) towards the cluster of donors (triangles). The group means (triangles) represent the same trend.

Figure 5: Microbiomes from patients with successful response to RBX2660 have microbiomes more similar to HMP microbiomes.

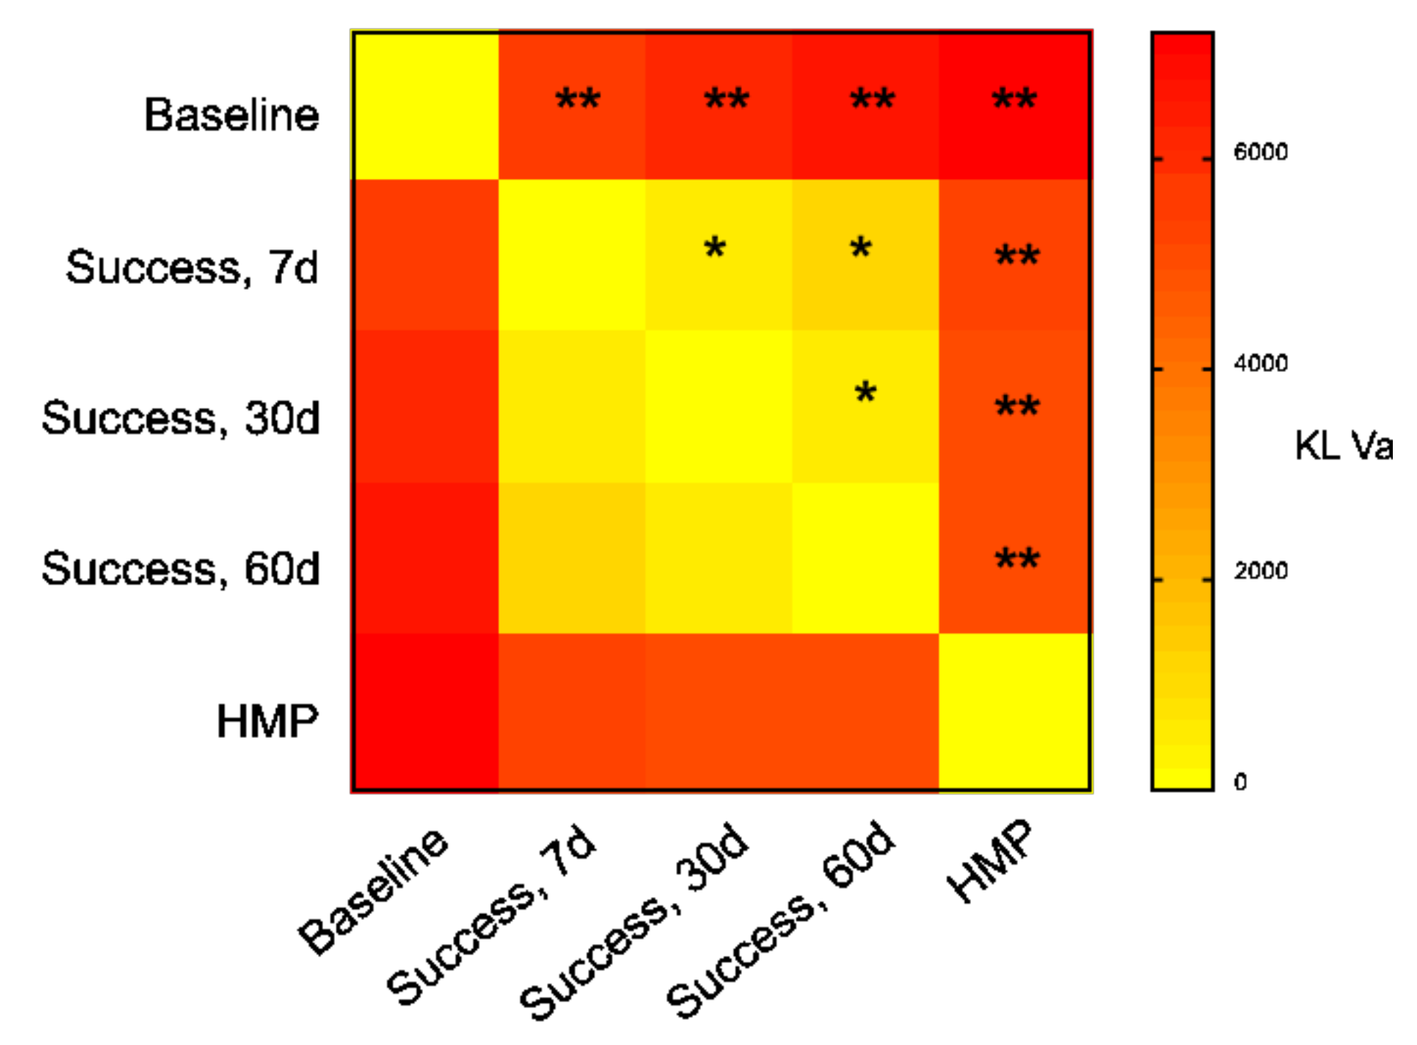
Baseline
Success, 7d
Success, 30d
Success, 60d
HMP
6000
4000
2000
0
KL Va

Kullback-Leibler (KL) divergence analysis assesses the difference between microbial diversity at different time points or between different samples. KL divergence increases as color ranges from yellow to red. Similarity between two microbiomes decreases as KL divergence increases. Differences between group mean microbiomes were compared using a generalized Wald type test to determine whether the difference between test groups were significant as noted. ** p<0.001; * p<0.05

Conclusion

Results show RBX2660 treatment shifts patient intestinal microbiomes closer to the healthy microbiome as defined by the Human Microbiome Project. In addition, RBX2660 significantly increases microbiome diversity compared to baseline.

Disclaimer: This analysis was funded by Rebiotix Inc., Roseville, MN.