Increased Microbial Diversity Found in Successful vs. Unsuccessful Recipients of Next-Generation FMT for Recurrent *Clostridium difficile* Infection

[a]Sahil Khanna, MBBS; [b]Courtney Jones, BS; [b]Lee Jones, MBA; [c]Frederic D Bushman, PhD; [c]Aubrey Bailey, MS

[a]Division of Gastroenterology and Hepatology, Mayo Clinic, Rochester, MN, [c]Rebiotix Inc., Roseville, MN; [c]Department of Microbiology, Perleman School of Medicine, University of Pennsylvania, Philadelphia, PA

Corresponding Author
Sahil Khanna, MBBS
Division of Gastroenterology and Hepatology
Mayo Clinic
200 1st Street SW,
Rochester, MN 55905, USA
khanna.sahil@mayo.edu

Background

- Patients with recurrent *Clostridium difficile* infection (CDI) have been found to have markedly decreased fecal microbiota diversity.[1-3] Diversity increases after successful fecal microbiota transplantation (FMT).[4,5]
- While the fecal microbiota of patients with recurrent CDI becomes more diverse, it remains dynamic following FMT.[5]

Objective

- We evaluated the gut microbiota of patients with recurrent CDI after successful and unsuccessful treatment with RBX2660, a Microbiota Restoration Therapy (MRT) sourced from live human-derived microbes.

Methods

- The fecal microbiota of a subset of 17 patients who participated in the previously reported Phase 2 PUNCH CD study assessing RBX2660 for recurrent CDI were characterized at 7 and 60 days post treatment using 16S rRNA gene sequencing in a post-hoc analysis.
- RBX2660 was delivered via enema.
- The patients (median 69.2 years; 65% female; 94% white) included 8 successes with 1 dose of RBX2660; 6 with 2 doses and 3 failures with 2 doses.
- Success was defined as the absence of CDI-associated diarrhea (passage of three or more unformed stools in 24 or fewer consecutive hours for at least two consecutive days through 8 weeks after the last dose of RBX2660).
- Failure was defined as a recurrence of CDI symptoms < 8 weeks after the last dose of RBX2660; going back on antibiotic treatment for CDI or a CDI-related hospitalization.

Patient Selection

- The patient cohort sequenced and analyzed was representative of the outcomes seen in the PUNCH CD study:
 - Single-dose success
 - Single-dose failure (opted out of 2nd dose of RBX2660)
 - Two-dose success
 - Two-dose failure

Sample Collection

- Patients supplied stool samples at baseline (pre-treatment) and at 7 days, 30 days, 60 days and 180 days (approximately) after treatment with RBX2660.
- Sample collection was restarted using the same schedule if a second dose of RBX2660 was received.
- Samples were collected at the patient's home (or care facility) and shipped overnight to Rebiotix in a cold pack.
- Aliquots of stool were stored at -80 °C until completion of the sample cycle.

Sequencing and Analysis

- All sequencing and analysis was performed by the CFAR Viral/ Molecular High Density sequencing core at the University of Pennsylvania.
- DNA sequencing was performed on an Illumina MiSeq platform.
- The 16S sequences were clustered into operational taxonomic units (OTUs) and used to determine within-sample diversity and taxonomic composition at each time point. Weighted Unifrac analysis was used to determine differences in between-sample diversity.
- Box and whisker plots were used to represent microbial diversity. Samples were rarefied to an even depth of 20,000 OTUs per sample to account for variation in sequencing effort.
- Abundance-weighted Unifrac distances were calculated and used to examine the shared community structures between samples.

Results

- There was no significant difference in diversity between successes and failures with RBX2660 at 7 days. A trend toward higher diversity was seen in patients who were successfully treated with RBX2660.
- At day 60, there was a significantly increased microbial richness in patients with successful treatments compared with failure, P =0.008. Successfully treated patients had increased microbial diversity, regardless of whether they had 1 or 2 doses of RBX2660, Figure 1.
- Unifrac analysis suggested that the microbial diversity of successfully treated patients was similar; the microbial diversity of failures was dissimilar by PERMANOVA test, P=0.05, Figure 2.

Figure 1. Rarefied to 20,000 OTU Observations Per Sample

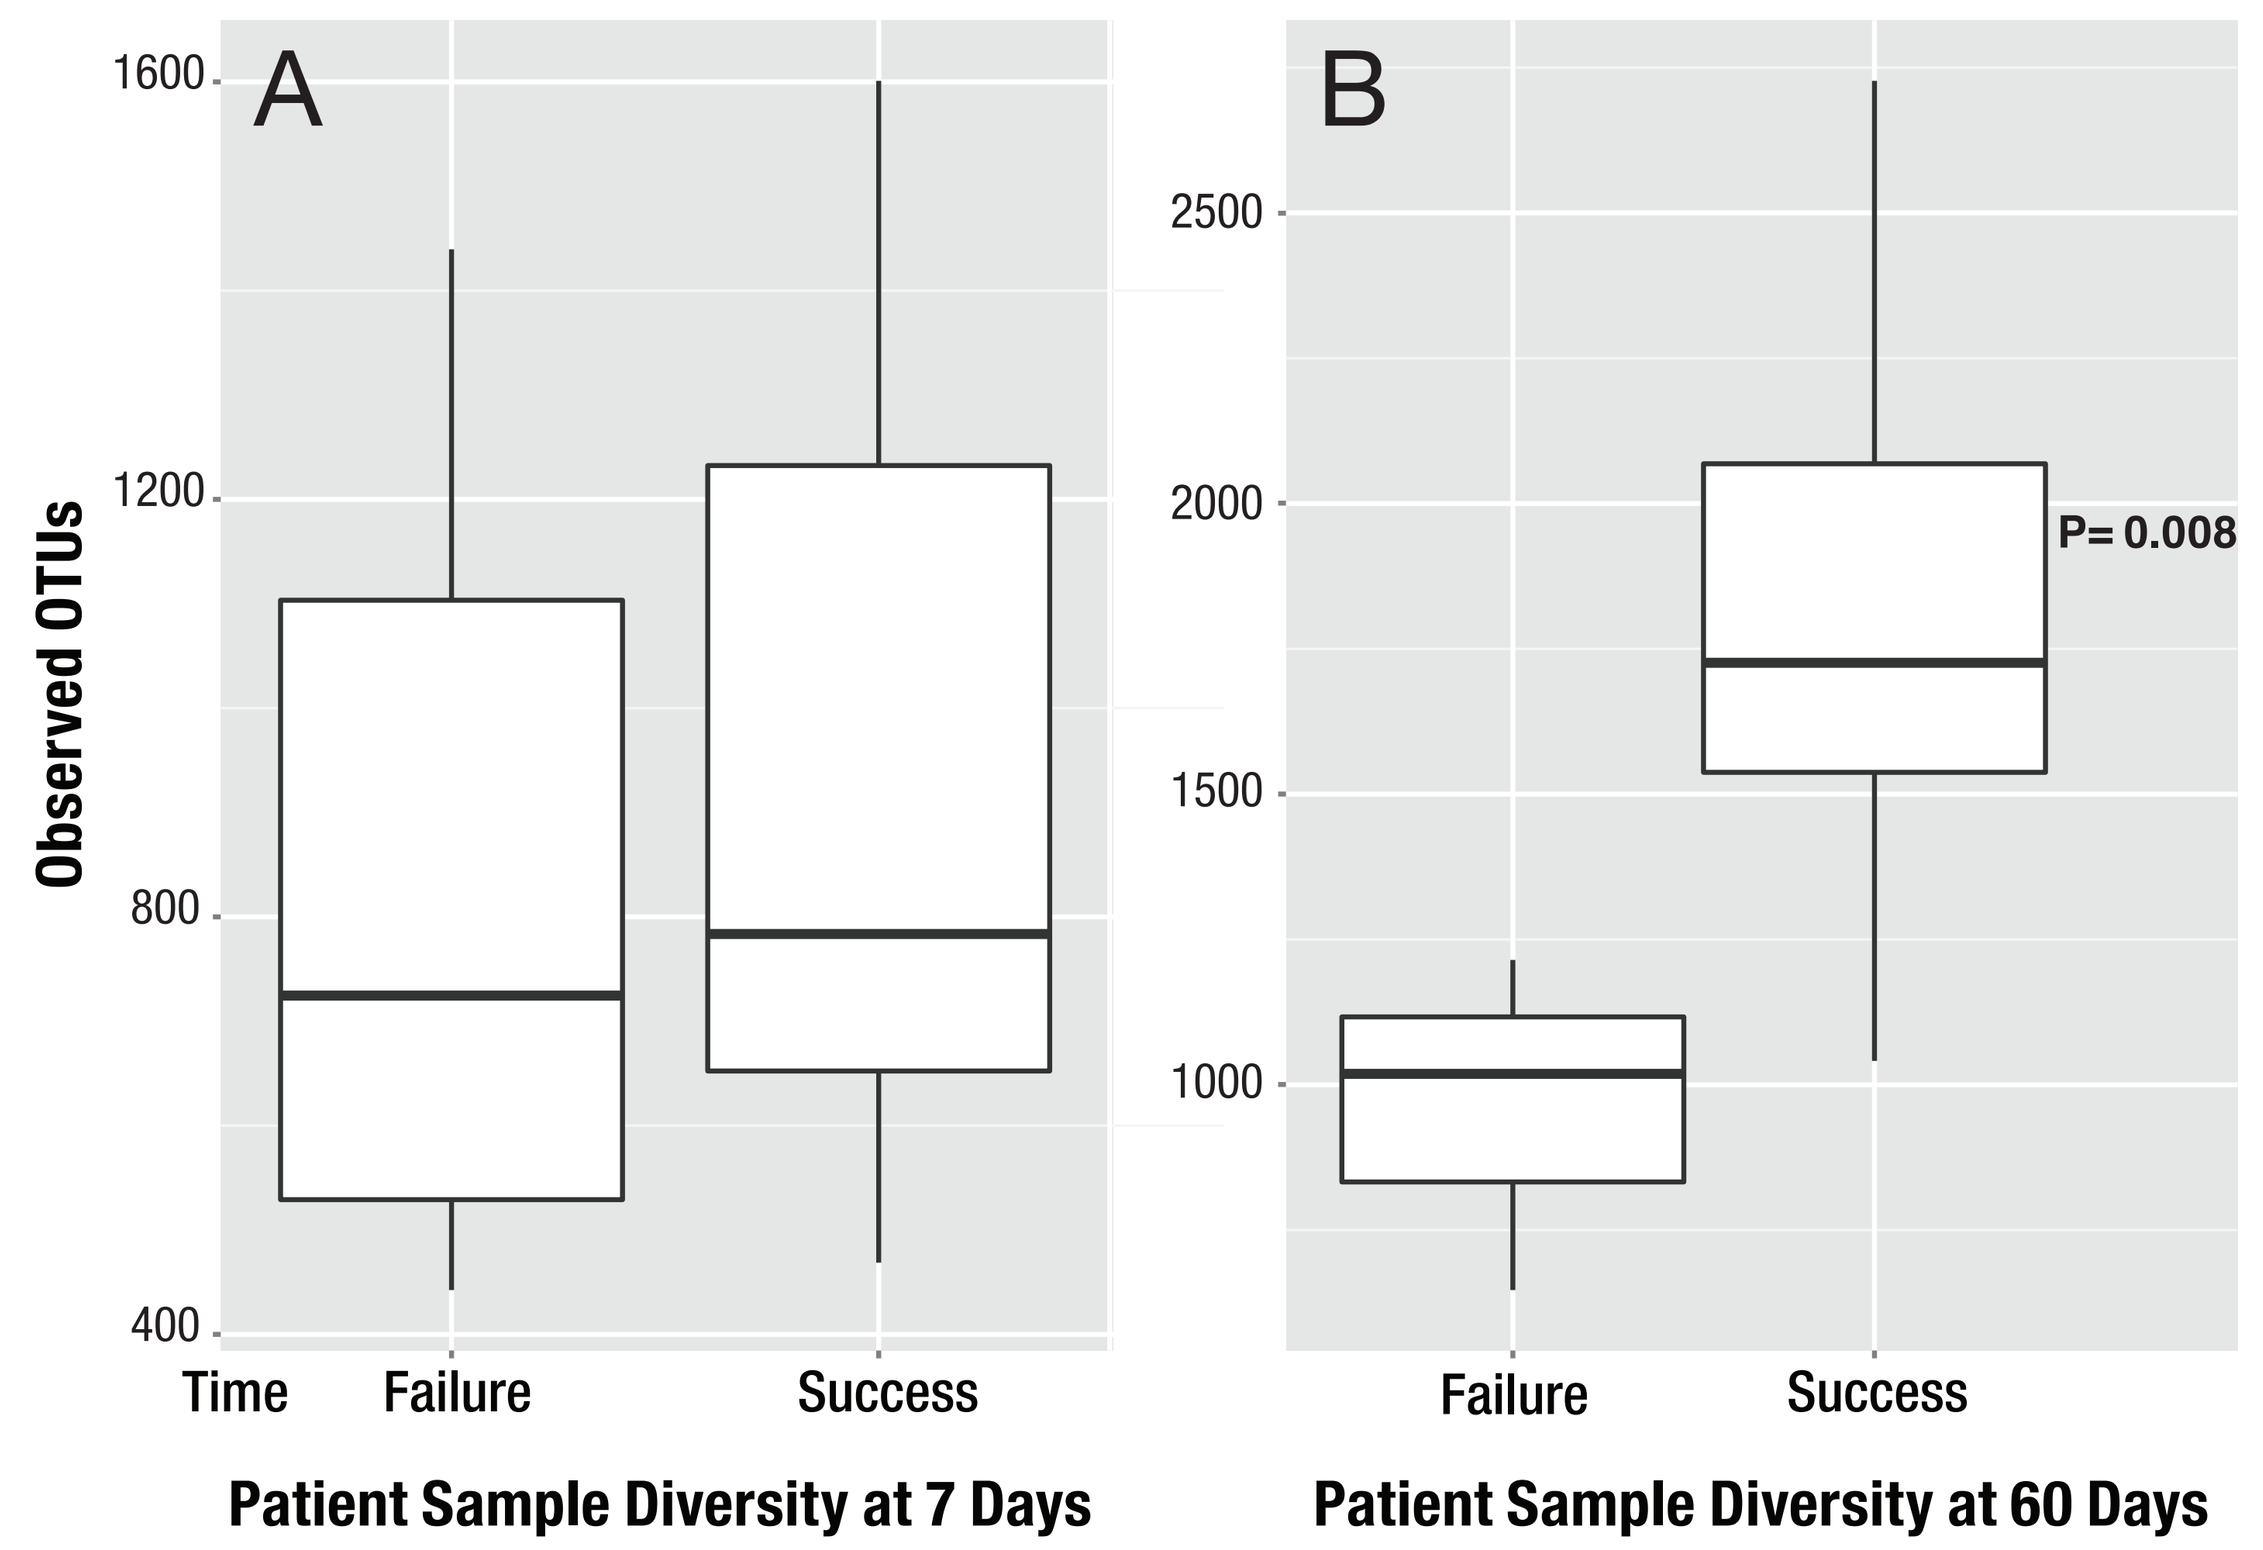
A
B
Observed OTUs
1600
1200
800
400
2500
2000
1500
1000
P= 0.008
Time
Failure
Success
Failure
Success
Patient Sample Diversity at 7 Days
Patient Sample Diversity at 60 Days

Figure 2. Weighted Unifrac Principal Coordinates Analysis at Day 60

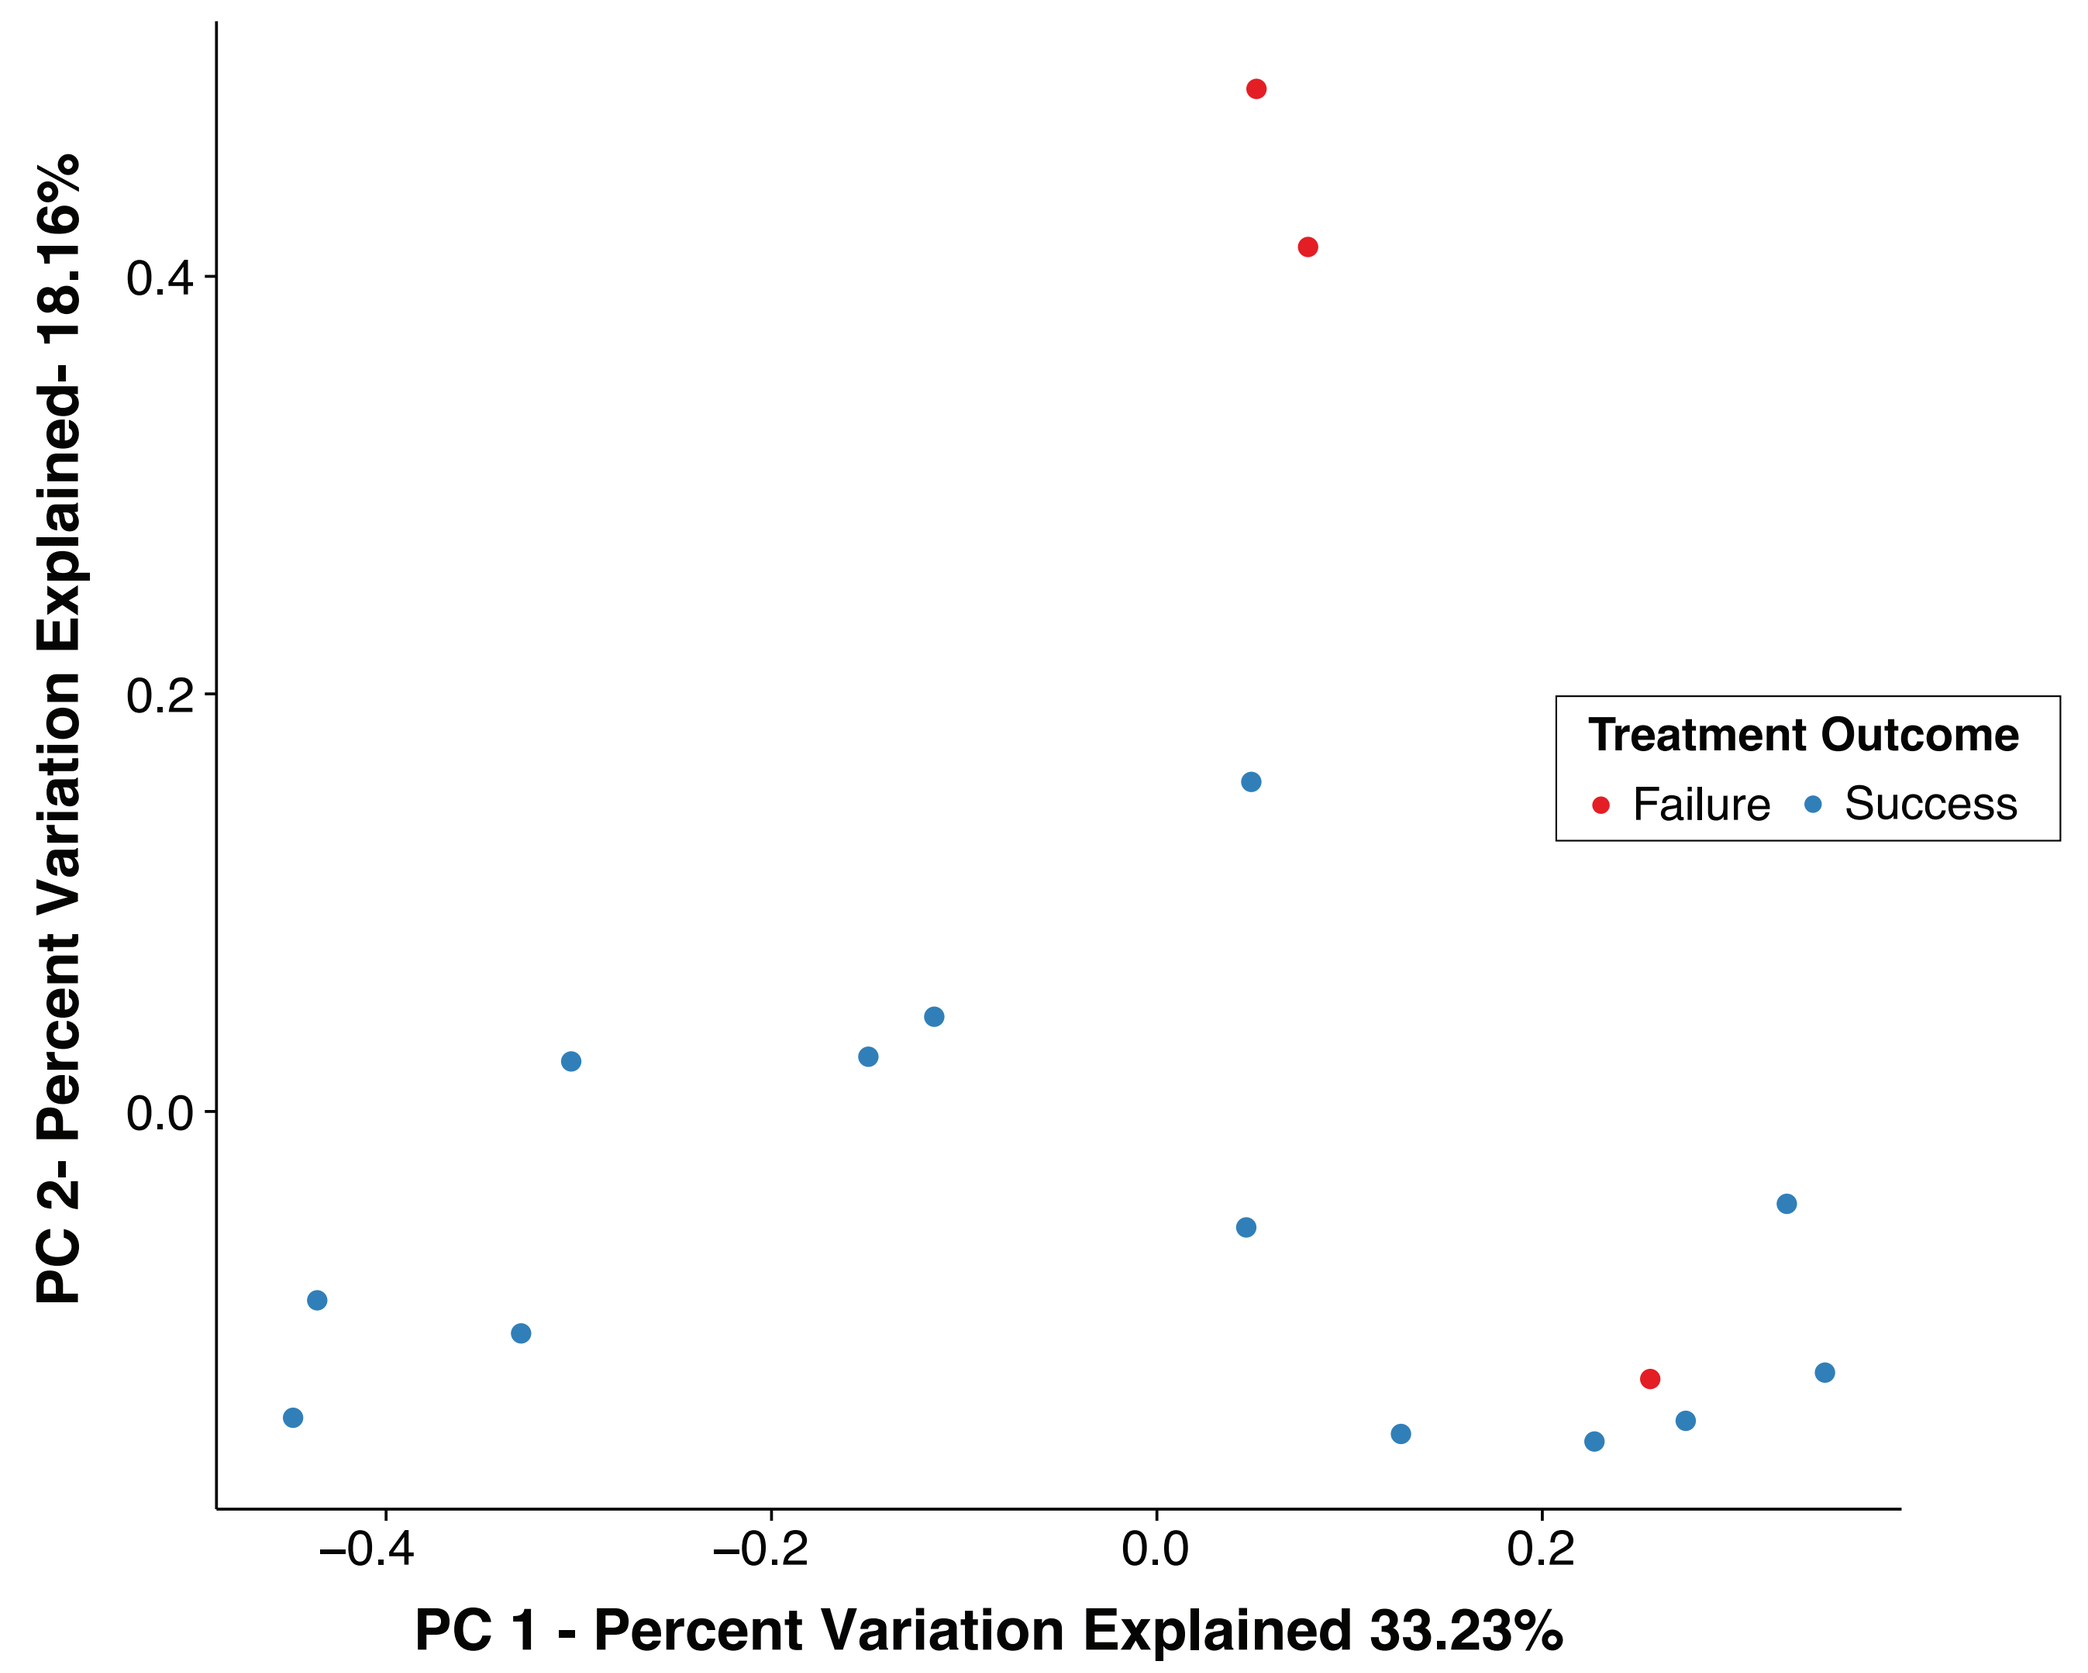
PC 2- Percent Variation Explained- 18.16%
PC 1 - Percent Variation Explained 33.23%
Treatment Outcome
Failure
Success
0.4
0.2
0.0
−0.4
−0.2
0.0
0.2

- Each point represents a patient who was sequenced (N=17). The variation between the samples is shown by the location of the point and proximity to other points (the closer any two points are to each other, the more similar the points are to each other).
- At day 60, the variation in the microbiomes of patients with successful outcomes was most closely related to other patients with successful outcomes. The microbiomes of patients who failed treatment with RBX2660 were different from those of patients who were successfully treated and from each other.

Conclusions

- Patients with recurrent CDI who responded to treatment with RBX2660 had a more diverse gut microflora at day 60 compared with patients who failed treatment but not at day 7.
- The results suggest a time dependence for re-establishing a diverse microbiome.
- The normal microbial diversity associated with successful treatment of recurrent CDI with RBX2660 is consistent with previously reported results obtained with FMT.
- The observed correlation between higher microbial diversity and treatment success could potentially serve as an early indicator of treatment outcomes as well as provide a biomarker for use in future MRT products.

References

1. Chang JY, Antonopoulos DA, Kalra A. et al. Decreased diversity of fecal microbiome in recurrent *Clostridium difficile*-associated diarrhea. *JID*; 2008;197:435-8.
2. Seekatz AM, Young VB. *Clostridium difficile* and the microbiota. *J Clin Invest.* 2014;124:4182-9.
3. Shankar V, Hamilton MJ, Khoruts A. Species and genus level resolution analysis of gut microbiota in *Clostridium difficile* patients following fecal microbiota transplantation. *Microbiome.* 2014;2:13.
4. Fuentes S, van Nood E, Tims S, et al. Reset of a critically disturbed microbial ecosystem: Faecal transplant in recurrent *Clostridium difficile* infection. *ISME J.* 2014;8:1621-33.
5. Weingarden A, Gonzalez A, Vazquez-Baeza Y, et al. Dynamic changes in short- and long-term bacterial composition following fecal microbiota transplantation for recurrent *Clostridium difficile* infection. *Microbiome.* 2015;3:10.