Does the Donor Matter? Donor vs. Patient Effects in the Outcome of Next-Generation Fecal Transplant for Recurrent *Clostridium difficile* Infection

Courtney Jones, BS[a], Robert Hardi, MD[b], Bill Shannon, PhD[c]

[a]Rebiotix Inc., Roseville, MN; [b]Capital Digestive Care, [c]BioRankings LLC, St. Louis, MO

Corresponding Author

Courtney Jones
2160 Paton Ave.
Roseville, MN 551113
Tel: 651-705-8770
E-mail: cjones@rebiotix.com

Background

- Fecal microbiota transplantation (FMT) is becoming an increasingly accepted treatment for recurrent *Clostridium difficile* infection (CDI).
- Overall resolution rates in the range of 90% have been reported in the literature[1-3] though multiple doses may be necessary to achieve this cure rate.[4-5]
- There are still many unknowns about the therapy including questions about donor vs. patient factors in therapy success.
- Conventional procedure has been to use a second donor in case of treatment failure.
- The relative importance of the donor was assessed in the context of a Phase 2 study of a next-generation FMT drug.

Methods

- A total of 34 patients with recurrent CDI enrolled at 11 sites in the U.S. received 1 or 2 doses of RBX2660 (microbiota suspension) via enema between August 2013 and January 2014 as part of the PUNCH CD study.
- Per protocol, a second dose was permitted if symptoms reoccurred < 8 weeks after the first dose.
- Patients who required 2 doses could receive RBX2660 manufactured from the same or different donors. The same pair of donors could also be used in a different order.
- Efficacy was defined as the absence of CDI at 8 weeks after the last treatment.
- A mathematical model was used to determine whether the specific donor effected the results.

RBX2660 (microbiota suspension)

- Designed to mimic FMT
- Raw material human stool
- Formulated to have a long shelf-life
- Supplied in 50g/150 mL dose; ready-to-use enema format
- Contains a minimum guaranteed quantity of live microbes
- Manufactured using standardized, quality controlled processes

Sourcing and Traceability

- Four donors were used to prepare the RBX2660 used in the study.
- Donations were made on site at Rebiotix Inc., Roseville, MN.
- Donations were not pooled.
- The same batch was used in 1-4 patients.
- Donors were randomized to patients for both the first and second doses.
- The product was manufactured in donor-specific batches that could be tracked to individual patients and outcomes.

Mathematical Model

- A mixed model was used to predict treatment success taking into account donors and dose number.
- Repeated measures data was included to account for patients who received 2 doses of RBX2660. In this analysis, a patient was classified as either a success or failure for both doses.
- Donor 1 was used as the reference donor for comparative purposes as no evidence was seen that the other donors had a different success rate.
- P < .05 indicates statistical significance; all analyses were done using the R statistical package.

Results

- A total of 34 patients (mean age 68.8 years, 67.6% female) received at least 1 dose of RBX2660.
- Nineteen patients received 1 dose and 15 patients received 2 doses.
- Success was not impacted by the donor or dose order.

LOGISTIC REGRESSION MODELING FOR ALL PATIENTS AND ALL DOSES

	Estimate	Std. Error	z Value	P r(>\|z\|)
Intercept	1.554	0.711	2.186	0.029
As factor – Donor 3	-0.115	0.934	-0.123	0.902
As factor – Donor 4	1.060	1.236	0.858	0.391
*As factor – Donor 5	16.221	2267.846	0.007	0.994
As factor – Dose 2	-0.559	0.872	-0.641	0.522

*Donor 5 contributed a comparatively small number of doses and was not significant compared with donor 1. There was a large standard deviation resulting from the small sample size.

- All P values in the logistical model were > .05, indicating no donor effect.

LOGISTIC REGRESSION MODELING FOR PATIENTS RECEIVING 2 DOSES OF RBX2660

	Estimate	Std. Error	z Value	P r(>\|z\|)
Intercept	1.174	0.730	1.609	0.108
As factor – Donor 3	-0.564	1.226	-0.460	0.645
As factor – Donor 4	1.723	1.271	0.569	0.569
*As factor – Donor 5	15.292	2399.545	0.006	0.995
As factor – Dose 2	0.101	1.061	0.095	0.924

*Donor 5 contributed a comparatively small number of doses and was not significant compared with donor 1. There was a large standard deviation resulting from the small sample size.

- All P values in the logistical model were > .05, indicating no donor effect.

Case Study: Same Donor; Same Batch; Different Results

Patient: 89-year-old white female with history of recurrent diarrhea.

Medical History:

- CDI diagnosed during an emergency room visit, positive for toxin A.
- Ciprofloxacin hydrochloride 500 mg, not tolerated; completed two 10-day courses of metronidazole 500 mg tid; two 14-day courses of vancomycin, 125 qid; and 1 course of fidaxomicin, 200 mg bid without resolution of symptoms.
- FMT was recommended; the patient was enrolled in the PUNCH CD study.

RBX2660 Administration:

- Sept. 16, 2013: first dose; well-tolerated; stool normalized.
- Sept. 30, 2013: CDI symptoms return.
- Oct. 3, 2013: second dose without antibiotic pre-treatment; well-tolerated.

Follow-up:

- No CDI symptoms through 6-month follow-up.

16S rRNA Analysis

- Patient's stool was characterized using 16S rRNA analysis at 7 days after the first dose of RBX2660 and at 7, 30, and 60 days after the second dose and compared with donor's fecal microbiota.
- The extraction was completed by Research and Testing Laboratory, Lubbock, TX; analysis was conducted by the Bushman Laboratory, University of Pennsylvania, Philadelphia, PA.

Operational Taxonomic Unit Heat Map Analysis

- A heat map of the patient's samples indicate the distribution of different families of bacteria over time (Figure 1).
- The color-coding represents the level of gene expression across a series of donor and patient stools ranging from "not found" (white) to 100% (red).
- The patient experienced a durable CDI cure after receiving a second dose of RBX2660 from exactly the same batch of product that failed the first time.
- The maps shows that the patient took on similar taxonomic characteristics as the donor. However, a copy of the donor's microbiome was never established in the patient over the course of the study.

Figure 1. Operational Taxonomic Unit Heat Map

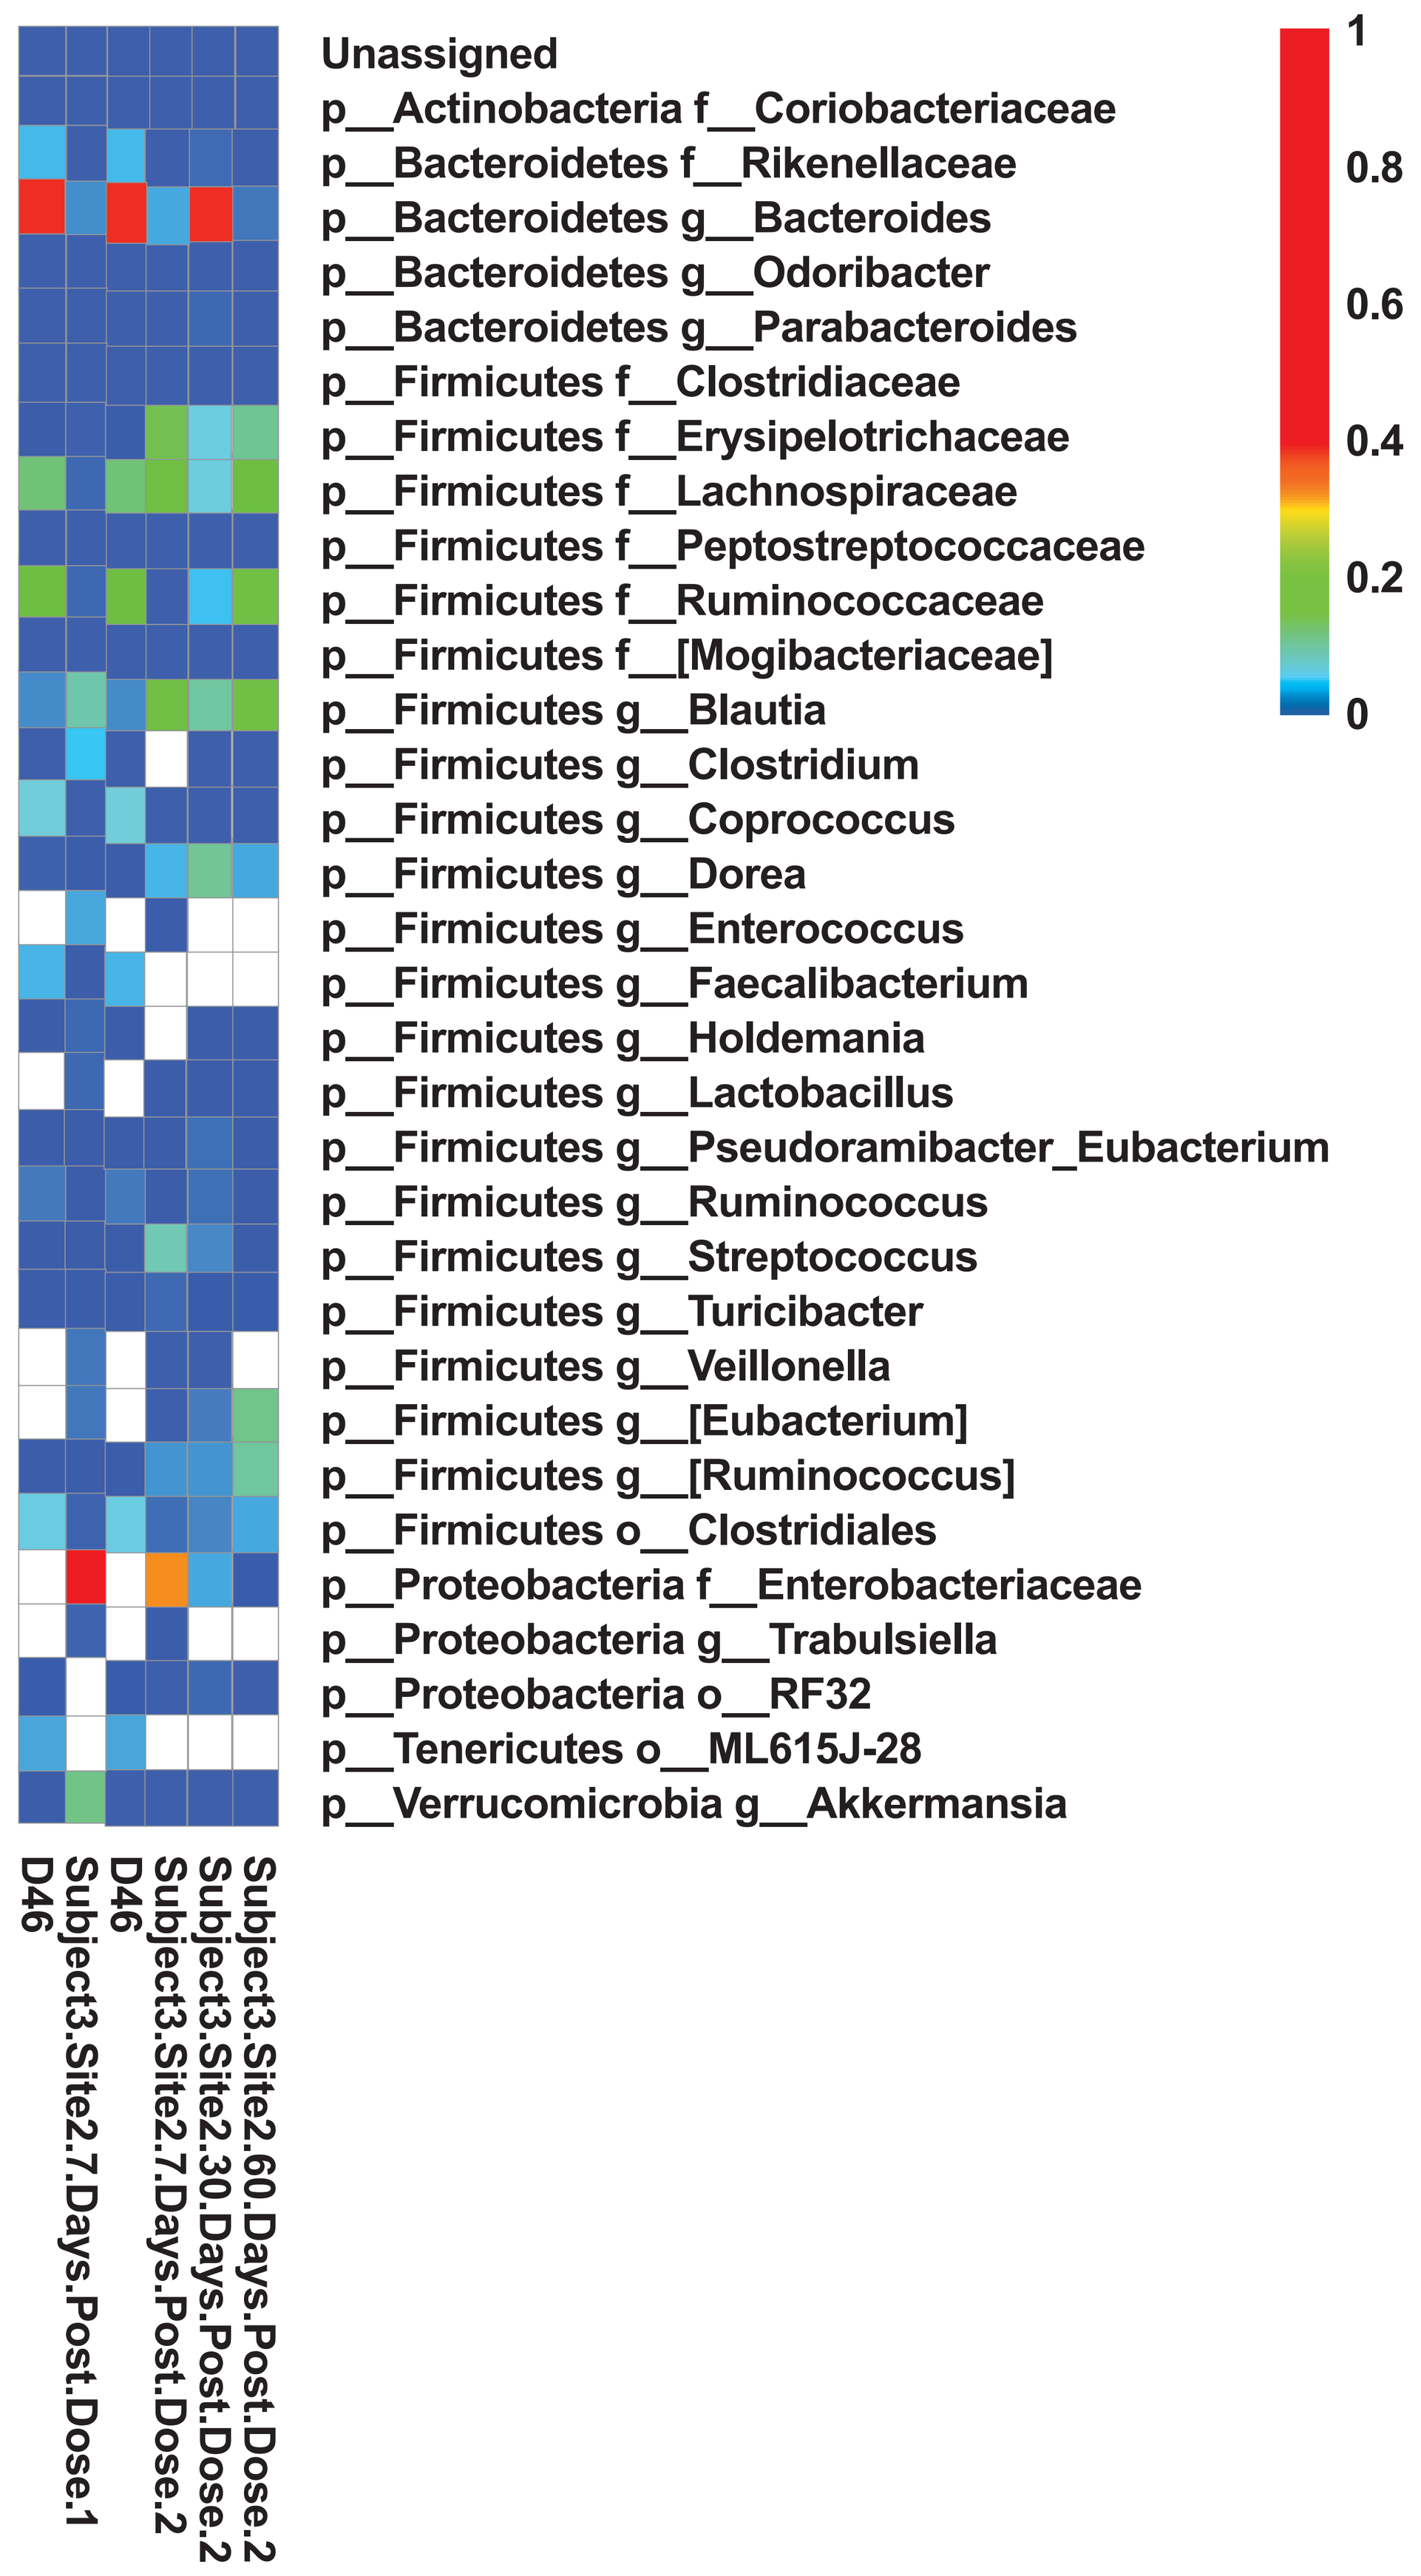

Unassigned
p__Actinobacteria f__Coriobacteriaceae
p__Bacteroidetes f__Rikenellaceae
p__Bacteroidetes g__Bacteroides
p__Bacteroidetes g__Odoribacter
p__Bacteroidetes g__Parabacteroides
p__Firmicutes f__Clostridiaceae
p__Firmicutes f__Erysipelotrichaceae
p__Firmicutes f__Lachnospiraceae
p__Firmicutes f__Peptostreptococcaceae
p__Firmicutes f__Ruminococcaceae
p__Firmicutes f__[Mogibacteriaceae]
p__Firmicutes g__Blautia
p__Firmicutes g__Clostridium
p__Firmicutes g__Coprococcus
p__Firmicutes g__Dorea
p__Firmicutes g__Enterococcus
p__Firmicutes g__Faecalibacterium
p__Firmicutes g__Holdemania
p__Firmicutes g__Lactobacillus
p__Firmicutes g__Pseudoramibacter_Eubacterium
p__Firmicutes g__Ruminococcus
p__Firmicutes g__Streptococcus
p__Firmicutes g__Turicibacter
p__Firmicutes g__Veillonella
p__Firmicutes g__[Eubacterium]
p__Firmicutes g__[Ruminococcus]
p__Firmicutes o__Clostridiales
p__Proteobacteria f__Enterobacteriaceae
p__Proteobacteria g__Trabulsiella
p__Proteobacteria o__RF32
p__Tenericutes o__ML615J-28
p__Verrucomicrobia g__Akkermansia
D46
Subject3.Site2.7.Days.Post.Dose.1
D46
Subject3.Site2.7.Days.Post.Dose.2
Subject3.Site2.30.Days.Post.Dose.2
Subject3.Site2.60.Days.Post.Dose.2
1
0.8
0.6
0.4
0.2
0

Conclusions

- Based on an analysis of small numbers, it appears that the specific donor does not affect the outcomes achieved with administration of RBX2260 for recurrent CDI.
- The results suggest that outcomes are patient-specific and it is not necessary to switch donors in order to achieve a cure if the first dose fails.
- Additional research with a larger patient cohort along with in-depth comparative analysis of donor and patient microbiota is needed to provide more details on patient-specific factors impacting success or failure with this therapy.

REFERENCES

1. Gough E, Shaikh H, Manges AR. Systematic review of intestinal microbiota transplantation (fecal bacteriotherapy) for recurrent *Clostridium difficile* infection. *Clinical Infectious Diseases 2011*;53(10):994–1002.
2. Kassam Z, Lee CH, Yuan Y, et al. Fecal microbiota transplantation for *Clostridium difficile* infection: systematic review and meta-analysis. *Am J Gastroenterol*. 2013;108:500–508.
3. Keller JJ, Kuijper EJ. Treatment of recurrent and severe *Clostridium difficile* infection. *Annu Rev Med*. 2015;66:373-86.
4. van Nood E, Vrieze A, Nieuwdorp M, et al. Duodenal infusion of donor feces for recurrent *Clostridium difficile*. *N Engl JMed*. 2013;368:407–415.
5. Lee CH, Belanger JE, Kassam Z et al. The outcome and long-term follow-up of 94 patients with recurrent and refractory *Clostridium difficile* infection using single to multiple fecal microbiota transplantation via retention enema. *Eur J Clin Microbiol Infect Dis.* 2014;33(8):1425-8.