Efficacy and Safety of RBX2660 for the Prevention of Recurrent *Clostridium difficile* Infection: Results of the PUNCH CD 2 Trial

Erik R. Dubberke, MD, MSPH,[a] Christine Lee, MD,[b] Robert Orenstein, DO,[c] and Sahil Khanna, MBBS, MS[d] Gail Hecht, MD,[e] Joseph Fraiz, MD[f]

[a]Department of Medicine, Washington University School of Medicine, St. Louis, MO; [b]Hamilton Regional Laboratory Medicine Program, Department of Pathology and Molecular Medicine, McMaster University, Hamilton, ON, Canada; [c]Division of Infectious Diseases, Mayo Clinic in Arizona, Phoenix, AZ; [d]Division of Gastroenterology and Hepatology Mayo Clinic, Rochester, MN; [e]Division of Gastroenterology and Nutrition, Loyola University Medical Center, Chicago, IL; [f]Infectious Disease of Indiana, Carmel, IN

Corresponding Author

Erik Dubberke, MD, MSPH
660 S Euclid, Box 8051,
St. Louis, MO 63110
E-mail: edubberk@dom.wustl.edu

Background

- Experience indicates microbiota-based therapies are highly efficacious at preventing *Clostridium difficile* (CDI) recurrences.
- There is a need for high-quality data on efficacy and safety.
- The PUNCH CD 2 trial was a randomized, placebo-controlled, double-blinded study to assess the safety and efficacy of RBX2660, a microbiota-based drug targeted at recurrent CDI.

Methods

- Patients with recurrent CDI were randomized to: 2 doses of RBX2660 (Group A); 2 doses of placebo (Group B); or 1 dose of RBX2660 and 1 dose of placebo (Group C) via enema with doses 7 days apart.
- Control of CDI symptoms with antibiotics was required prior to enrollment. Antibiotics were discontinued 24-48 hours prior to the first enema.
- Failures could receive up to 2 doses of open-label active treatment, 7 days apart, Figure 1.
- Safety was assessed via a patient diary: in clinic at 1, 4, and 8 weeks and via telephone at 2, 3 and 5-7 weeks and at 3, 6, 12 and 24 months. Durability was assessed concurrently and is on-going until 24 months.

Figure 1. PUNCH CD 2 Trial Design

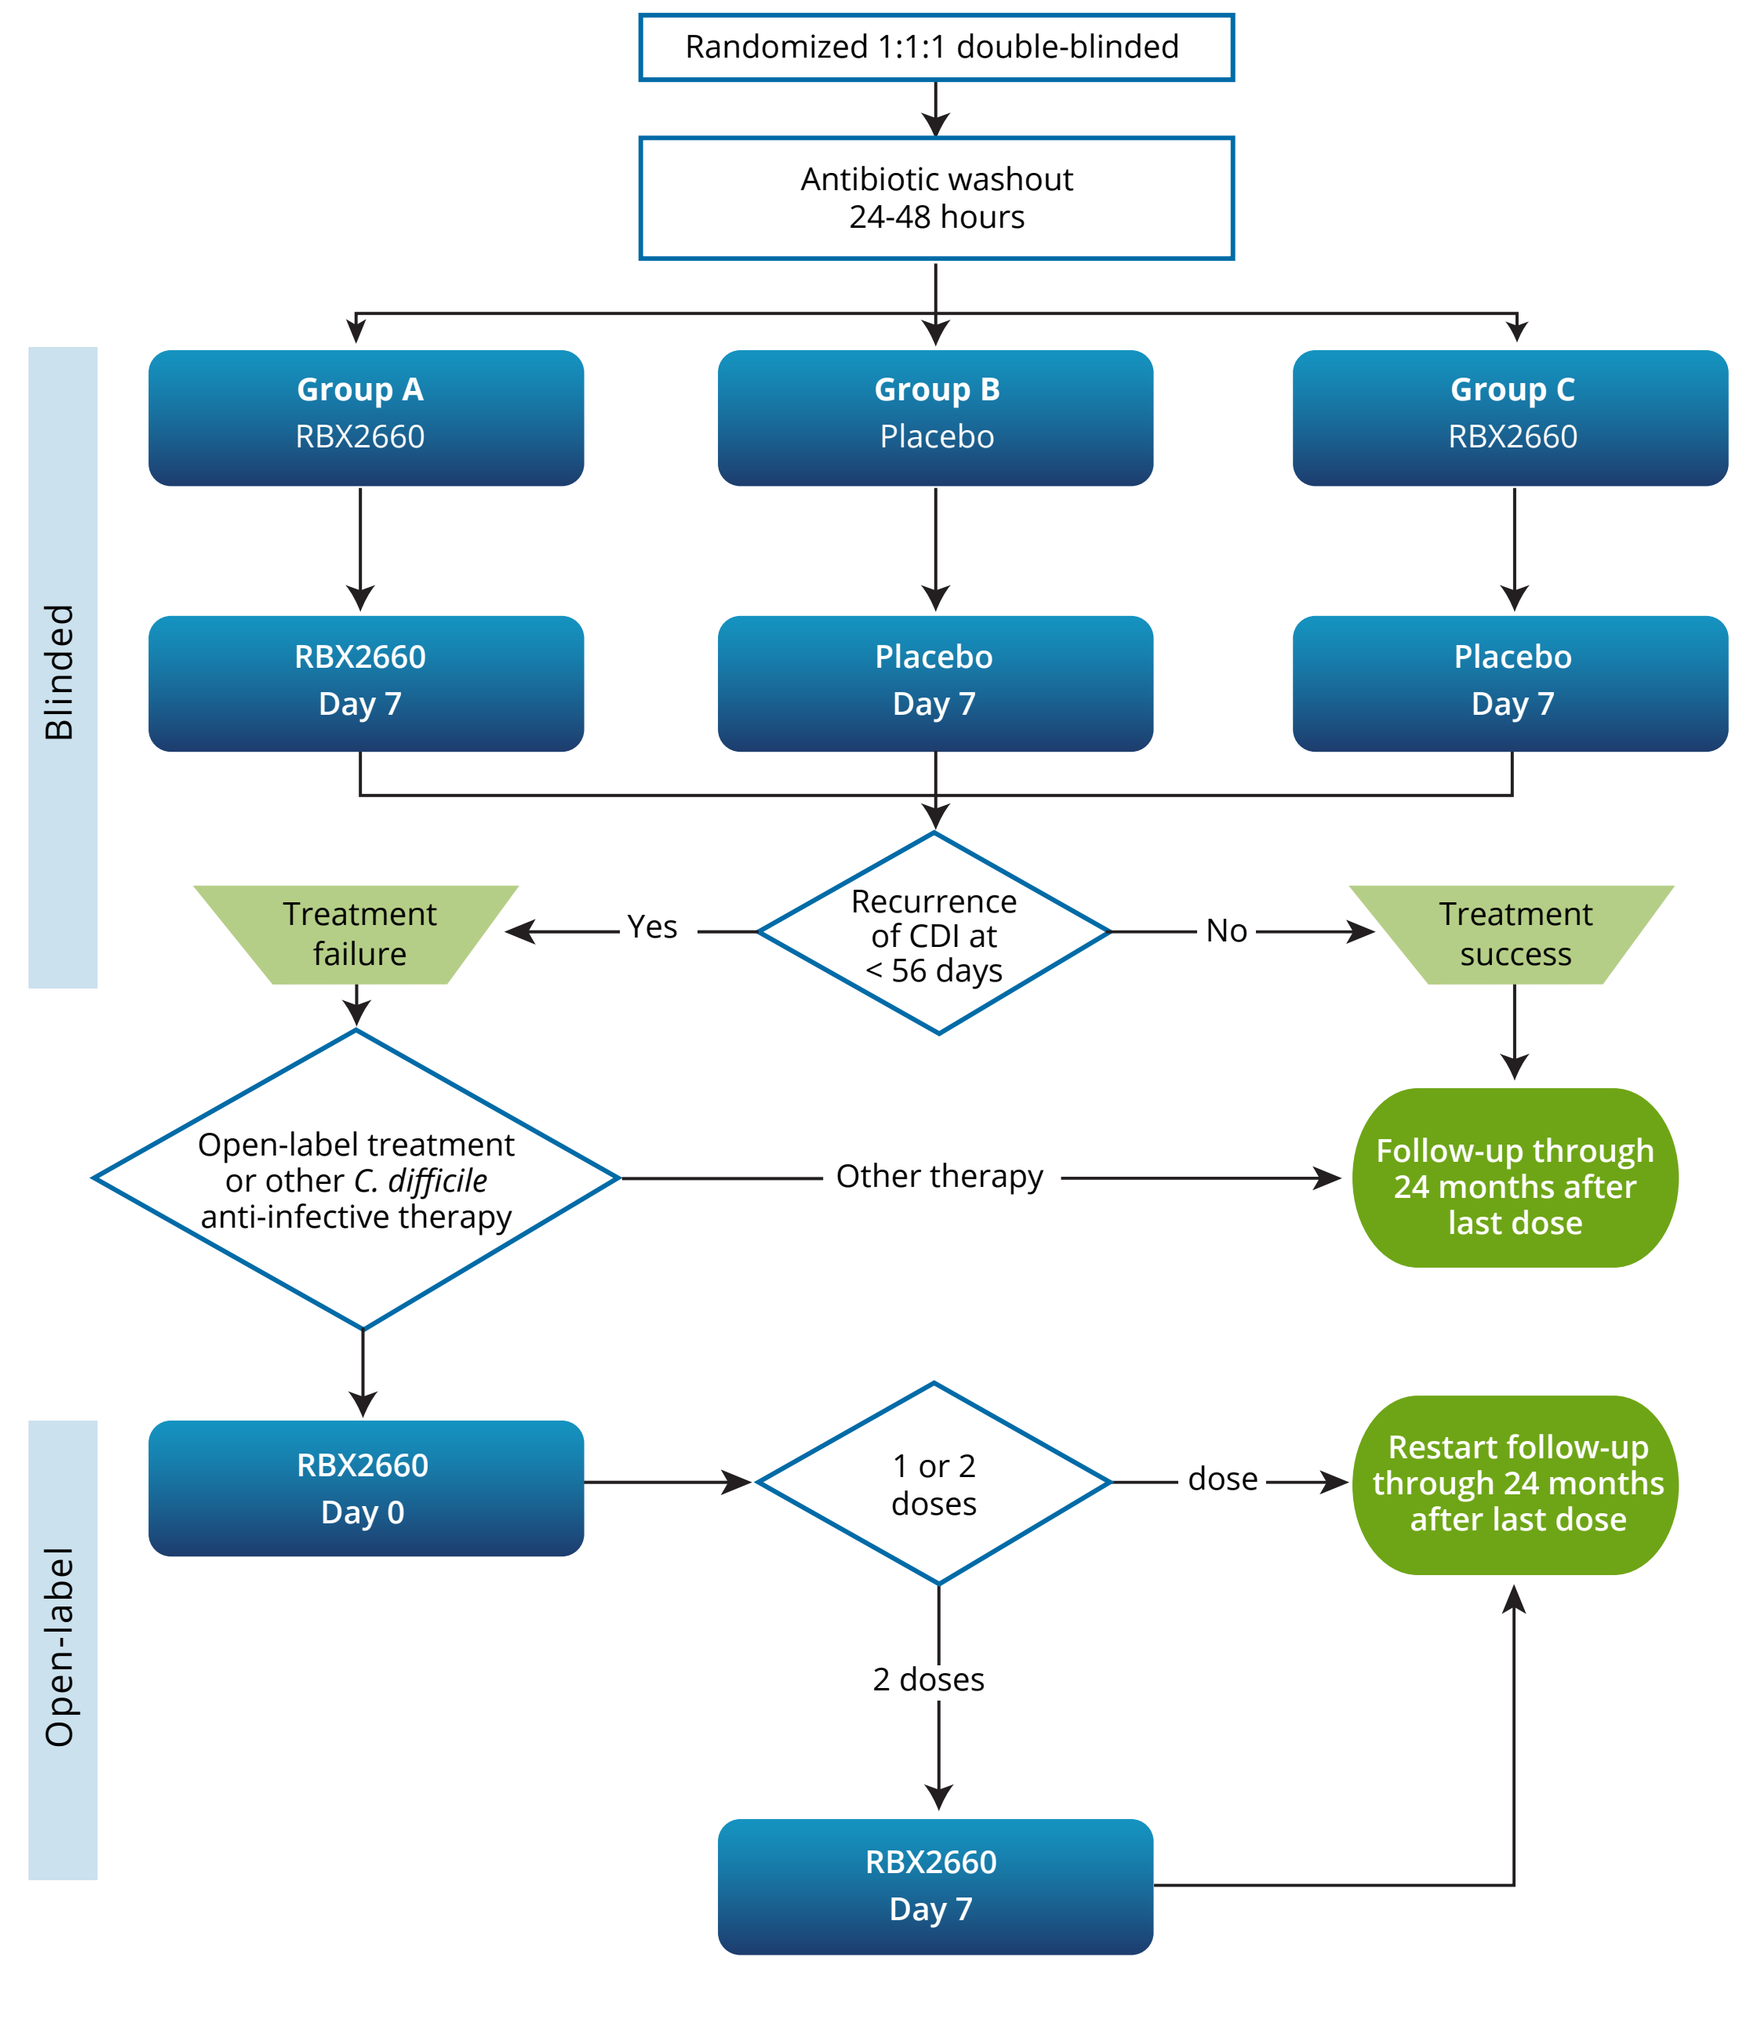
Randomized 1:1:1 double-blinded
Antibiotic washout 24-48 hours
Group A RBX2660
Group B Placebo
Group C RBX2660
RBX2660 Day 7
Placebo Day 7
Placebo Day 7
Blinded
Recurrence of CDI at < 56 days
Yes
No
Treatment failure
Treatment success
Open-label treatment or other C. difficile anti-infective therapy
Other therapy
Follow-up through 24 months after last dose
Open-label
RBX2660 Day 0
1 or 2 doses
dose
Restart follow-up through 24 months after last dose
2 doses
RBX2660 Day 7

Objectives

- Assess dosing strategy for RBX2660
 - Efficacy of 1 vs. 2 doses; timing of doses
- Long-term safety of RBX2660

Major Inclusion Criteria

- Age ≥ 18 years old
- A positive stool test for *Clostridium difficile* within 60 days prior to enrollment
- At least two recurrences of CDI after a primary episode
 Or
- At least two episodes of severe CDI resulting in hospitalization

Major Exclusion Criteria

- History of inflammatory bowel disease (ulcerative colitis, Crohn's disease or microscopic colitis); irritable bowel syndrome; chronic diarrhea; celiac disease
- Colostomy
- Evidence of active colitis
- Known exposure to antibiotics within 6 months after study enrollment
- Compromised immune system
- White blood cell count <1000 cells/μL

RBX2660

- Microbiota-based drug manufactured from live human-derived microbes using standardized processes and controls.
- Each unit of RBX2660 is identified by a unique batch number and is traceable to a specific donor and recipient.

Definition of Success

- The absence of CDI associated diarrhea -without the need for retreatment with a *Clostridium difficile* anti-infective therapy or fecal transplant through 8 weeks after administration of the second dose of the assigned treatment in the blinded phase.

Definition of Failure *(All 4 Criteria Required)*

- Presence of diarrhea, with or without other CDI symptoms, at <8 weeks after last treatment
- A positive stool test for Clostridium difficile
- Need for retreatment for CDI
- No other cause for diarrhea identified

Results

- A total of 127 patients at 21 sites in the U.S. and Canada were enrolled between December 10, 2014 through November 13, 2015 were included in the intent-to-treat analysis:
 - Group A (2 doses of RBX2660): n=41
 - Group B (2 doses of placebo): n=44
 - Group C (1 dose of RBX2660 and 1 dose of placebo: n=42
- The median age was: 63.0, range: 18-92 years; sex: 62% female; median prior CDI episodes (4 range: 2-14), Table 1.

Efficacy

BLINDED:

- Group C (1 dose) was superior to Group B (placebo): 66.7%, 28/42 vs. 45.5%, 20/44, P=0.048.
- There was no difference between Group A: 2 doses (61%, 25/41) vs. Group C: 1 dose (66.7%), P=0.589.
- The combined success in Groups A and C (1 or 2 doses) was superior to Group B: placebo, 63.9% vs. 45.5%, P=0.046.

OPEN-LABEL:

- Of patients in groups A and C who went on to receive open-label treatment, 70% succeeded (21/30).
- All 24 patients who failed placebo (experienced recurrence) went on to receive open-label treatment with RBX2660. Of these patients, 87.5% (21/24) were successful when treated with RBX2660.

OVERALL:

- For all patients who were randomized to receive at least 1 active treatment (Groups A and C), the overall success (blinded and open-label) was 89.2% (n=74/83) compared with a 45.5% (20/44) placebo response, P < 0.0001, Figure 2.

Safety

- There were 580 adverse events (AEs) reported in 94 patients and 45 serious AEs in 26 patients with a mean follow-up of 8.3 months, Table 2.
- AEs were primarily gastrointestinal (n=242, 41.7%).
- There were no unanticipated AEs and there was no significant difference in proportion of AEs or serious AEs among the treatment groups.

Durability

- Of the patients who received at least 1 active treatment and were successful, the long-term CDI-free rate (median follow-up: 8.3 months; range: 1.6 to 14.9 months) was 95.8%.
- Of these patients, 4.2% developed a new episode of CDI confirmed by a positive test > 8 weeks after that last RBX2660 treatment.
- One recurrence occurred after antibiotics for a dog bite and one during a hospital stay for small bowel obstruction. Reasons were unknown for 2 cases.

Conclusions

- RBX2660 administered via enema is a safe and effective and effective treatment for recurrent CDI.
- Overall efficacy of RBX2660 was 89.2% in a randomized, double-blinded, placebo-controlled study in patients with recurrent CDI.
- AEs were primarily gastrointestinal; there were no unanticipated AEs.
- RBX2660 provided a durable cure.
- The results were consistent with those achieved in the open-label PUNCH CD trial.[1]

Table 1. Baseline Characteristics in the Intent-to-Treat Population

Characteristic	2 Doses of RBX2660 (N=41)	2 Doses of Place-bo (N=44)	1 Dose of RBX2660 and 1 Dose of Placebo (N=42)
Median age (IQR) - years	66.0 (28.0)	62.0 (24.5)	63.0 (33.0)
Female sex- no. (%)	25 (61)	30 (68)	24 (57)
Race-White – no. (%)	39 (95)	42 (96)	40 (95)
Antibiotic used at screening – no. (%)			
Vancomycin	38 (93)	39 (89)	36 (86)
Fidaxomicin	1 (2)	2 (5)	2 (5)
Other	2 (5)	3 (6)	4 (10)
Median number of CDI episodes (range) – no., interquartile range	4.0 (2)	3.0 (1)	4.0 (1)
Mean duration of CDI episode, days	18.8 ± 14	19.5 (17)	16.4 (11)
Healthcare-acquired CDI infection – no. (%)	10 (5.7)	10 (6.0)	13 (7.8)
Mean duration of hospitalization, days	14.4 ± 12.4	5.3 ± 2.9	14.2 ± 26.6

CDI=*Clostridium difficile* infection

Figure 2. Overall Success of RBX2660 in Patients Randomized to Receive at Least One Active Treatment

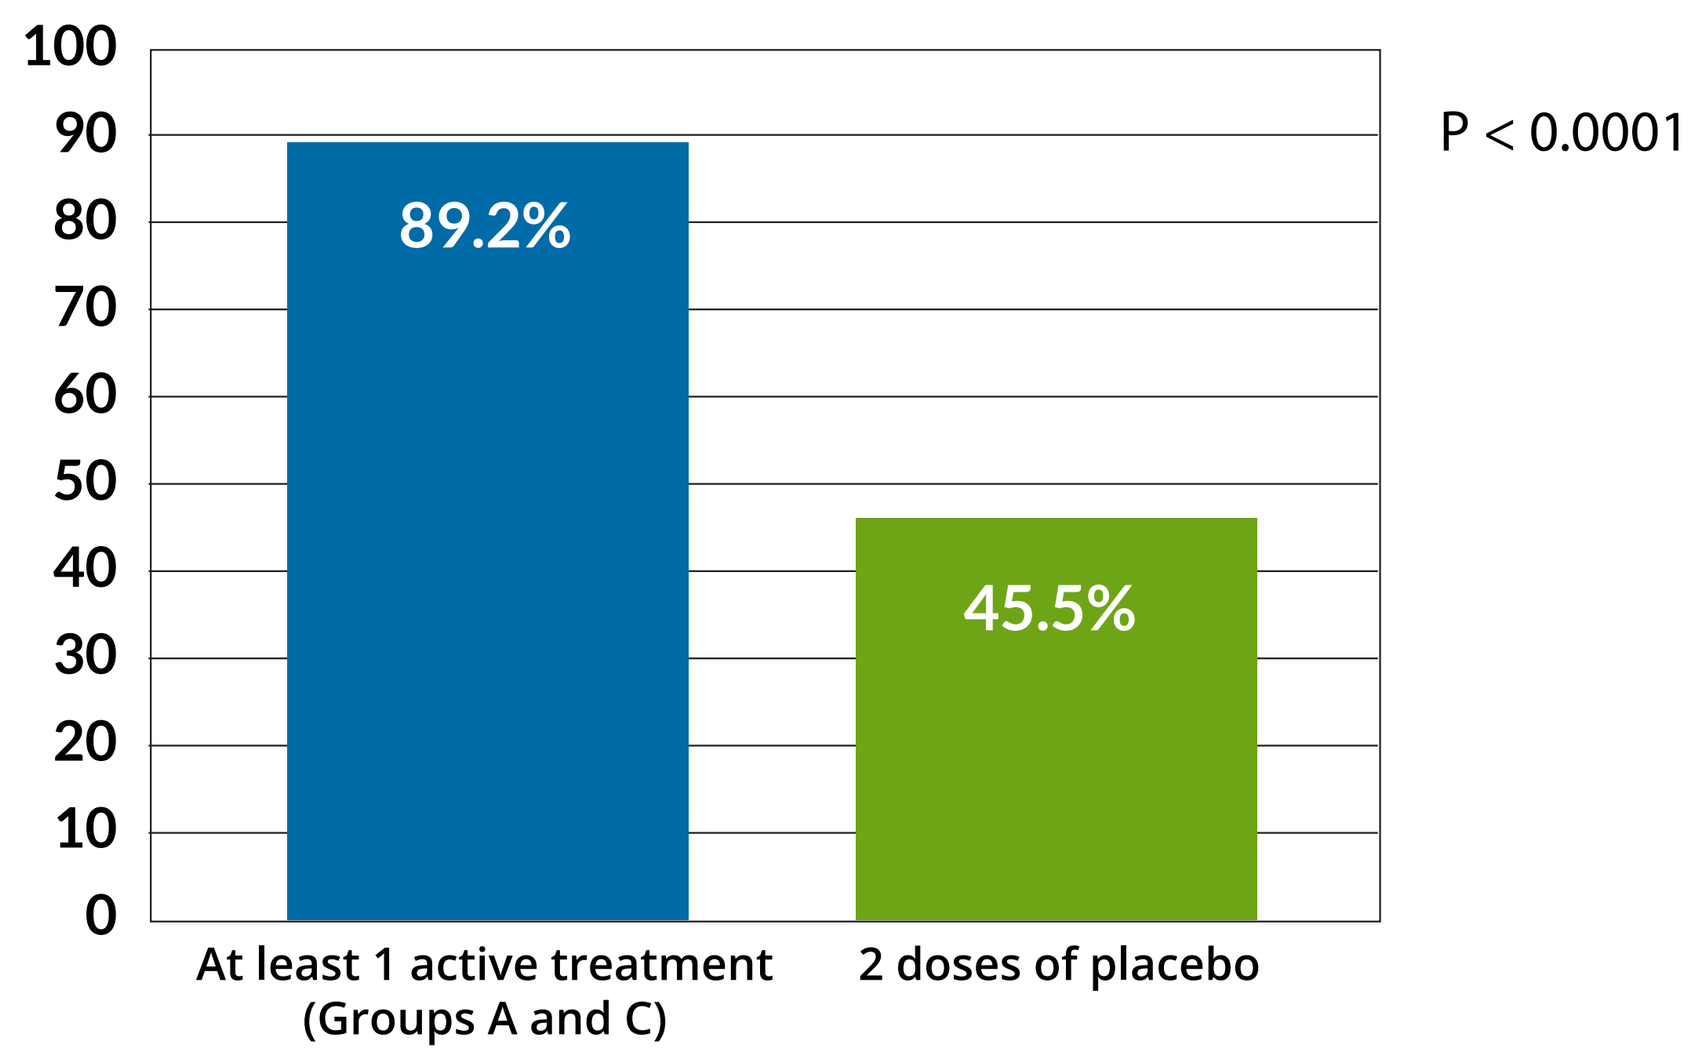
100
90
80
70
60
50
40
30
20
10
0
89.2%
45.5%
P < 0.0001
At least 1 active treatment (Groups A and C)
2 doses of placebo

Table 2. Adverse Events, Mean Follow-up: 8.3 Months

Adverse Events	n (%)
Gastrointestinal disorders	242 (41.7)
General disorders	57 (9.8)
Infections	52 (9)
Unspecified	37 (6.4)
Respiratory disorders	26 (4.5)
Injury, poisoning, procedural complications	23 (4)
Nervous system disorders	21 (3.6)
Metabolism and nutrition disorders	20 (3.5)
Musculoskeletal and connective tissue disorders	14 (2.4)
Cardiac disorders	13 (2.2)
Skin and subcutaneous tissue disorders	13 (2.2)
Renal and urinary disorders	11 (2)
Blood and lymphatic system disorders	10 (2)
Psychiatric disorders	10 (2)
Vascular disorders	8 (1.4)
Investigations	7 (1.2)
Reproductive and breast disorders	4 (0.7)
Eye disorders	3 (0.5)
Hepatobiliary disorders	3 (0.5)
Neoplasms, benign, malignant and unspecified	3 (0.5)
Surgical and medical procedures	1 (0)
Ear and labyrinth disorders	1
Endocrine disorders	1
Total	**580 (100)**

References

1. Orenstein R, Dubberke E, Hardi R, et al. Safety and durability of RBX2660 (microbiota suspension) for recurrent *Clostridium difficile* infection: results of the PUNCH CD study. *Clin Infect Dis.* 2016;62:596-602.

The PUNCH CD 2 trial was sponsored by Rebiotix Inc., Roseville, MN.