#P2222

Impact of RBX2660 on the Intestinal Microbiota of Patients with Recurrent *Clostridium difficile* Infection Enrolled in the Randomized, Placebo-Controlled PUNCH CD 2 Trial

Courtney Jones, BS[a], Bill Shannon, PhD, MBA[b] , Sharina Carter, BS[b]

[a]Rebiotix Inc., Roseville, MN; [b]BioRankings, St. Louis, MO

Background

- Perturbation of the intestinal microbiota, primarily by antibiotics, is a risk factor for *Clostridium difficile* infection (CDI).
- Restoration of a variable intestinal microbiota protects against recurrence of CDI.
- RBX2660 is a microbiota-based drug under study for the prevention of recurrent CDI.
- We evaluated the impact of RBX2660 treatment on the intestinal microbiota of patients enrolled in the PUNCH CD 2 trial, a multicenter, randomized, double-blinded, placebo-controlled phase 2b study.

Methods

- Patients in the 3-arm PUNCH CD 2 trial were randomized 1:1:1 to receive either: 2 doses of RBX2660 (Group A); 2 doses of placebo (Group B); or 1 dose of RBX2660 and 1 dose of placebo (Group C). Success was defined as the absence of *Clostridium difficile*-associated diarrhea at 8 weeks following completion of the last treatment.
- Failure was defined as recurrence of CDI symptoms; a positive stool test; a need for CDI retreatment; and no other cause for CDI symptoms within 8 weeks.
- Longitudinal 16s rRNA analysis using the Illumina MiSeq platform was performed on a total of 120 stool samples from patients in the 3 arms of the study (Group A: n=23; Group B: n=22; Group C: n= 22) at baseline, 7 days and 30 days after the second blinded dose.
- The variable region V4 was targeted to identify the operational taxonomic units (OTUs) in each sample.

Results

- At baseline, the microbiota profiles were similar across all samples.
- OTU analysis demonstrated higher diversity between baseline and follow-up time points for all patients who responded successfully to their assigned treatment.
- The higher diversity at follow-up points was also true for placebo responders (12 patients in Group B who received two doses of placebo and did not experience recurrent CDI symptoms prior to 8-week follow-up).
- Specifically, successful Group A patients had an increase in *Akkermansia* and *Bacteroides* from baseline; successful Group B and C patients had a decreased abundance of *Enterobacter (Figures 1-3)*.
- Of the three groups of patients, those in Group A (two doses of active treatment) showed the greatest increase in bacterial diversity (number of OTUs) and abundance of taxa between baseline and all follow-up points (Figure 4).

Figure 1. Group A (2 Doses of RBX2660): Comparative Microbiota Profiles Showing Baseline vs. Success

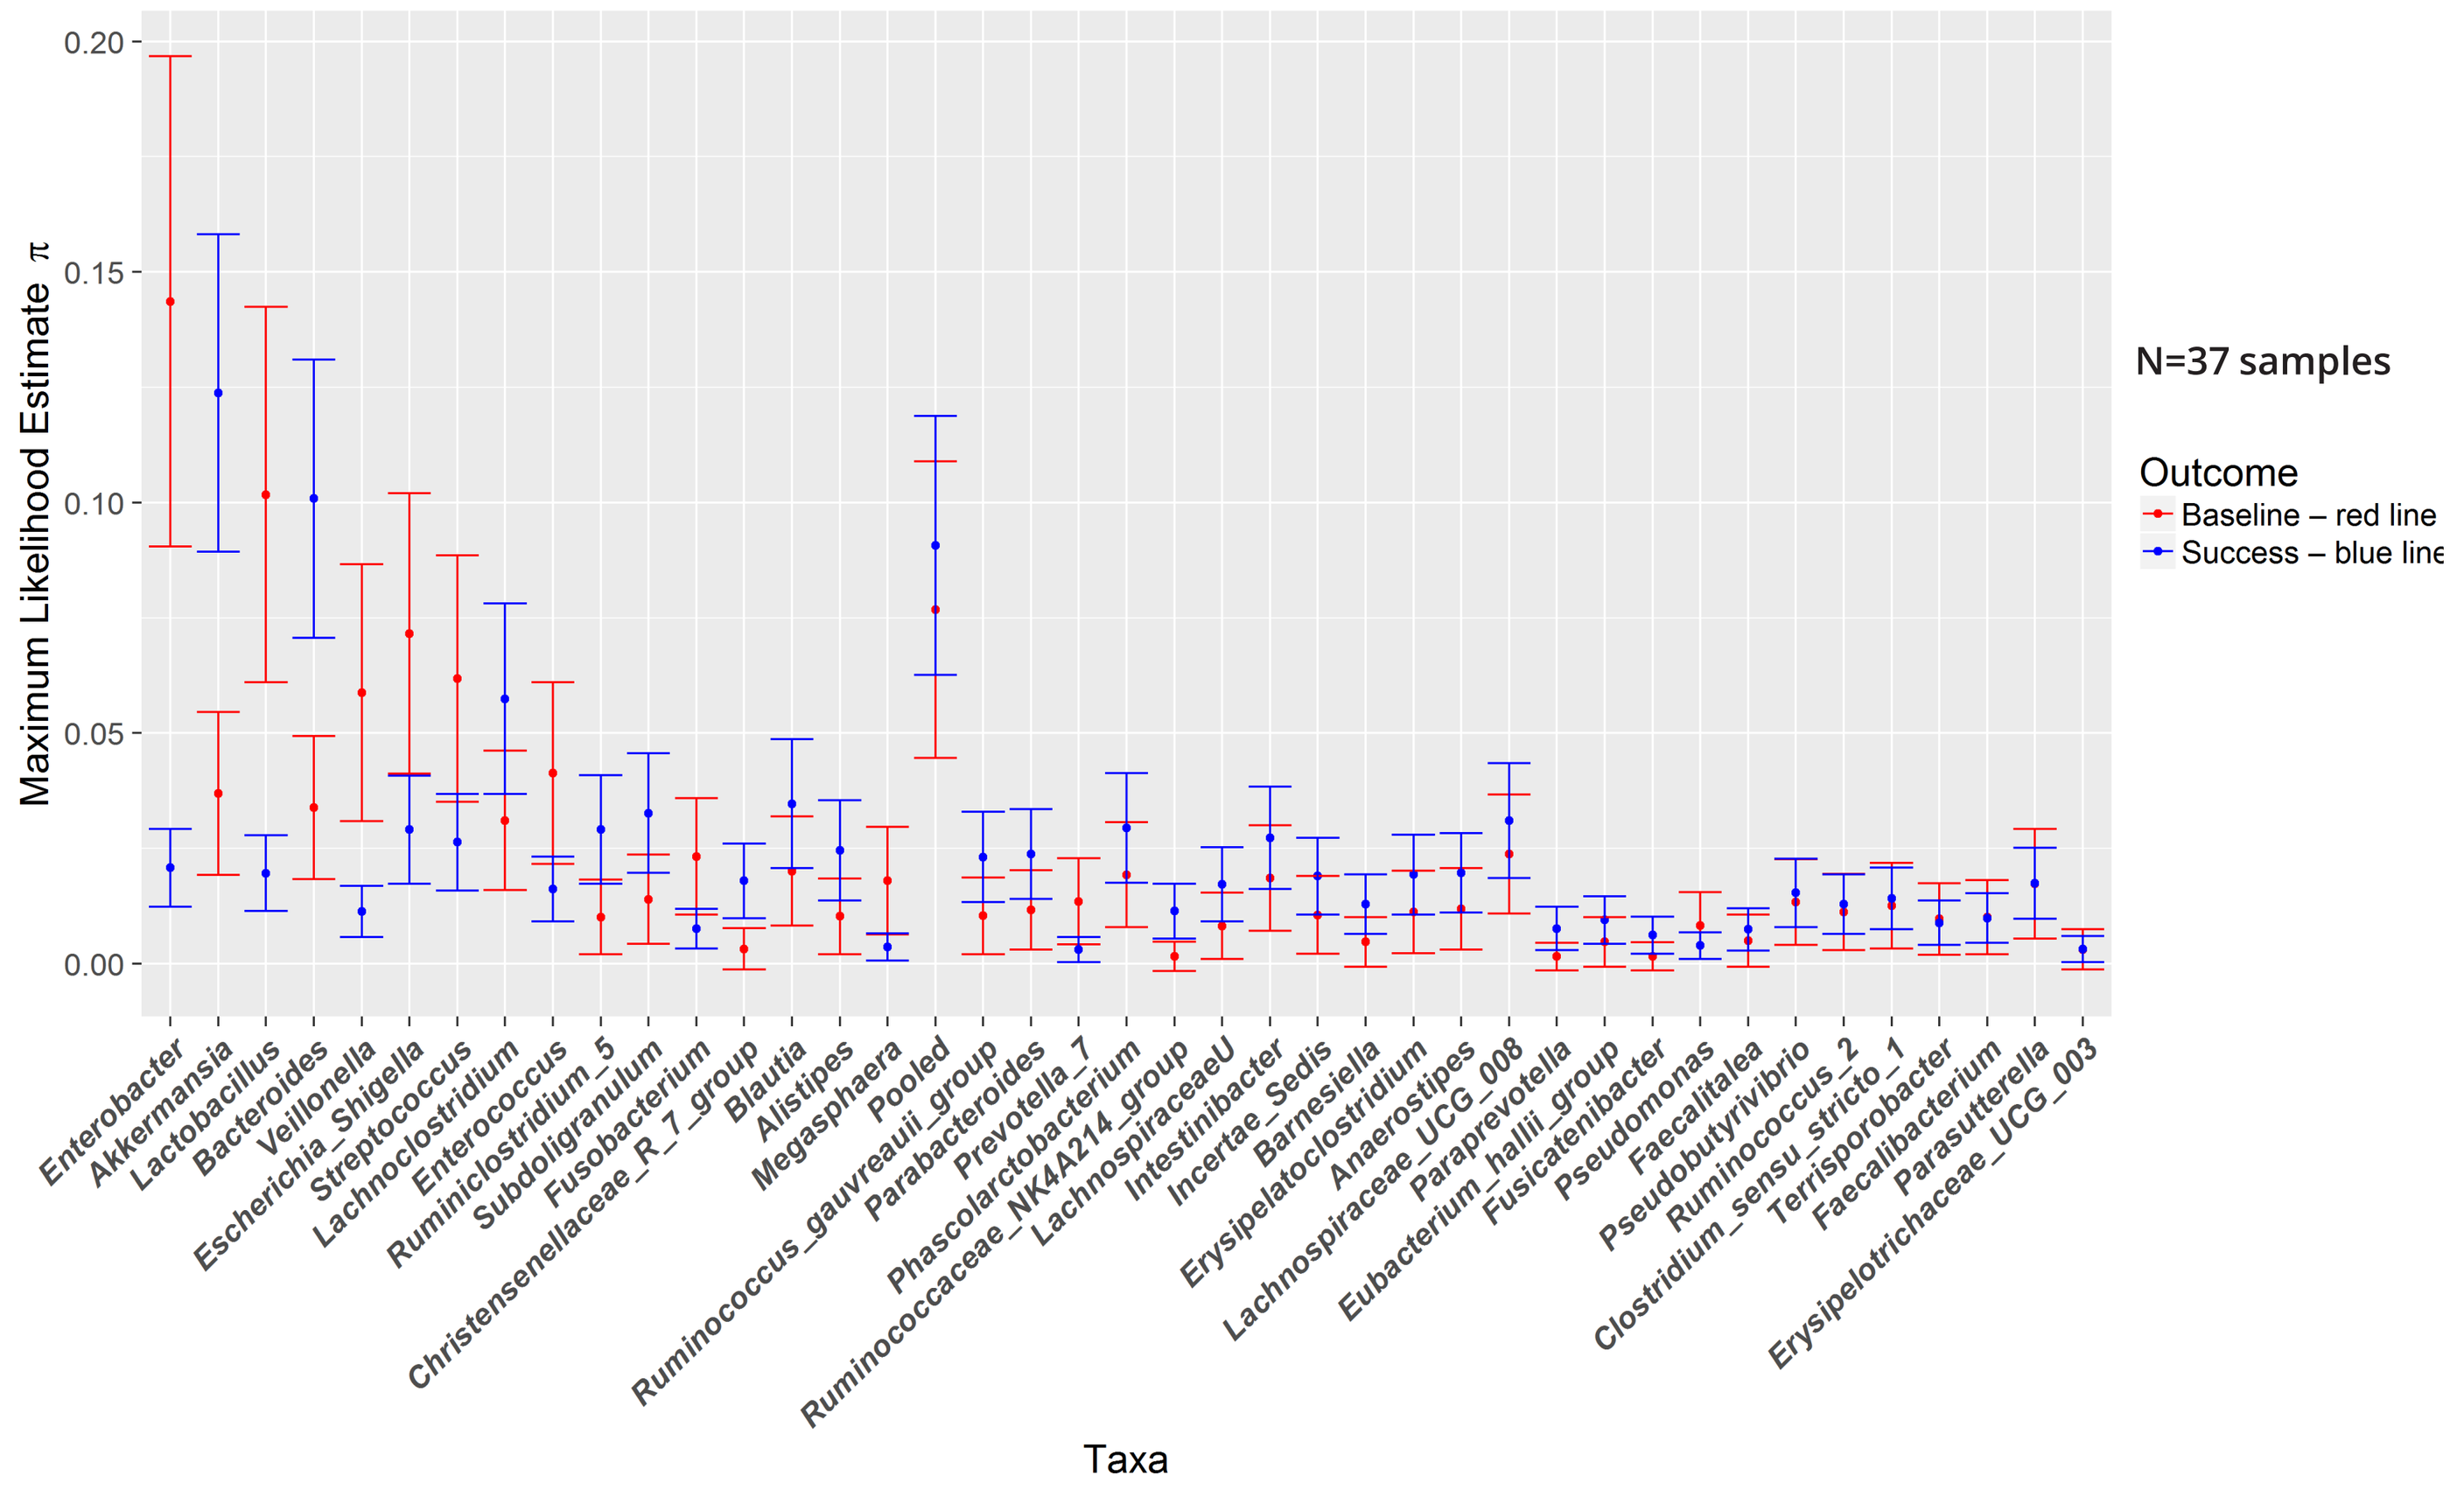

0.20
0.15
0.10
0.05
0.00
Maximum Likelihood Estimate π
Enterobacter
Akkermansia
Lactobacillus
Bacteroides
Veillonella
Escherichia_Shigella
Streptococcus
Lachnoclostridium
Enterococcus
Ruminiclostridium_5
Subdoligranulum
Fusobacterium
Christensenellaceae_R_7_group
Blautia
Alistipes
Megasphaera
Pooled
Ruminococcus_gauvreauii_group
Parabacteroides
Prevotella_7
Phascolarctobacterium
Ruminococcaceae_NK4A214_group
LachnospiraceaeU
Intestinibacter
Incertae_Sedis
Barnesiella
Erysipelatoclostridium
Anaerostipes
Lachnospiraceae_UCG_008
Paraprevotella
Eubacterium_hallii_group
Fusicatenibacter
Pseudomonas
Faecalitalea
Pseudobutyrivibrio
Ruminococcus_2
Clostridium_sensu_stricto_1
Terrisporobacter
Faecalibacterium
Parasutterella
Erysipelotrichaceae_UCG_003
Taxa
N=37 samples
Outcome
Baseline – red line
Success – blue line

The maximum likelihood estimate of π is shown with the 95% confidence interval for each group. The “Pooled” group consists of the least abundant taxa which jointly represent <5% of the overall abundance.

Figure 2. Group B (2 Doses of Placebo): Comparative Microbiota Profiles of Showing Baseline vs. Success

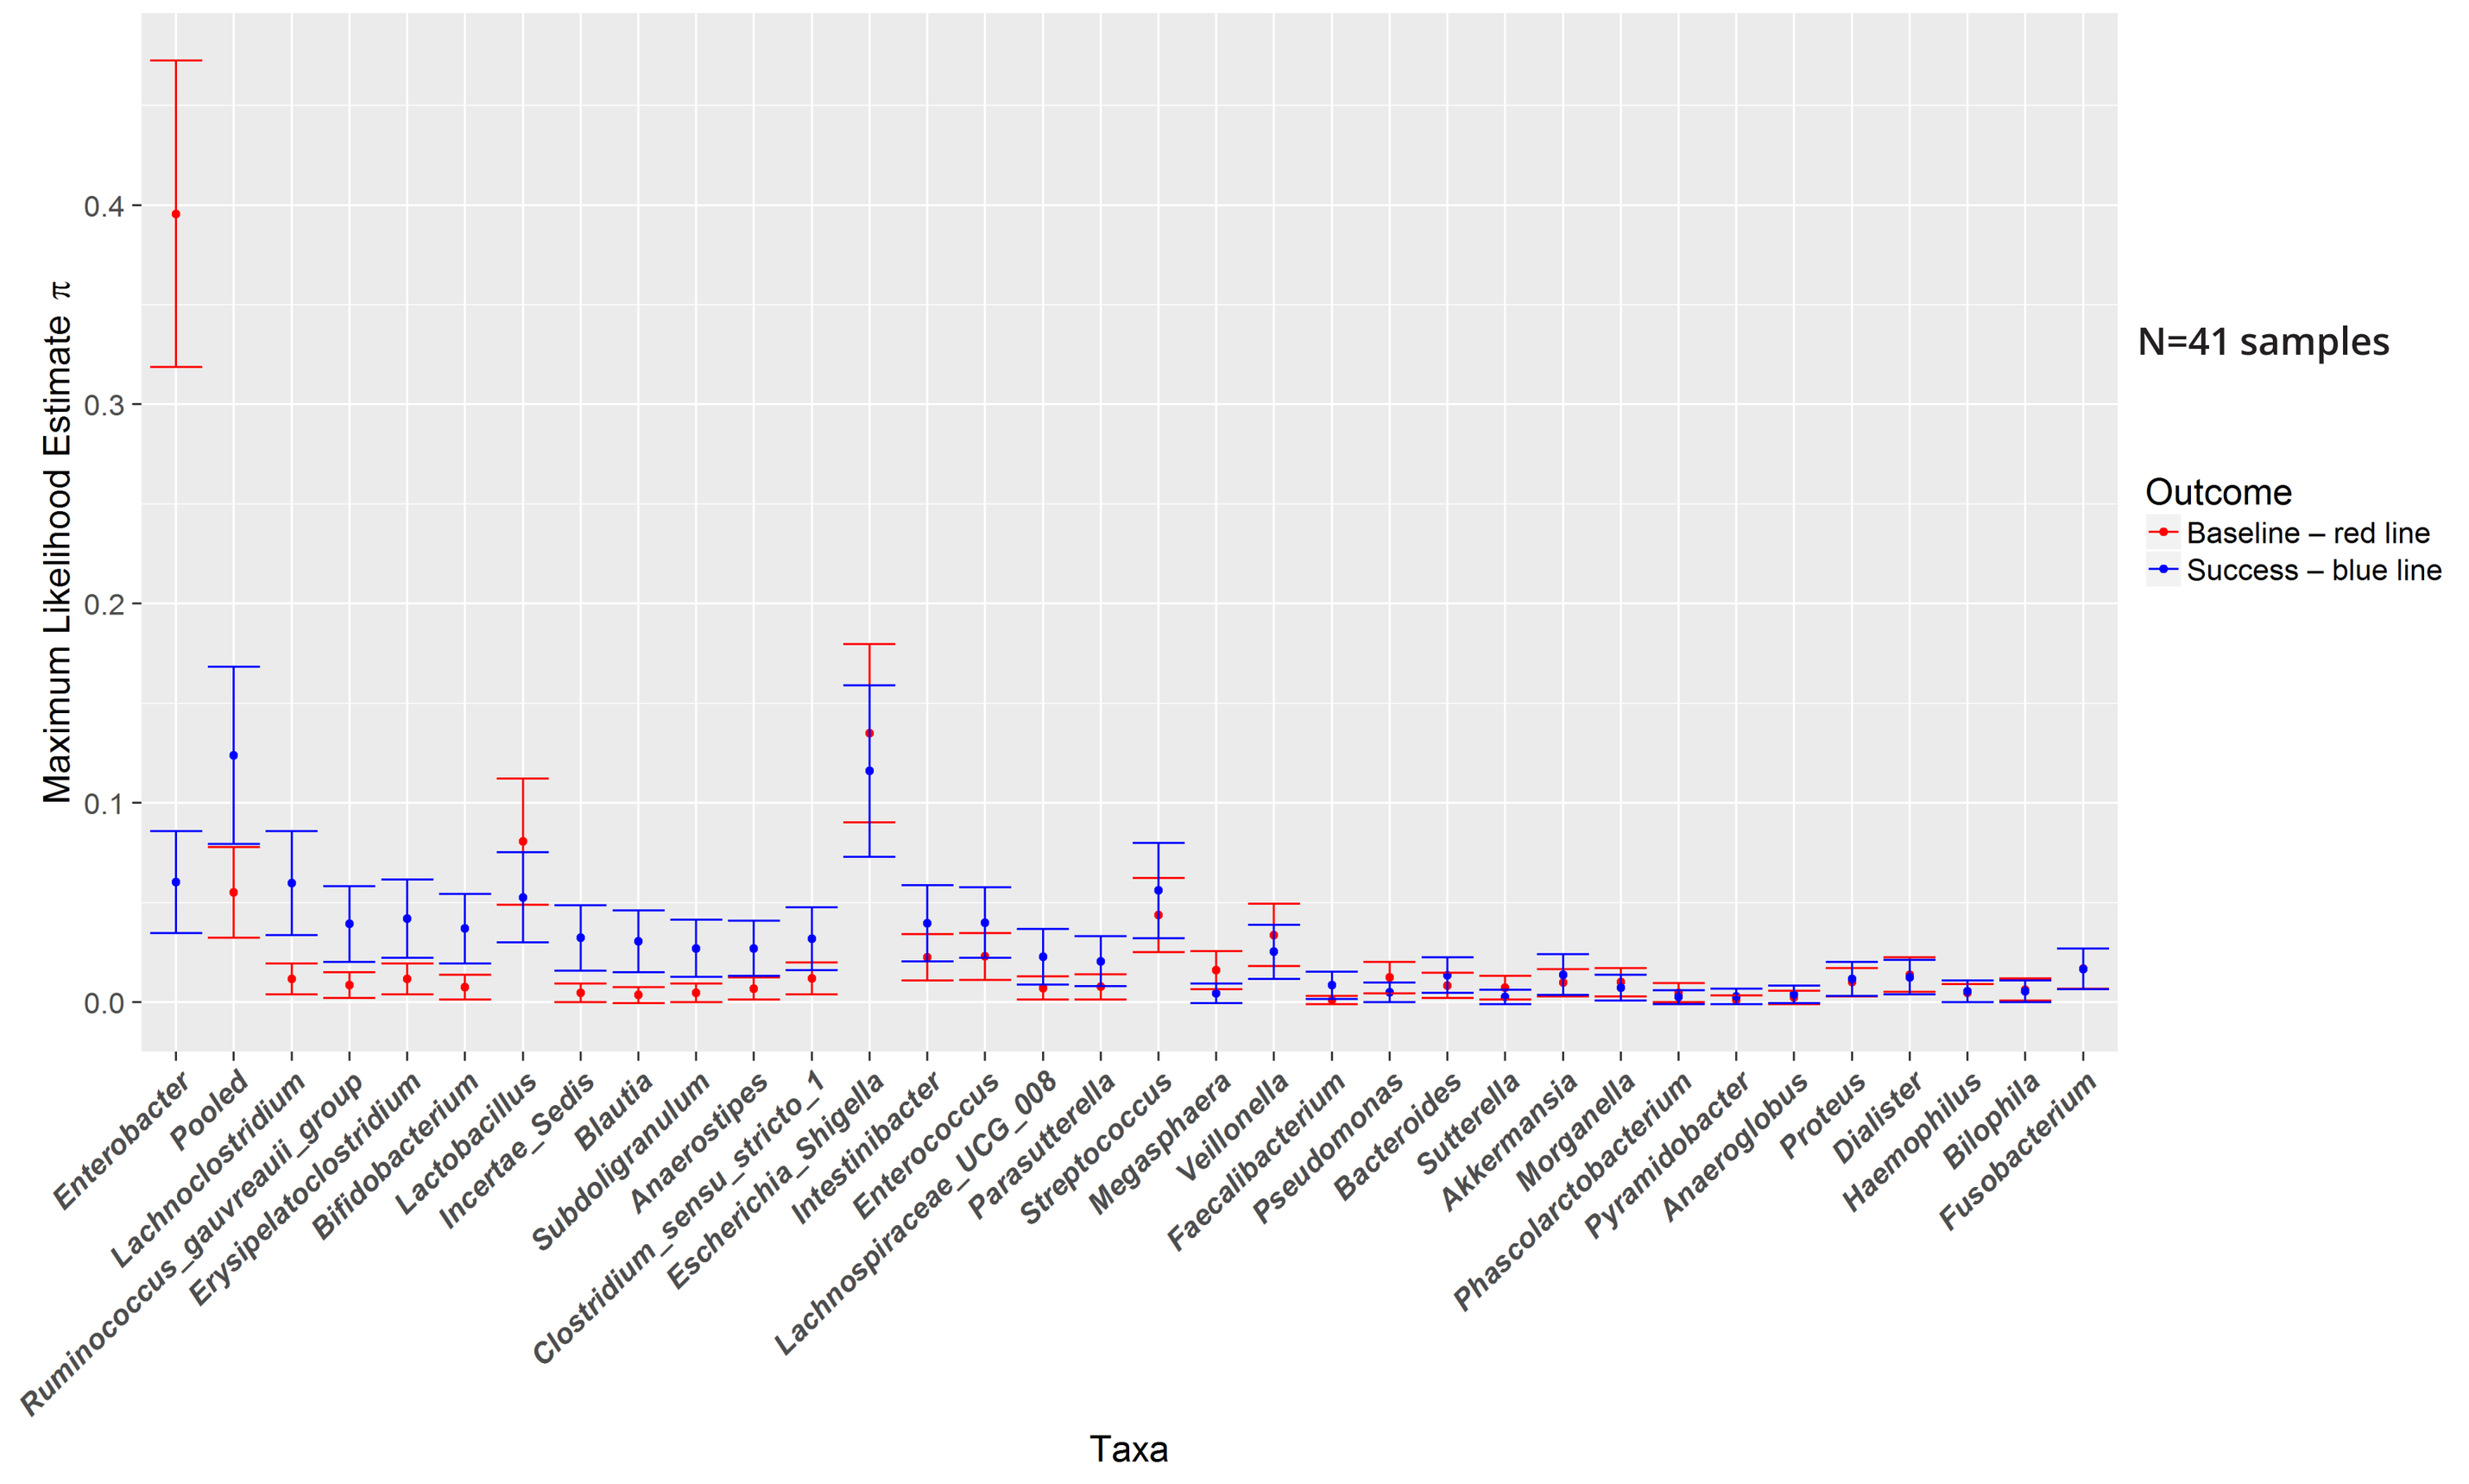

0.4
0.3
0.2
0.1
0.0
Maximum Likelihood Estimate π
Enterobacter
Pooled
Lachnoclostridium
Ruminococcus_gauvreauii_group
Erysipelatoclostridium
Bifidobacterium
Lactobacillus
Incertae_Sedis
Blautia
Subdoligranulum
Anaerostipes
Clostridium_sensu_stricto_1
Escherichia_Shigella
Intestinibacter
Enterococcus
Lachnospiraceae_UCG_008
Parasutterella
Streptococcus
Megasphaera
Veillonella
Faecalibacterium
Pseudomonas
Bacteroides
Sutterella
Akkermansia
Morganella
Phascolarctobacterium
Pyramidobacter
Anaeroglobus
Proteus
Dialister
Haemophilus
Bilophila
Fusobacterium
Taxa
N=41 samples
Outcome
Baseline – red line
Success – blue line

The maximum likelihood estimate of π is shown with the 95% confidence interval for each group. The “Pooled” group consists of the least abundant taxa which jointly represent <5% of the overall abundance.

Figure 3. Group C (1 Dose of RBX2660 and 1 Dose of Placebo): Comparative Microbiota Profiles Profiles Showing Baseline vs. Success

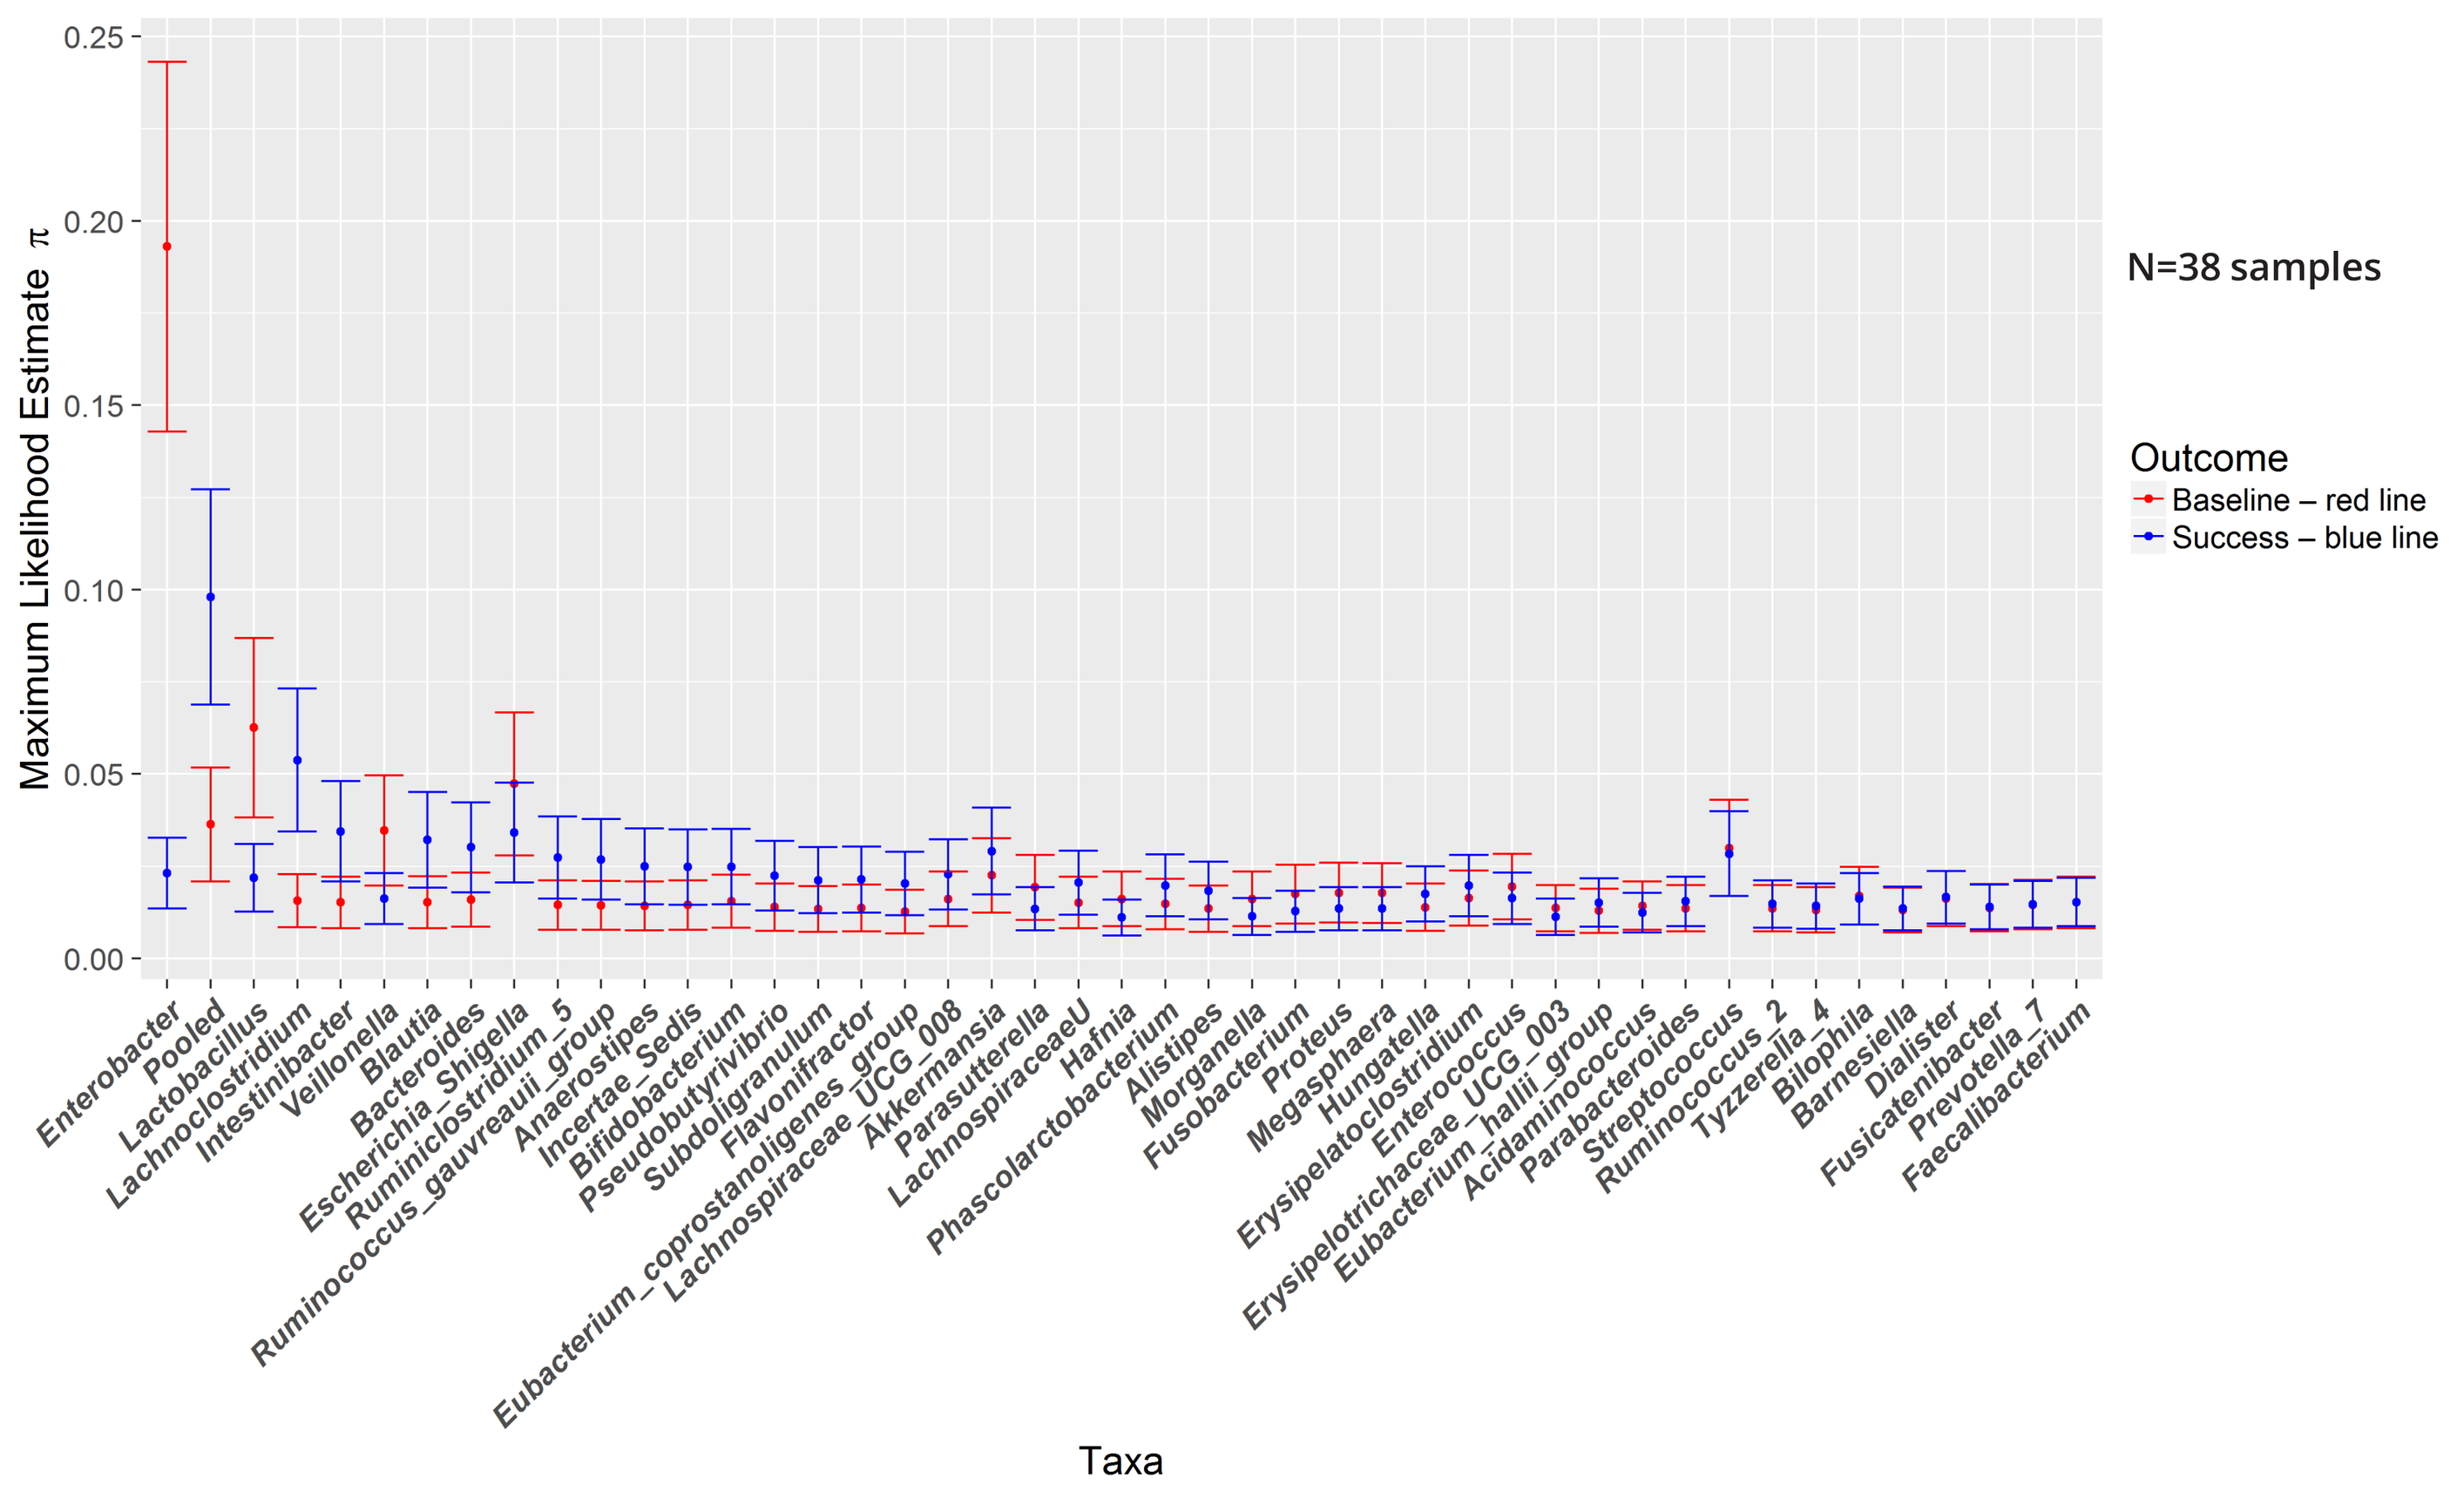

0.25
0.20
0.15
0.10
0.05
0.00
Maximum Likelihood Estimate π
Enterobacter
Pooled
Lactobacillus
Lachnoclostridium
Intestinibacter
Veillonella
Blautia
Bacteroides
Escherichia_Shigella
Ruminiclostridium_5
Ruminococcus_gauvreauii_group
Anaerostipes
Incertae_Sedis
Bifidobacterium
Pseudobutyrivibrio
Subdoligranulum
Flavonifractor
Eubacterium_coprostanoligenes_group
Lachnospiraceae_UCG_008
Akkermansia
Parasutterella
LachnospiraceaeU
Hafnia
Phascolarctobacterium
Alistipes
Morganella
Fusobacterium
Proteus
Megasphaera
Hungatella
Erysipelatoclostridium
Enterococcus
Erysipelotrichaceae_UCG_003
Eubacterium_hallii_group
Acidaminococcus
Parabacteroides
Streptococcus
Ruminococcus_2
Tyzzerella_4
Bilophila
Barnesiella
Dialister
Fusicatenibacter
Prevotella_7
Faecalibacterium
Taxa
N=38 samples
Outcome
Baseline – red line
Success – blue line

The maximum likelihood estimate of π is shown with the 95% confidence interval for each group. The “Pooled” group consists of the least abundant taxa which jointly represent <5% of the overall abundance.

Figure 4. Kullback-Leibler (KL) Divergence Comparison for Arm A (2 doses of RBX2660)

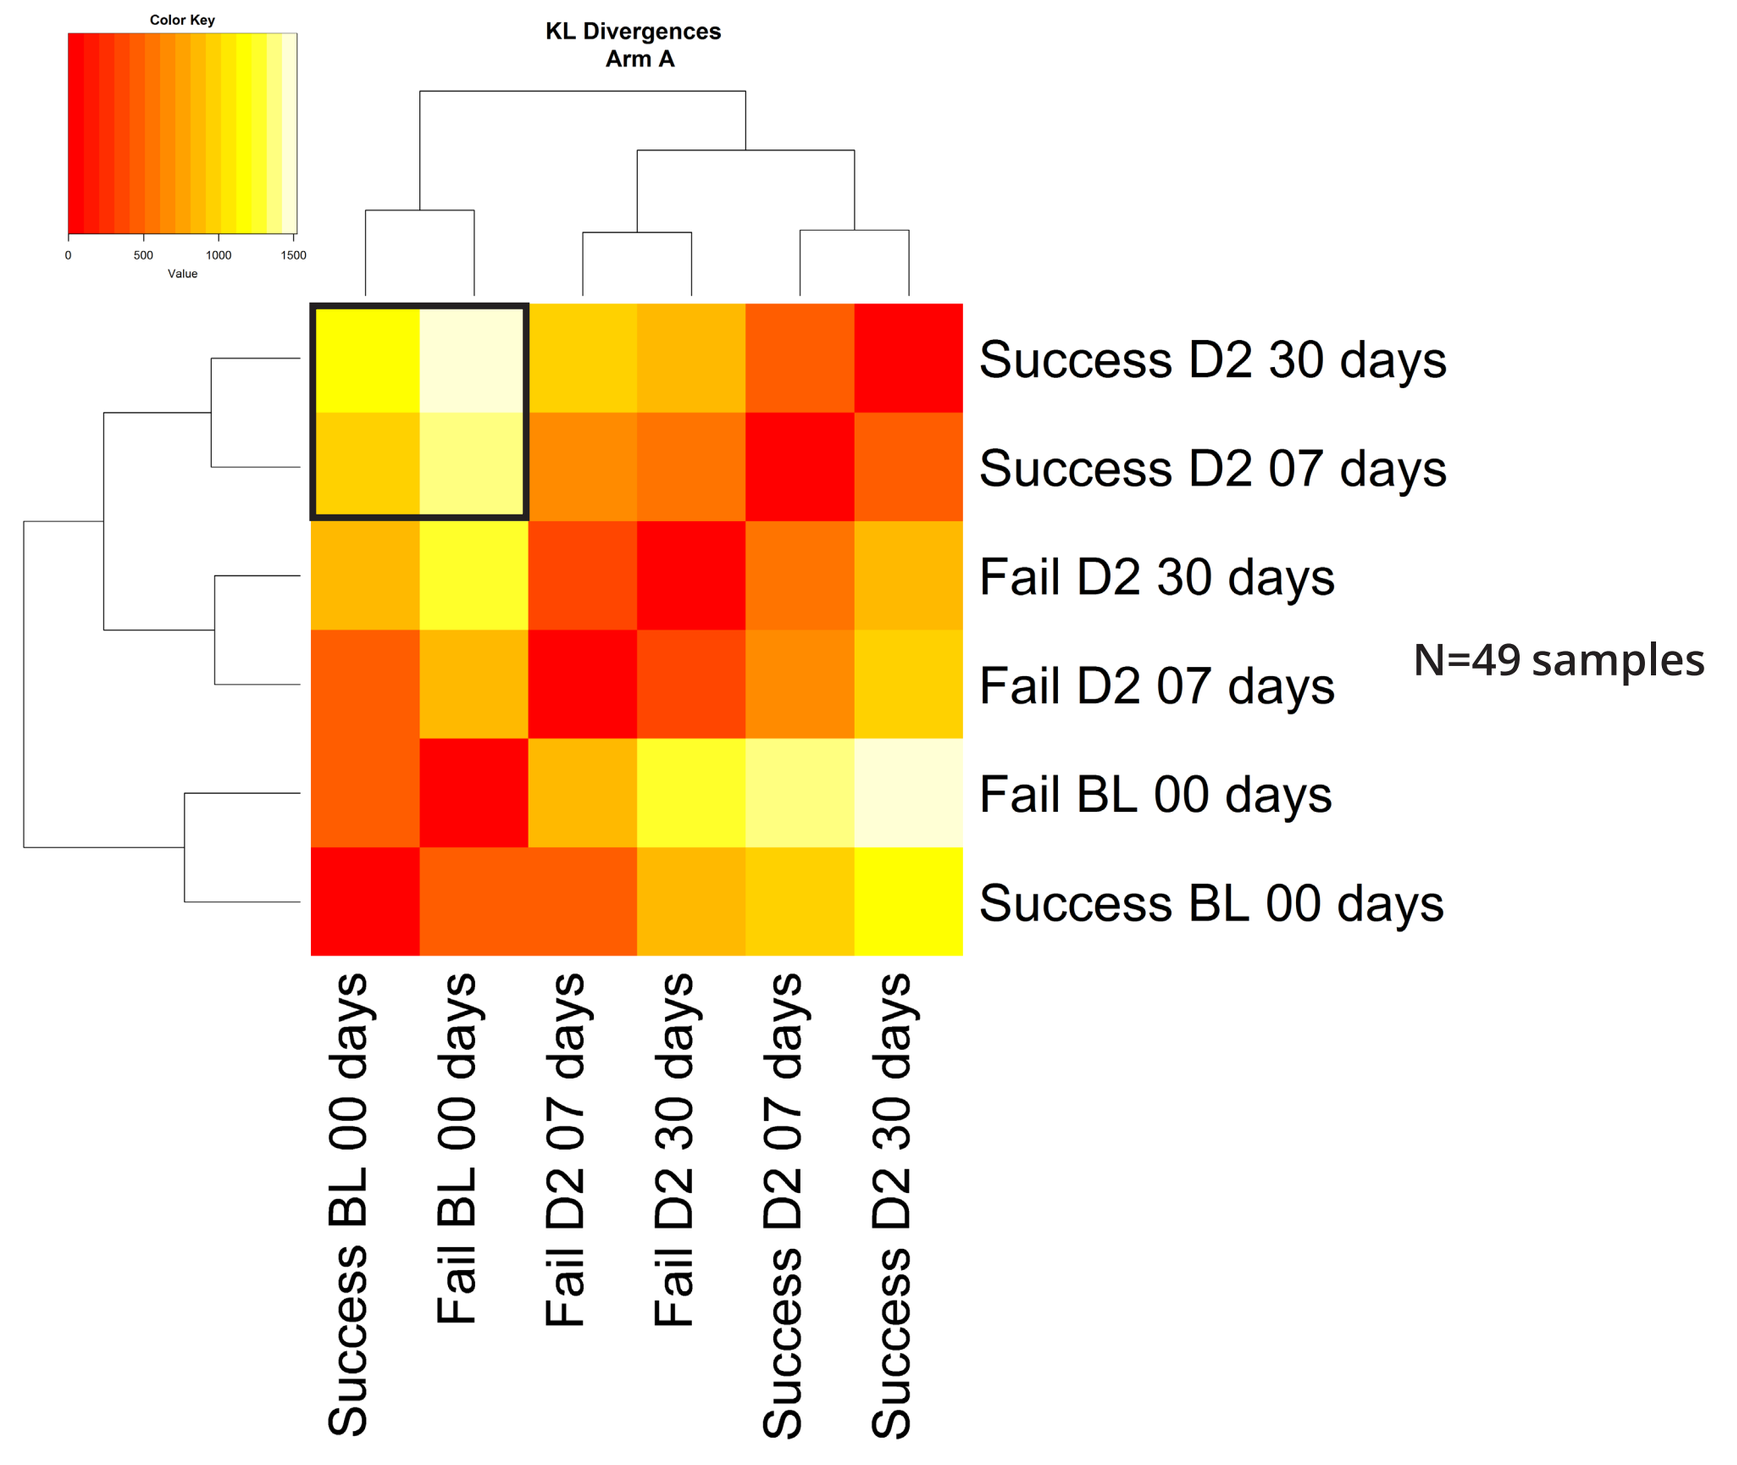

Color Key
0
500
1000
1500
Value
KL Divergences
Arm A
Success D2 30 days
Success D2 07 days
Fail D2 30 days
Fail D2 07 days
Fail BL 00 days
Success BL 00 days
Success BL 00 days
Fail BL 00 days
Fail D2 07 days
Fail D2 30 days
Success D2 07 days
Success D2 30 days
N=49 samples

BL=baseline; D2=Dose 2

A Kullback-Leibler (KL) divergence analysis was used to compare microbiota populations stratified by treatment outcome and time. The plot suggests that for successful patients, the microbiota at 7 and 30 days is very different from baseline (highlighted area). The yellow shading in the heat map corresponds to higher KL divergence.

Conclusions

- In this randomized, placebo-controlled study of RBX2660 for recurrent CDI, 16s rRNA analysis found that patients who succeeded with their assigned therapy had a more diverse intestinal microbiota at 7 and 30 days compared with baseline.

Corresponding Author

Courtney Jones
2660 Patton Road
Roseville, MN 55113
Tel: 651-705-8770
E-mail: cjones@rebiotix.com

This analysis was funded by Rebiotix Inc., Roseville, MN.