Enema Administration of RBX2660 (microbiota suspension) for Recurrent *C. difficile* Infection: Lessons Learned from the PUNCH CD Study

Arnab Ray, MD[a], Robert Hardi, MD[b], Mayur S. Ramesh, MD[c], Mary Kay Sobcinski, RN, MHA[d]

[a]Ochsner Clinic, New Orleans, LA; [b]Capital Digestive Care, Chevy Chase, MD; [c]Henry Ford Health System; [d]Rebiotix Inc., Roseville, MN

Purpose

Fecal transplantation is gaining increasing acceptance as an effective method of treating recurrent *Clostridium difficile* infection (CDI). However, there are many unknowns about the therapy including questions about administration. Administration via colonoscopy, nasogastric or nasoduodenal tube, and enema have been reported in the literature. Definitive superiority has not been established for any one method.

We report on lessons learned about enema administration of RBX2660 (microbiota suspension) for the treatment of recurrent CDI in the context of a 60-day interim analysis of the PUNCH CD phase 2 multicenter safety study. RBX2660, a biologic drug consisting of live human-derived microbes, is a next generation version of fecal transplantation.

Methods

RBX2660 was administered via enema to unprepped patients with recurrent CDI who were treated with standardized oral vancomycin. A second RBX2660 treatment was permitted if CDI reoccurred in <8 weeks. Antibiotics were not required prior to the second treatment. Enema administration was chosen because of simplicity, convenience and safety compared with colonoscopic or nasoenteral administration which are associated with additional risk, especially in the recurrent CDI population (elderly with multiple comorbidities).

Results

A total of 34 patients (mean age 66.8 years; range 26.7 to 89.6 years) received at least one RBX2660 treatment, and 31 were included in the 60-day interim analysis (2 patients dropped out after the first dose and 1 died of respiratory failure). Overall efficacy was 87.1%. The most common position for enema administration was left lateral decubitus (used in 94.1% of patients for the first treatment and in 100% for the second treatment). The knee/chest position was used in the remaining cases. The mean time patients remained in position after RBX2660 administration was 44.5 min. (range 15.0-75 min.) for the first treatment; 47.7 min. (range 15.0-60 min.) for the second treatment. Forceful enema expulsion was reported in 4 patients after the first treatment and in 4 after the second treatment. The mean retention time prior to expulsion was 15.8 min. for the first treatment and 7.8 min. for the second treatment. Patients remained in position after explusion. Six of the 8 patients who expelled the enema were treatment successes. A patient who expelled only a small amount of the enema 20 minutes after treatment and a second patient who expelled a moderate amount 5 minutes after treatment were treatment failures.

Figure 1. Enema Position

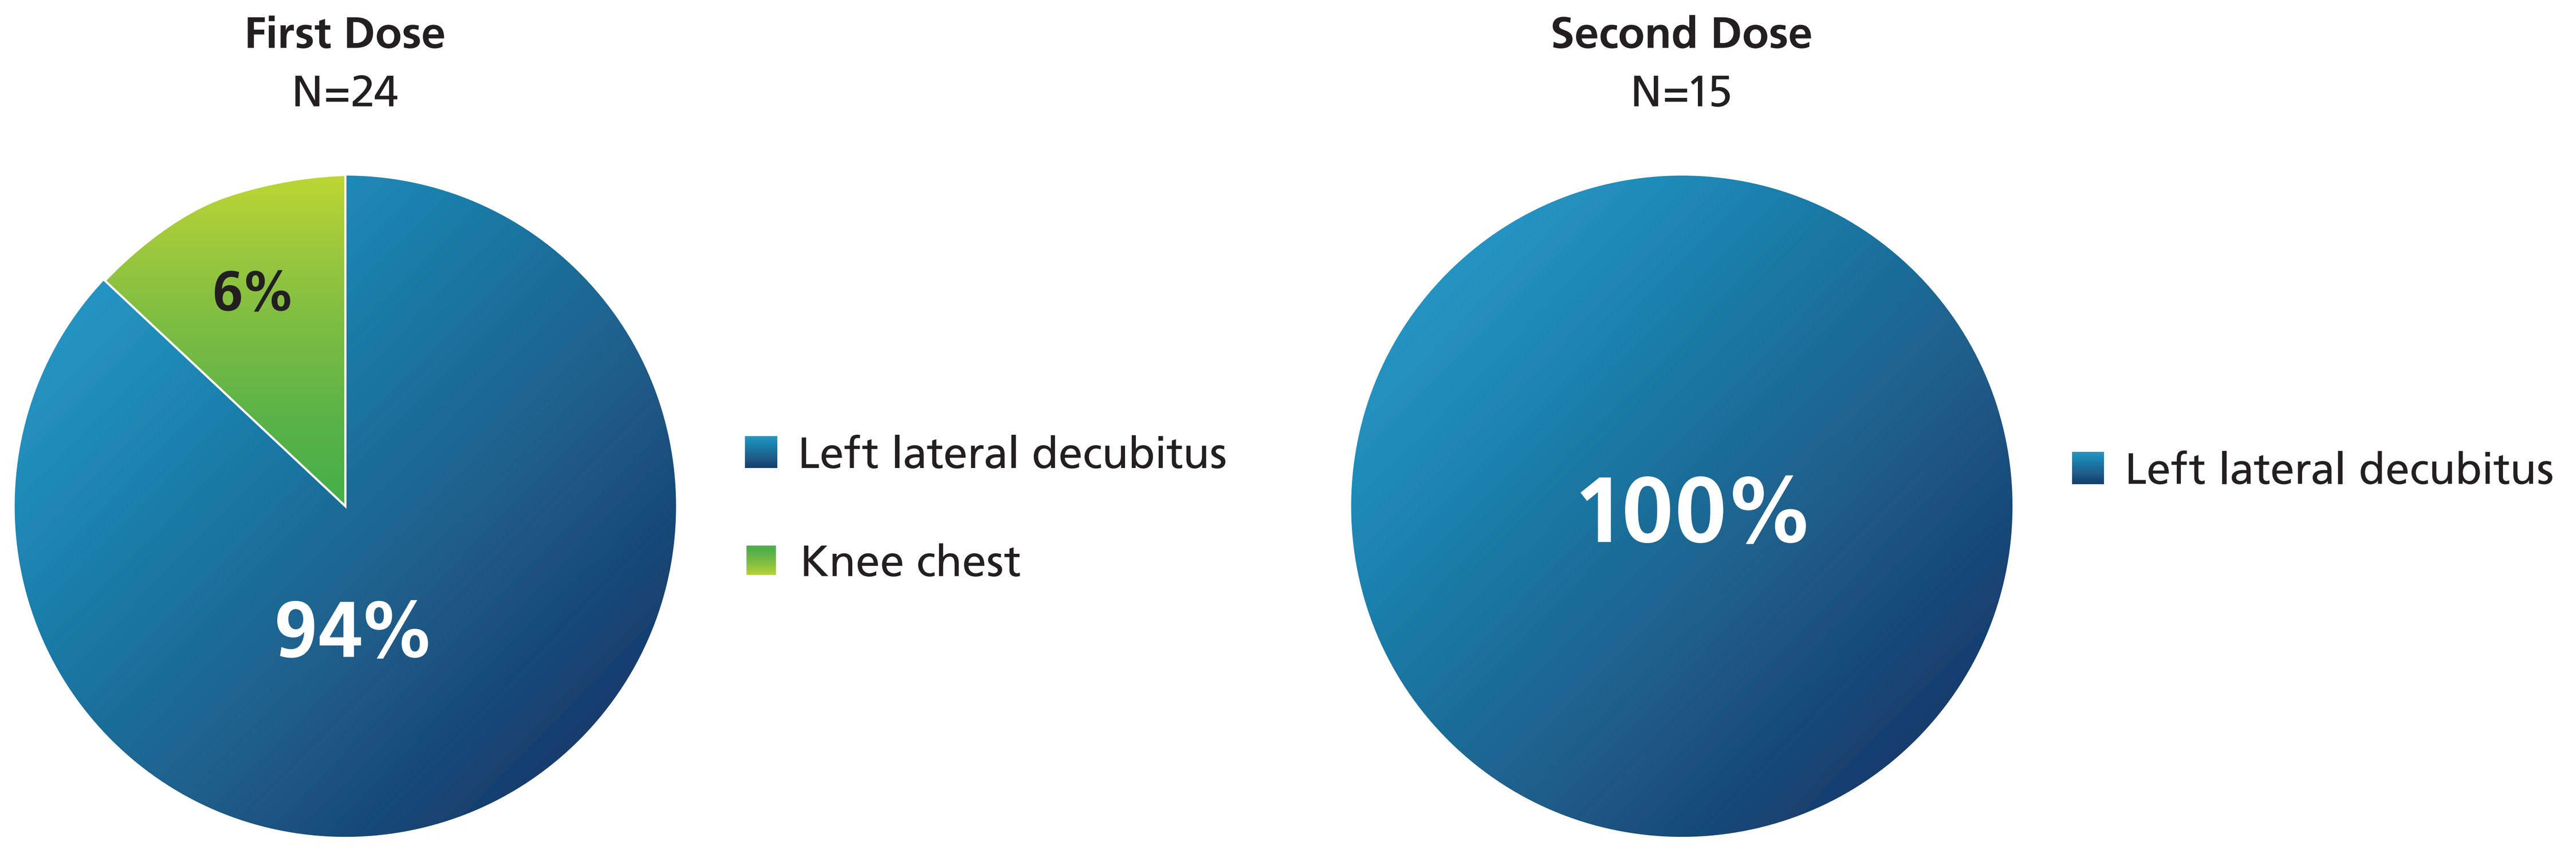
First Dose
N=24
6%
94%
Left lateral decubitus
Knee chest
Second Dose
N=15
100%
Left lateral decubitus

Table 1.
Enema Treatment Parameters

	RBX2660 Dose 1 N=34	RBX2660 Dose 2 N=15
Adherence to antibiotic regimen	92% (n=31)	N/A
Time since last bowel movement (min)		
Mean	382.3	291.1
Range	20 - 1,440	0 - 1,260
Duration in position after enema (min)		
Mean	44.5	47.7
Range	15 - 75	15 - 60
Forceful expulsion of enema	11.8% (n=4)	26.7% (n=4)
Time enema was retained prior to forceful expulsion (min)		
Mean	15.8	7.8
Range	2 - 45	1 - 20
Leakage during observation period	20.6% (7/34)	20% (3/15)
Estimated leakage (%)		
Scant	42.9 (3/7)	100 (3/3)
Small	42.9 (3/7)	0
Moderate	14.3 (1/7)	0
Large	0	0

Conclusion

Enema administration of RBX2660 in the first multicenter study of a biologic drug containing live human-derived microbes was simple and well tolerated with efficacy results consistent with those reported in the literature for fecal transplantation.