Clearance of Vancomycin-Resistant *Enterococcus* Concomitant with Administration of a Microbiota-Based Drug Targeted at Recurrent *Clostridium difficile* Infection

Erik Dubberke, MD, MSPH, Washington University School of Medicine, St. Louis, MO; Courtney Jones, BS, Rebiotix Inc., Roseville, MN

Background:

- Vancomycin-resistant *Enterococcus* (VRE) is a major nosocomial pathogen and is a well-known complication among transplant and immune compromised patients.[1]
- VRE colonisation and *Clostridium difficile* infection (CDI) share similar risk factors; antibiotic-induced perturbation of the gut microbiota may contribute to both.[2]
- VRE carriers are at increased risk for infection and a source of transmissions to others.
- Controlling transmission in a healthcare setting can be challenging.[3]
- Fecal microbiota transplantation (FMT) may be effective in clearing VRE and restoring colonisation resistance.[4]
- We report on clearance of VRE in stool in a secondary analysis of the PUNCH CD study, a Phase 2 study assessing the safety and efficacy of RBX2660 (microbiota suspension), in a population of patients with recurrent CDI.

Methods:

- A total of 34 patients with recurrent CDI enrolled at 11 sites in the U.S. received 1 or 2 doses of RBX2660, a next-generation FMT drug, between August 2013 and January 2014 as part of the PUNCH CD study.
- Patients were requested to voluntarily submit stool samples at baseline and at 7, 30 and 60 days and 6-months after the last administration of RBX2660.

PUNCH CD Study

Primary Objective: Assess the safety of RBX2660 for recurrent CDI infection
Design: Phase 2 open label
Participants and Location: 40 patients at 11 sites across the U.S.

Major inclusion criteria:

- Age ≥ 18 years old
- At least two recurrences of CDI after a primary episode and has completed at least two rounds of standard-of-care oral antibiotic therapy
 Or,
- At least two episodes of severe CDI resulting in hospitalization
- Taking or will start a course of oral antibiotics for CDI symptoms for 10-14 days
- At least 7 days of oral vancomycin (125 mg 4 x/day (500 mg) for the last 7 days of the regimen

Major exclusion criteria:

- History of IBD, ulcerative colitis, Crohn's disease, or microscopic colitis; IBS; chronic diarrhea; celiac disease
- Colostomy
- Evidence of active, severe colitis
- Planned surgery requiring perioperative antibiotics within 6 months of study enrollment
- Compromised immune system
- Neutropenia (white blood cell count <1000 cells/μL)

Follow-up: 7 days; 30 days; 60 days; 3 months; 6 months

RBX2660 (microbiota suspension)

- Designed to mimic FMT
- Raw material human stool
- Donors rigorously screened
- Contains a minimum guaranteed quantity of microbes
- Formulated to have a long shelf-life
- Manufactured using standardised, quality controlled processes
- Supplied in a ready-to-use enema format, 50g/150 mL dose

VRE TESTING PROTOCOL

Test type: Bile esculin azide agar with 6 μg/ml vancomycin (Remel, Lenaxa, Kansas, USA)
Specimens: Stool samples shipped fresh to laboratory via overnight delivery
Laboratory: Fairview Diagnostic Laboratories, Minneapolis, MN, USA
VRE confirmation: Blood agar and etest
Handling specification: Fresh stool, within 72 hours of collection; refrigerated if not tested directly after deposit

RELATIONSHIP OF RBX2660 ADMINISTRATION TO VRE TESTING

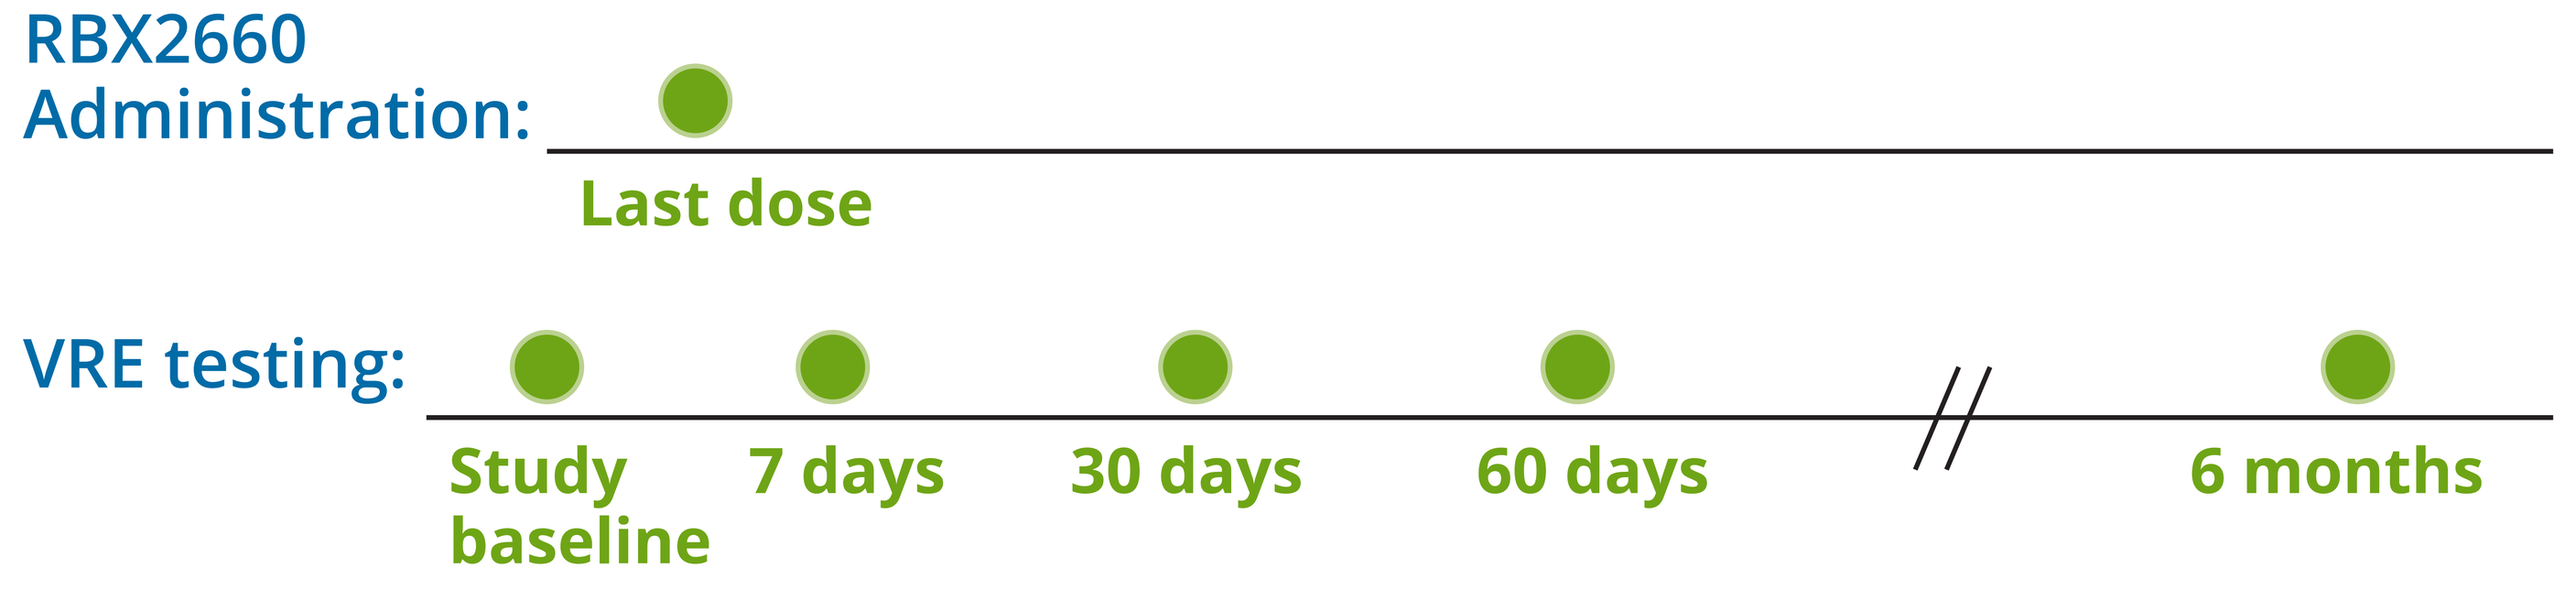
RBX2660 Administration:
Last dose
VRE testing:
Study baseline
7 days
30 days
60 days
6 months

Results:

PATIENTS

- During the study period, 30 patients with recurrent CDI had at least 1 stool sample available for VRE testing. All submitted samples were deemed acceptable by the independent laboratory used for testing.
- All stool samples for 19 patients tested VRE negative; 11 patients were VRE positive at the first test (baseline or 7-day follow-up).
- Of the patients who tested VRE positive at least once, 72.7% (n=8) converted to negative as of the last available follow-up.

BASELINE CHARACTERISTICS

Parameter	Total n=30	Always VRE (-) n=19	At least 1 VRE (+) n=11
Mean age, range, y	66.7 (26.7-89.5)	61.7 (26.7-77.8)	75.5 (47.6-89.5)
Female sex	20 (66.6%)	13 (68.4%)	7 (63.6%)
Caucasian	29 (96.6%)	19 (100%)	10 (90.9%)
Mean BMI, range k/m^2	24.4 (15-37)	23.9 (15-31)	25.3 (19-37%)
History of cardiovascular disease	16 (53.3%)	10 (52.6%)	6 (54.5%)
History of gastrointestinal comorbidity	18 (60%)	11 (57.9%)	7 (63.6%)
History of neurological comorbidity	5 (16.6%)	5 (26.3%)	0 (0%)

VRE STATUS AFTER RBX2660 ADMINISTRATION

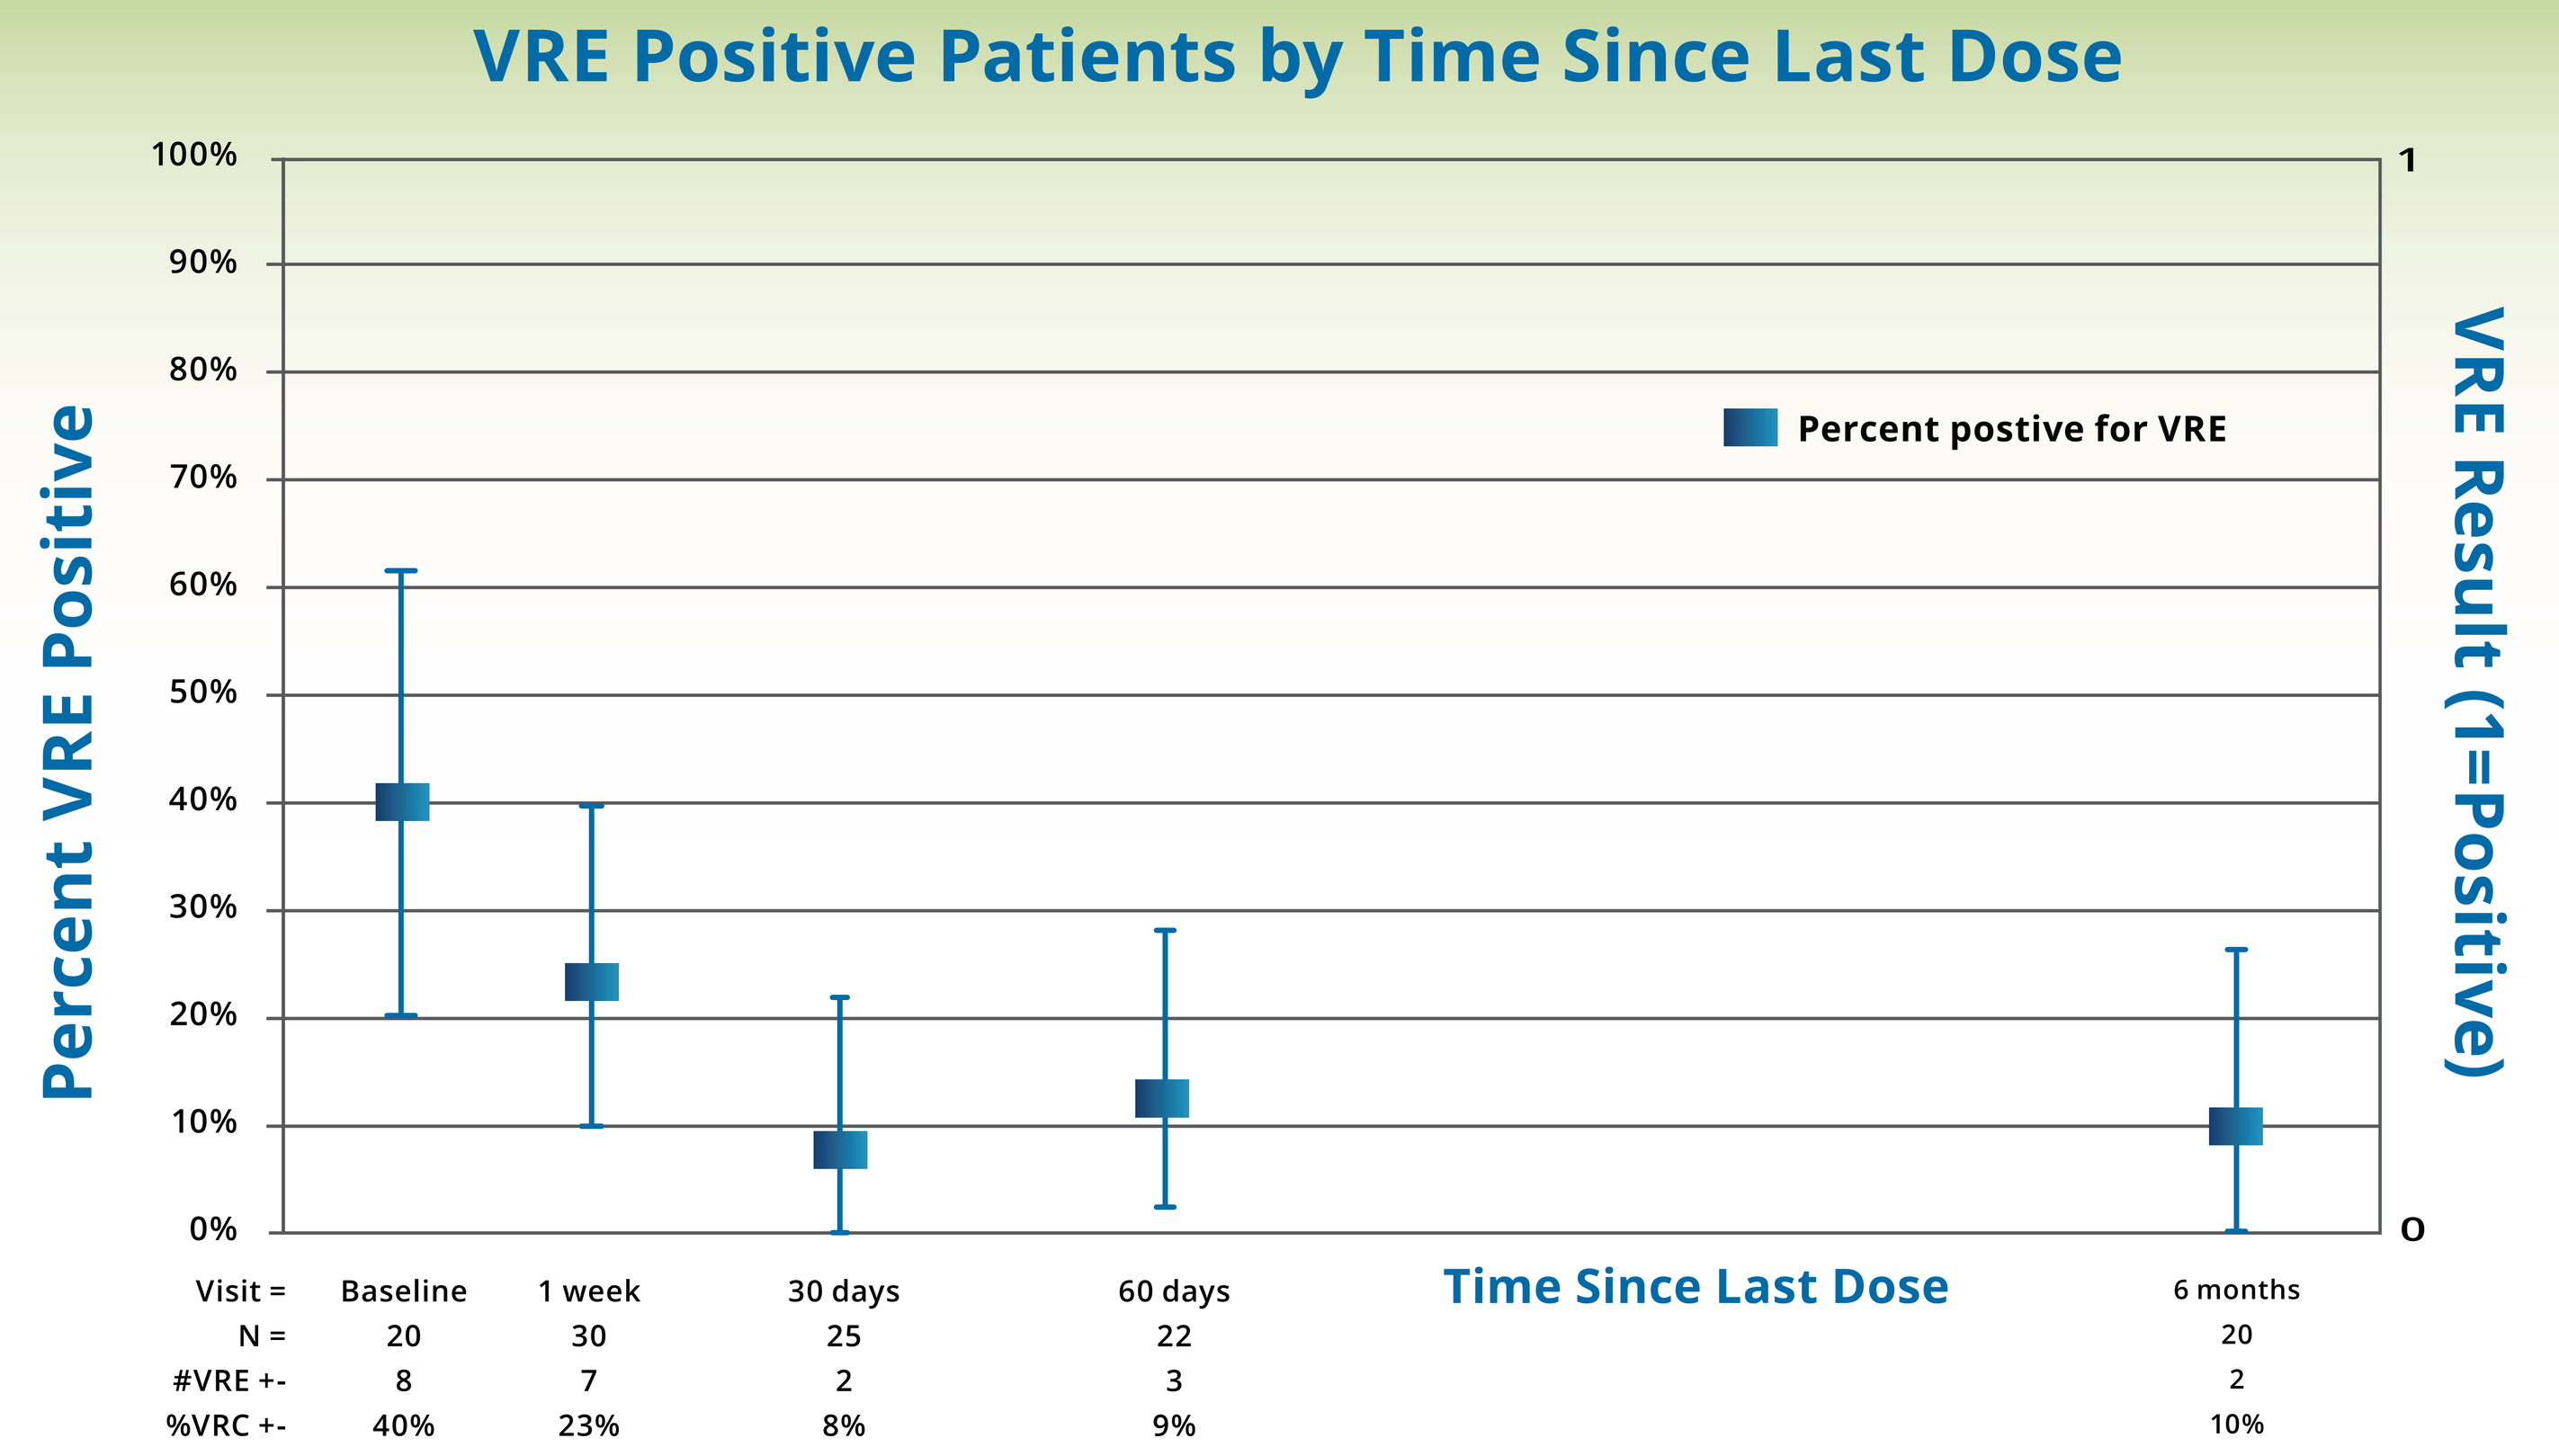
VRE Positive Patients by Time Since Last Dose
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Percent VRE Positive
VRE Result (1=Positive)
1
0
Percent postive for VRE
Time Since Last Dose
Visit = Baseline 1 week 30 days 60 days 6 months
N = 20 30 25 22 20
#VRE +- 8 7 2 3 2
%VRC +- 40% 23% 8% 9% 10%

- The percentage of patients testing VRE positive after the last dose of RBX2660 decreased over time.
- Status of the 3 VRE positive patients who did not convert at last follow-up:
 - 1 patient died (respiratory failure unrelated to RBX2660 administration); follow-up data was not available.
 - 1 patient remained VRE positive at all follow-ups.
 - 1 patient retested positive at 6 months after having negative tests in the interim; extenuating factors: multiple rounds of vancomycin for CDI recurrence.

Duration of Antibiotic Use Post-RBX2660 Administration

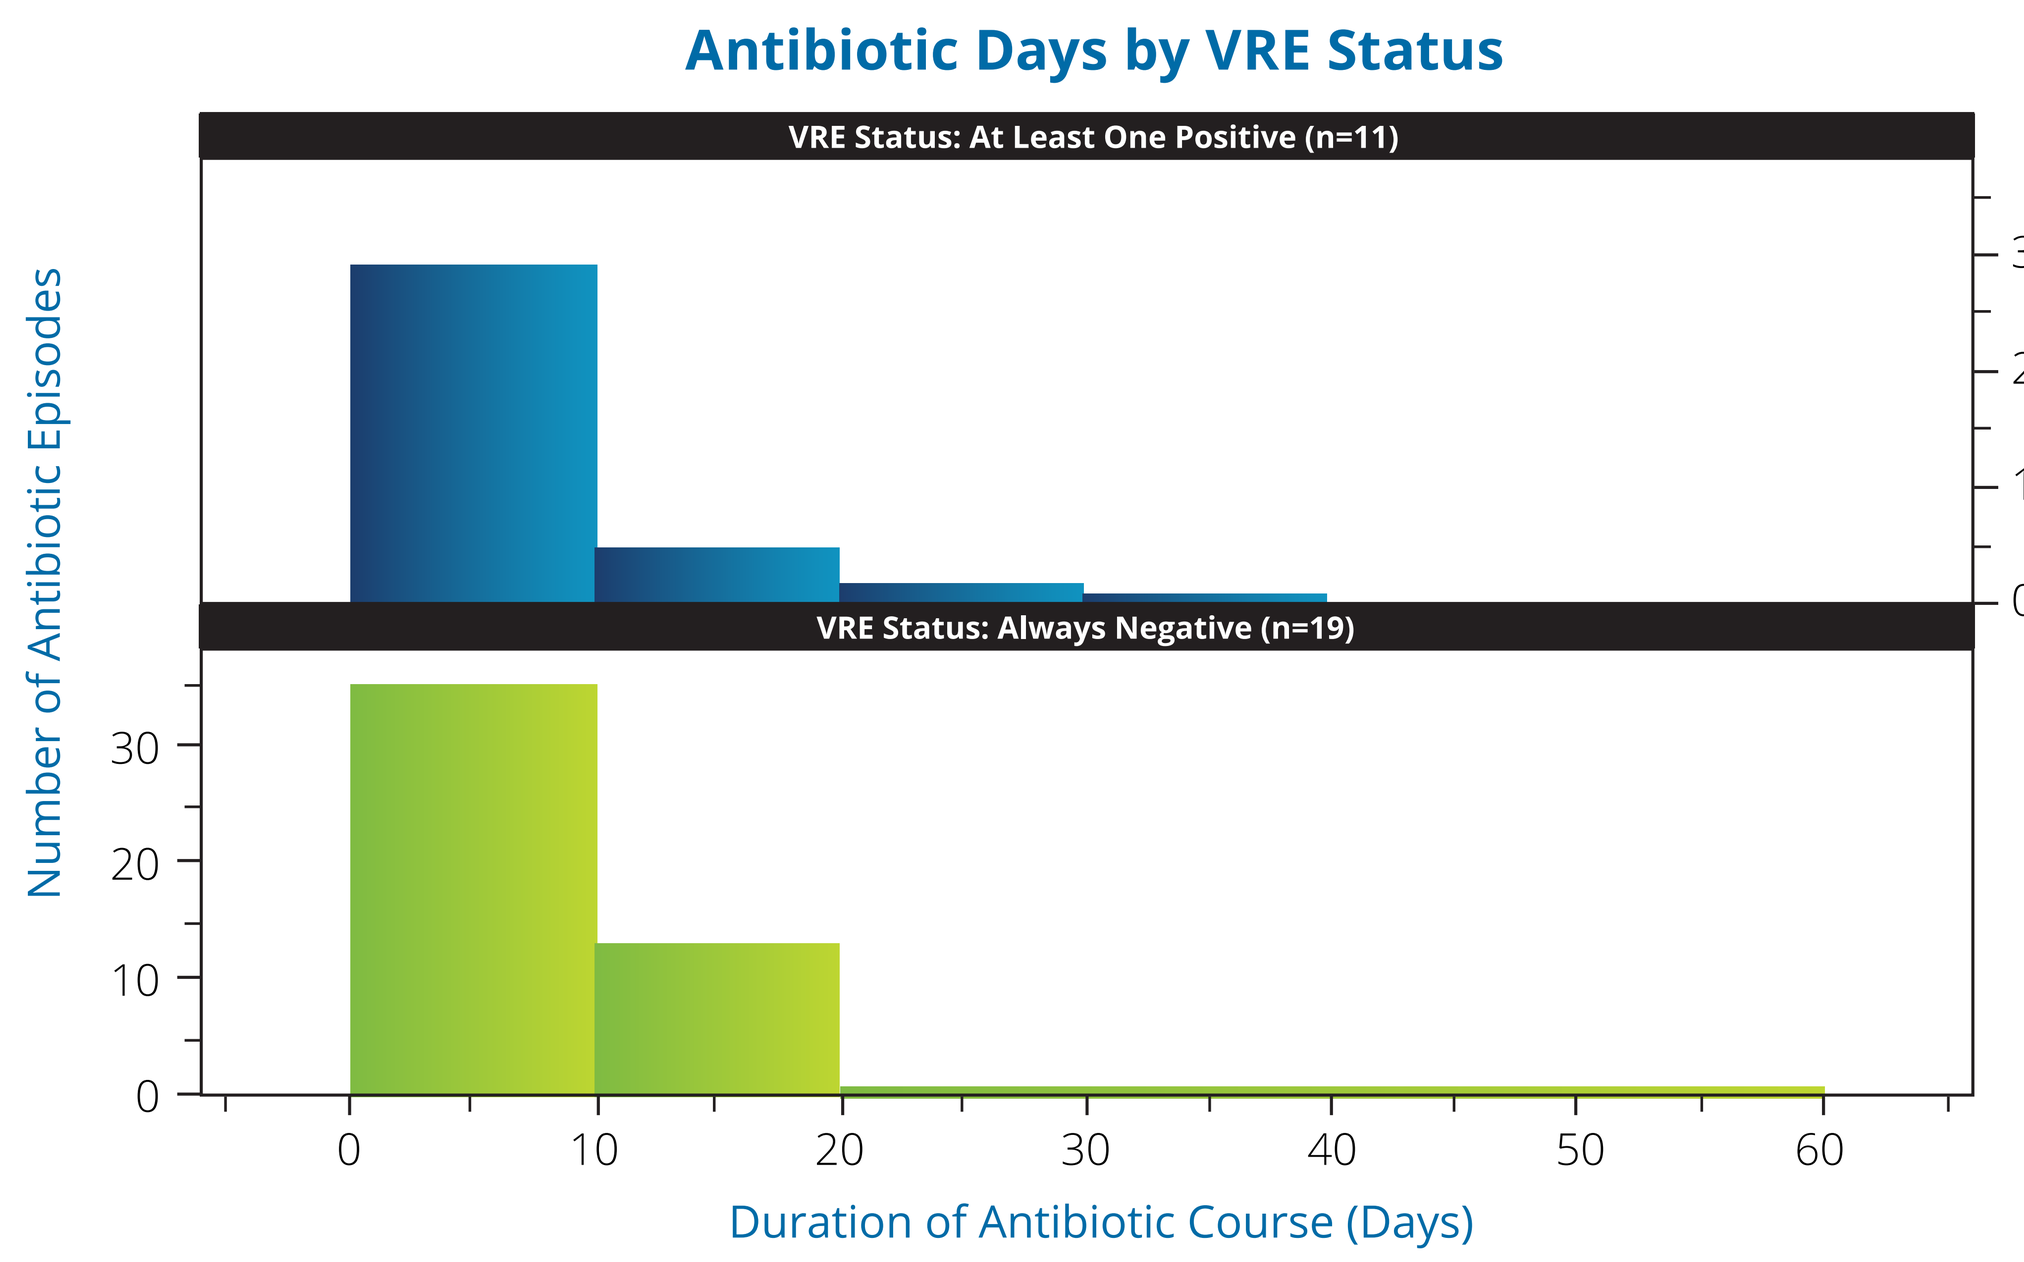
Antibiotic Days by VRE Status
VRE Status: At Least One Positive (n=11)
VRE Status: Always Negative (n=19)
Number of Antibiotic Episodes
0
10
20
30
0 10 20 30 40 50 60
Duration of Antibiotic Course (Days)

- Exposure to antibiotics post-RBX2660 was similar over time regardless of VRE status.
- Mean duration of antibiotic exposure post-RBX2660 administration was 1.6 (range: 0-10) days for patients who always tested VRE negative; 6.4 (range 1.0-14) days for patients who had at least 1 positive VRE test.
- Analysis of variance shows that antibiotic use was concentrated in a few patients: 14.3 days.

Relationship between Clearance of Recurrent CDI and VRE

- Considering patients who had at least 1 positive VRE test (n=11:
 - 88% (n=7 of 8) who cleared recurrent CDI after RBX2660 administration were VRE-negative at the last test.
 - 66% (n=2 of 3) who failed to clear recurrent CDI after RBX2660 administration were VRE-negative at the last test.

Limitations

- Small secondary analysis conducted in the context of a Phase 2 open-label assessment of RBX2660 for recurrent CDI.
- Patient compliance with stool submission post-RBX2660 administration varied over the course of follow-up. A total of 30/34 (88%) of patients treated with RBX2660 submitted at least 1 stool for VRE testing.

Conclusions:

- This secondary analysis demonstrates the possibility of converting a high percentage of VRE positive patients to negative with a next-generation FMT drug.
- Although the association between clearance of recurrent CDI and VRE clearance was not statistically significant, the percentage of patients successfully treated for CDI who also cleared their VRE was 22% higher than among those not successfully treated for CDI.
- Additional study in a larger prospective trial is need to confirm the results and to further assess the relationship between VRE and CDI clearance.

References:

1. Tavadze M, Rybicki L, Mossad S, et al. Risk factors for vancomycin-resistant enterococcus bactermia and its influence on survival after allogenic hematopoietic cell transplantation. *Bone Marrow Transplantation.* 2014;49:1310-16.
2. Fujitani S, George WL, Morgan MA, Nichols S, Murthy AR. Implications for vancomycin-resistant *Enterococcus* colonization associated with Clostridium difficile infections. *Am J Infect Control.* 2011;39:188-93.
3. Jung E, Byun S, Lee Ho, et al. Vancomycin-resistant *Enterococcus* colonization in the intensive care unit: Clinical outcomes and attributable costs of hospitalization. *Am J Infect Control.* 2014;42:1062-6.
4. Tosh PK, McDonald LC. Infection control in the multidrug-resistant era: *Tending the human microbiome.* CID. 2012;54:707-13.

Corresponding Author

Erik Dubberke, MD, MSPH
Division of Infectious Disease
Washington University School of Medicine
660 S Euclid Ave, Box 8051
St Louis, MO 63110
Tel: 314-454-8293
E-mail: edubberk@DOM.wustl.edu