Poster# 1870

Microbiome Profile is Distinct in Patients with Successful Response to Microbiota-Based Drug RBX2660 Relative to Placebo Responders

Corresponding Author

Ken Blount, PhD
2660 Patton Road
Roseville, MN 55113
Phone: 651-705-8770
Email: kblount@rebiotix.com

Erik R. Dubberke MD, MSPH[1], Robert Orenstein DO[2], Christine H. Lee MD[3], Sahil Khanna MBBS[4], Gail Hecht MD[5], Ken Blount PhD[6], Bill Shannon PhD MBA[7]

[1]Department of Internal Medicine, Washington University in St. Louis, St. Louis, MO, USA; [2]Division of Infectious Diseases, Mayo Clinic in Arizona, Phoenix, AZ, USA; [3]Hamilton Regional Laboratory Medicine Program, Department of Pathology and Molecular Medicine, McMaster University, Hamilton, ON, Canada, Vancouver Island Health Authority, Victoria, Department of Pathology and Laboratory Medicine, University of British Columbia, BC, Canada; [4]Division of Gastroenterology and Hepatology Mayo Clinic, Rochester, MN, USA; [5]Division of Gastroenterology and Nutrition, Loyola University Medical Center, Chicago, IL, USA; [6]Rebiotix, Inc. Roseville, MN, USA; [7]BioRankings LLC St. Louis, MO, USA

Background

- Recurrent *Clostridium difficile* infections (rCDI) are associated with an altered microbiome composition and diversity compared to healthy patients
- RBX2660, a standardized microbiota-based drug designed to rehabilitate patients' microbiomes, was superior to placebo for preventing rCDI in a Phase2B clinical trial
- The Phase 2B placebo-treated patients demonstrated a response rate that is consistent with the results from placebo-treated groups of other randomized-controlled rCDI trials
- This study sought to understand whether there was a significant difference in the extent of microbiome recovery between patients who responded in the RBX2660 treatment groups and the placebo treatment group

Methods

- Patients with rCDI were enrolled in the PUNCH CD2 trial and randomized to receive blinded treatment by enema of 2 doses of RBX2660 (Group A), 2 doses of placebo (Group B), or 1 dose of RBX2660 and 1 dose of placebo (Group C), with doses 7 days apart (Figure 1)
- Success was defined as the absence of CDI at 8 weeks following completion of the last treatment. Patients were classified as a treatment failure if all four (4) of the following criteria were met: recurrence of diarrhea less than 8 weeks after administration of the last assigned study enema, a positive laboratory diagnosis of *C. difficile* as conducted and reported by the study investigator, a need for retreatment for CDI, and no other cause for diarrhea
- Patients voluntarily submitted stool samples at baseline (pre-treatment) 7, 30, and 60 days after treatment
- 16s rRNA analysis using the Illumina MiSeq platform was performed on stool samples collected from subjects with successful outcomes
- The variable region V4 was targeted to identify the operational taxonomic units (OTUs) in each sample
- Relative abundance data from subject samples were grouped longitudinally and compared among groups using a Bray-Curtis dissimilarity calculation with non-metric multi-dimensional scaling. Additional analyses were performed based on the Dirichlet-Multinomial distribution to compare group mean relative taxonomic abundances (pi) and within group over dispersion (theta)
- Differentiation between sample communities were visualized using a Kullback-Leibler (KL) divergence analysis model (BioRankings, St. Louis, MO), a measure of the difference between microbial diversity at different time points or between different samples

Figure 1: PUNCH™ CD2 Trial Design

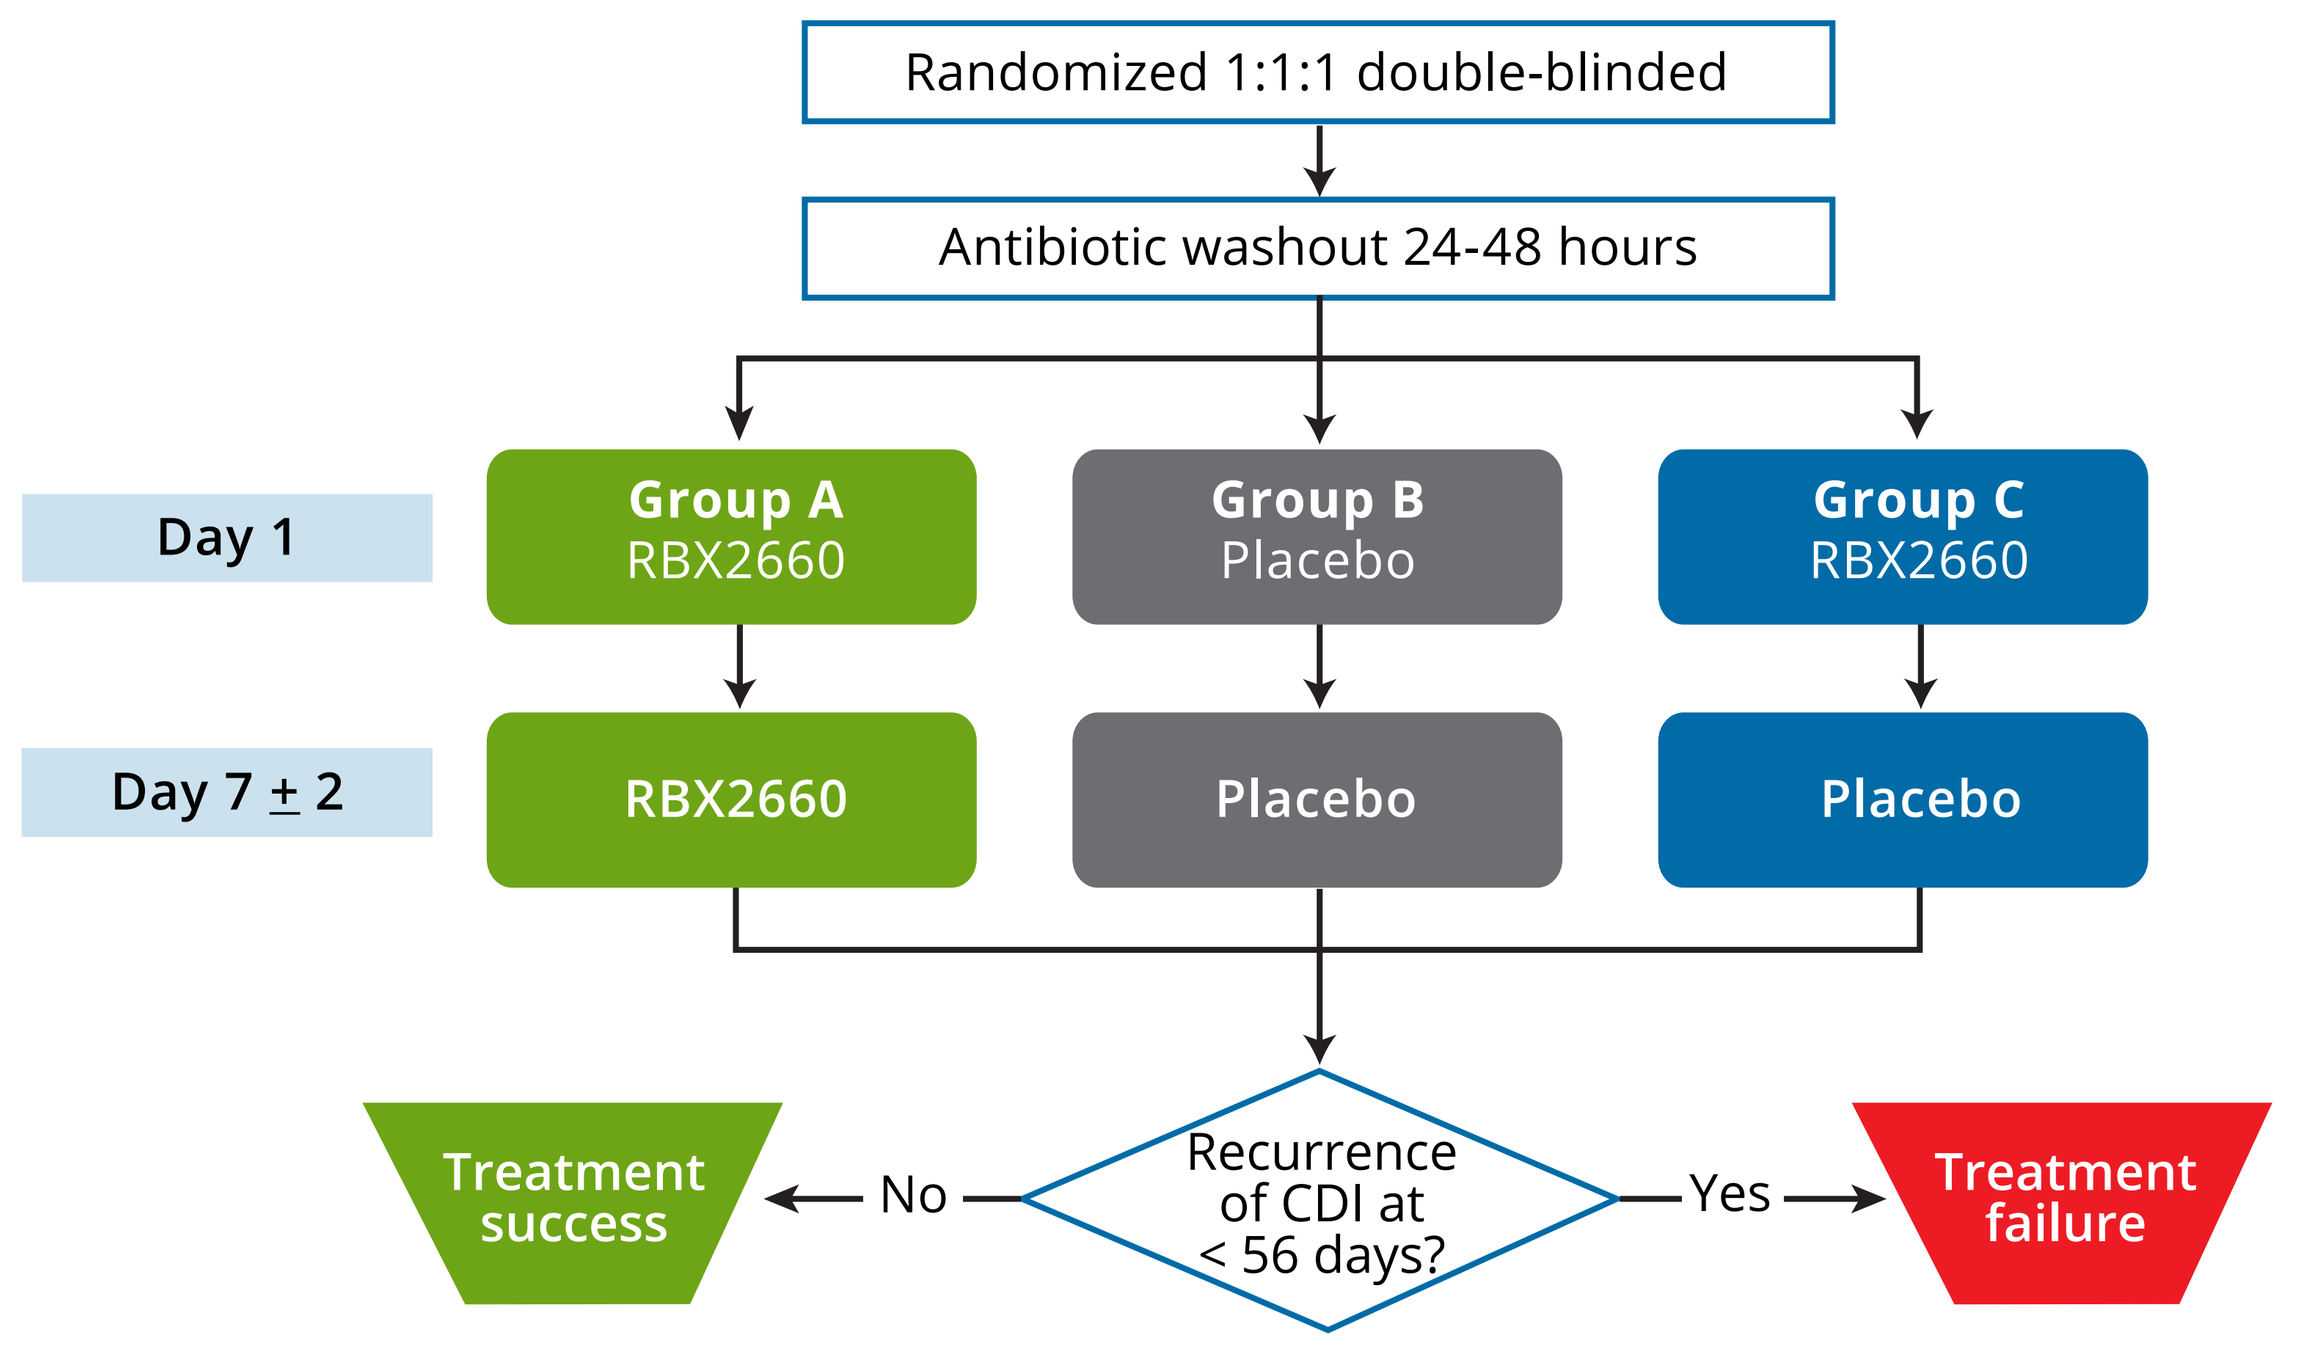

Randomized 1:1:1 double-blinded
Antibiotic washout 24-48 hours
Day 1
Group A RBX2660
Group B Placebo
Group C RBX2660
Day 7 ± 2
RBX2660
Placebo
Placebo
Treatment success
No
Recurrence of CDI at < 56 days?
Yes
Treatment failure

PUNCH™ CD2 Clinical Trial Results

- A single dose of RBX2660 was significantly better than placebo (67%, Group C vs 46% Group B, p=0.48) (Figure 2; Intent-to-Treat analysis)
- A second dose of RBX2660 one week later (Group A) did not provide additional clinical benefit
- Groups A and C combined were superior to placebo (p=0.046)

Figure 2: Patients who received at least one dose of microbiota-based drug RBX2660 showed greater efficacy than placebo-treated patients following blinded treatment.

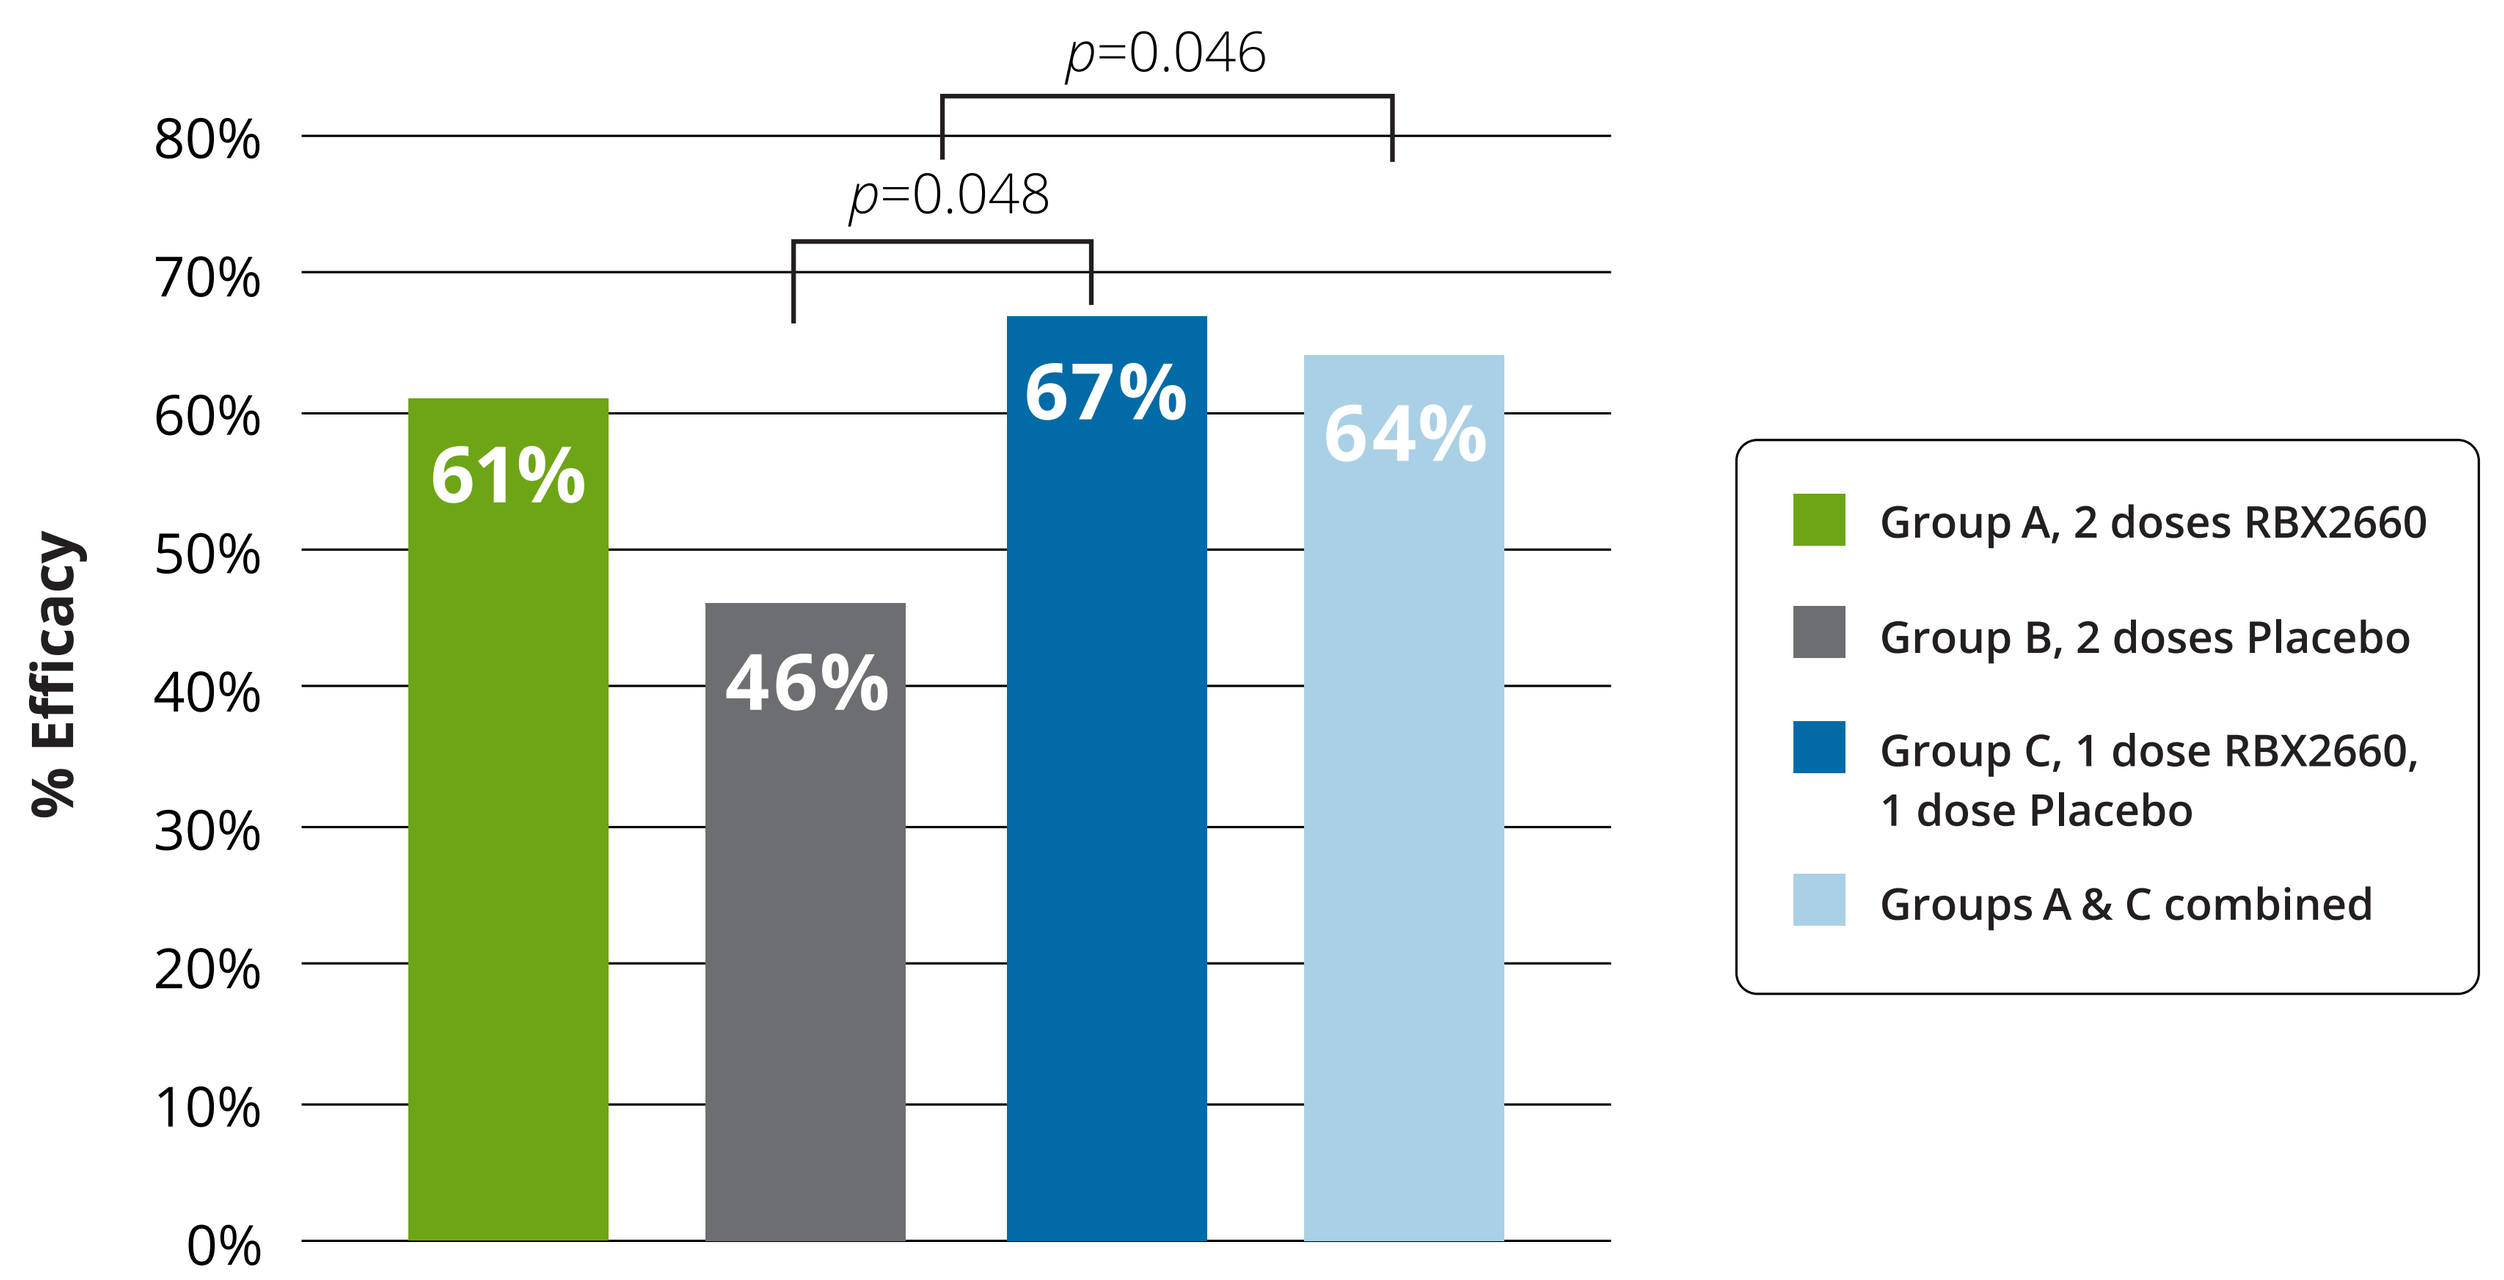

% Efficacy
80%
70%
60%
50%
40%
30%
20%
10%
0%
p=0.046
p=0.048
61%
46%
67%
64%
Group A, 2 doses RBX2660
Group B, 2 doses Placebo
Group C, 1 dose RBX2660, 1 dose Placebo
Groups A & C combined

Results

- 157 stool samples collected from 57 patients classified as responders (A, n=21; B, n=15; C, n=21). Microbiome data for all patients at study entry were combined as a treatment-naïve baseline. For MDS and relative abundance analyses, Groups A and C were pooled as "active" and compared to placebo treatments longitudinally (Table 1)

Table 1

	Patients	# Patient samples				Product samples
		Baseline	7 days	30 days	60 days	
Active Responders (A&C)	42	24	35	27	27	136
Placebo Responders (B)	15	12	10	10	12	

- At 7, 30, and 60 days, microbiomes from RBX2660-treated patients had high KL divergence from baseline and significantly different means from baseline (p<0.001) (Figure 3)
- RBX2660 treatment increased the relative abundances of Bacteroidia, and decreased Gammaproteobacteria and Bacilli more than placebo treatment (Figure 4)
- Recurrent CDI patient microbiomes were dissimilar to RBX2660 product profile at baseline. Active responders became more similar following RBX2660 treatment (Figure 5)

Figure 3: Active responders diverge more from their respective baseline over time than Placebo responders do. Likewise, Active responders become less divergent from RBX2660 over time than Placebo responders do.

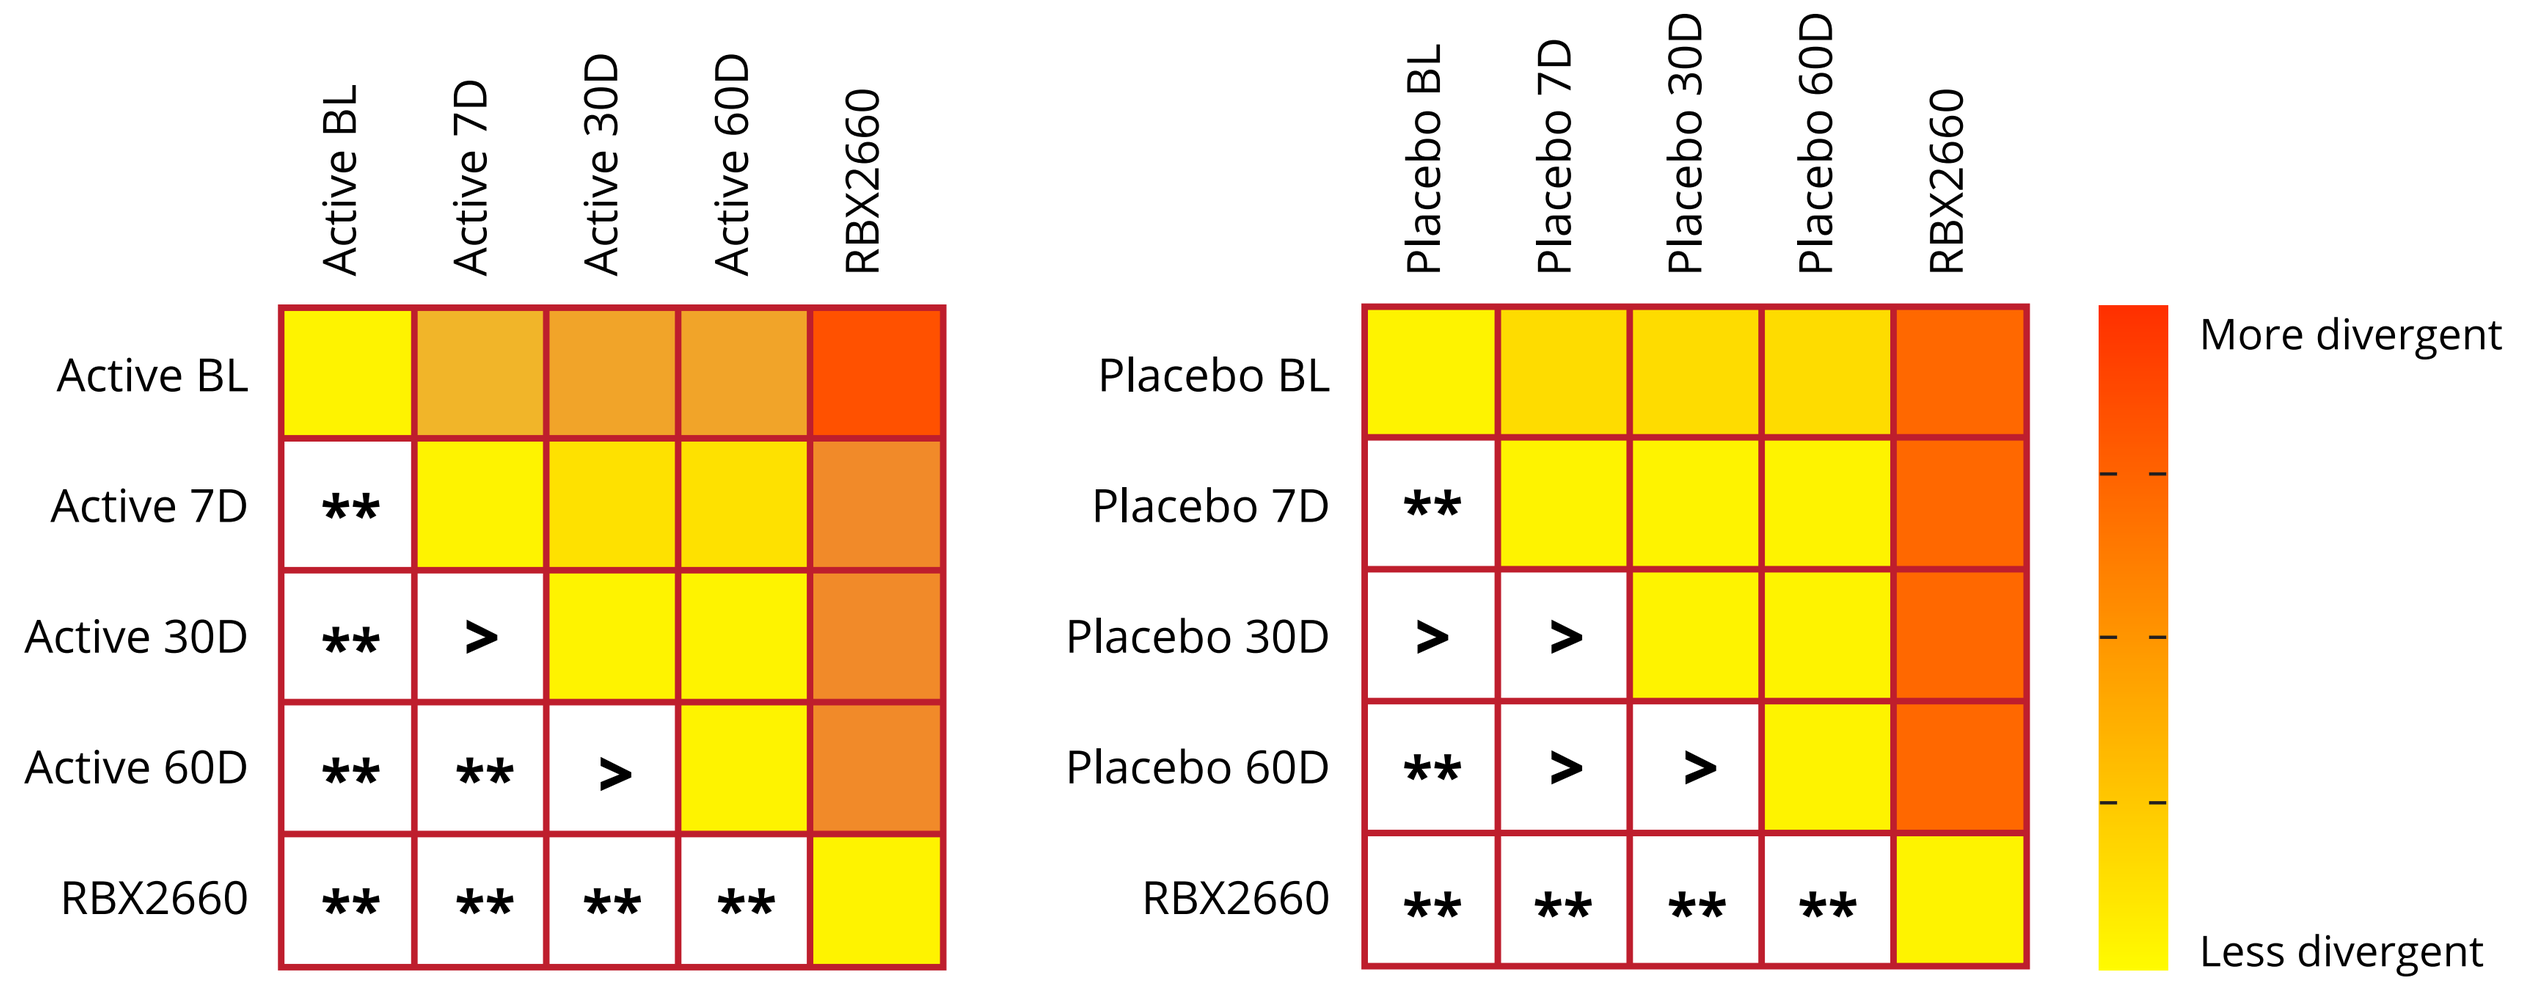

Active BL
Active 7D
Active 30D
Active 60D
RBX2660
Placebo BL
Placebo 7D
Placebo 30D
Placebo 60D
More divergent
Less divergent

Kullback-Leibler (KL) divergence analysis assesses the difference between microbial diversity at different time points or between different samples. KL divergence increases as color ranges from yellow to red. Similarity between two microbiomes decreases as KL divergence increases.
** indicates p<0.001; > indicates p>0.05

Figure 4: Active responders trend toward higher Bacteroides and lower Gammaproteobacteria and Bacilli after treatment, both of which are characteristic of a healthier microbiome

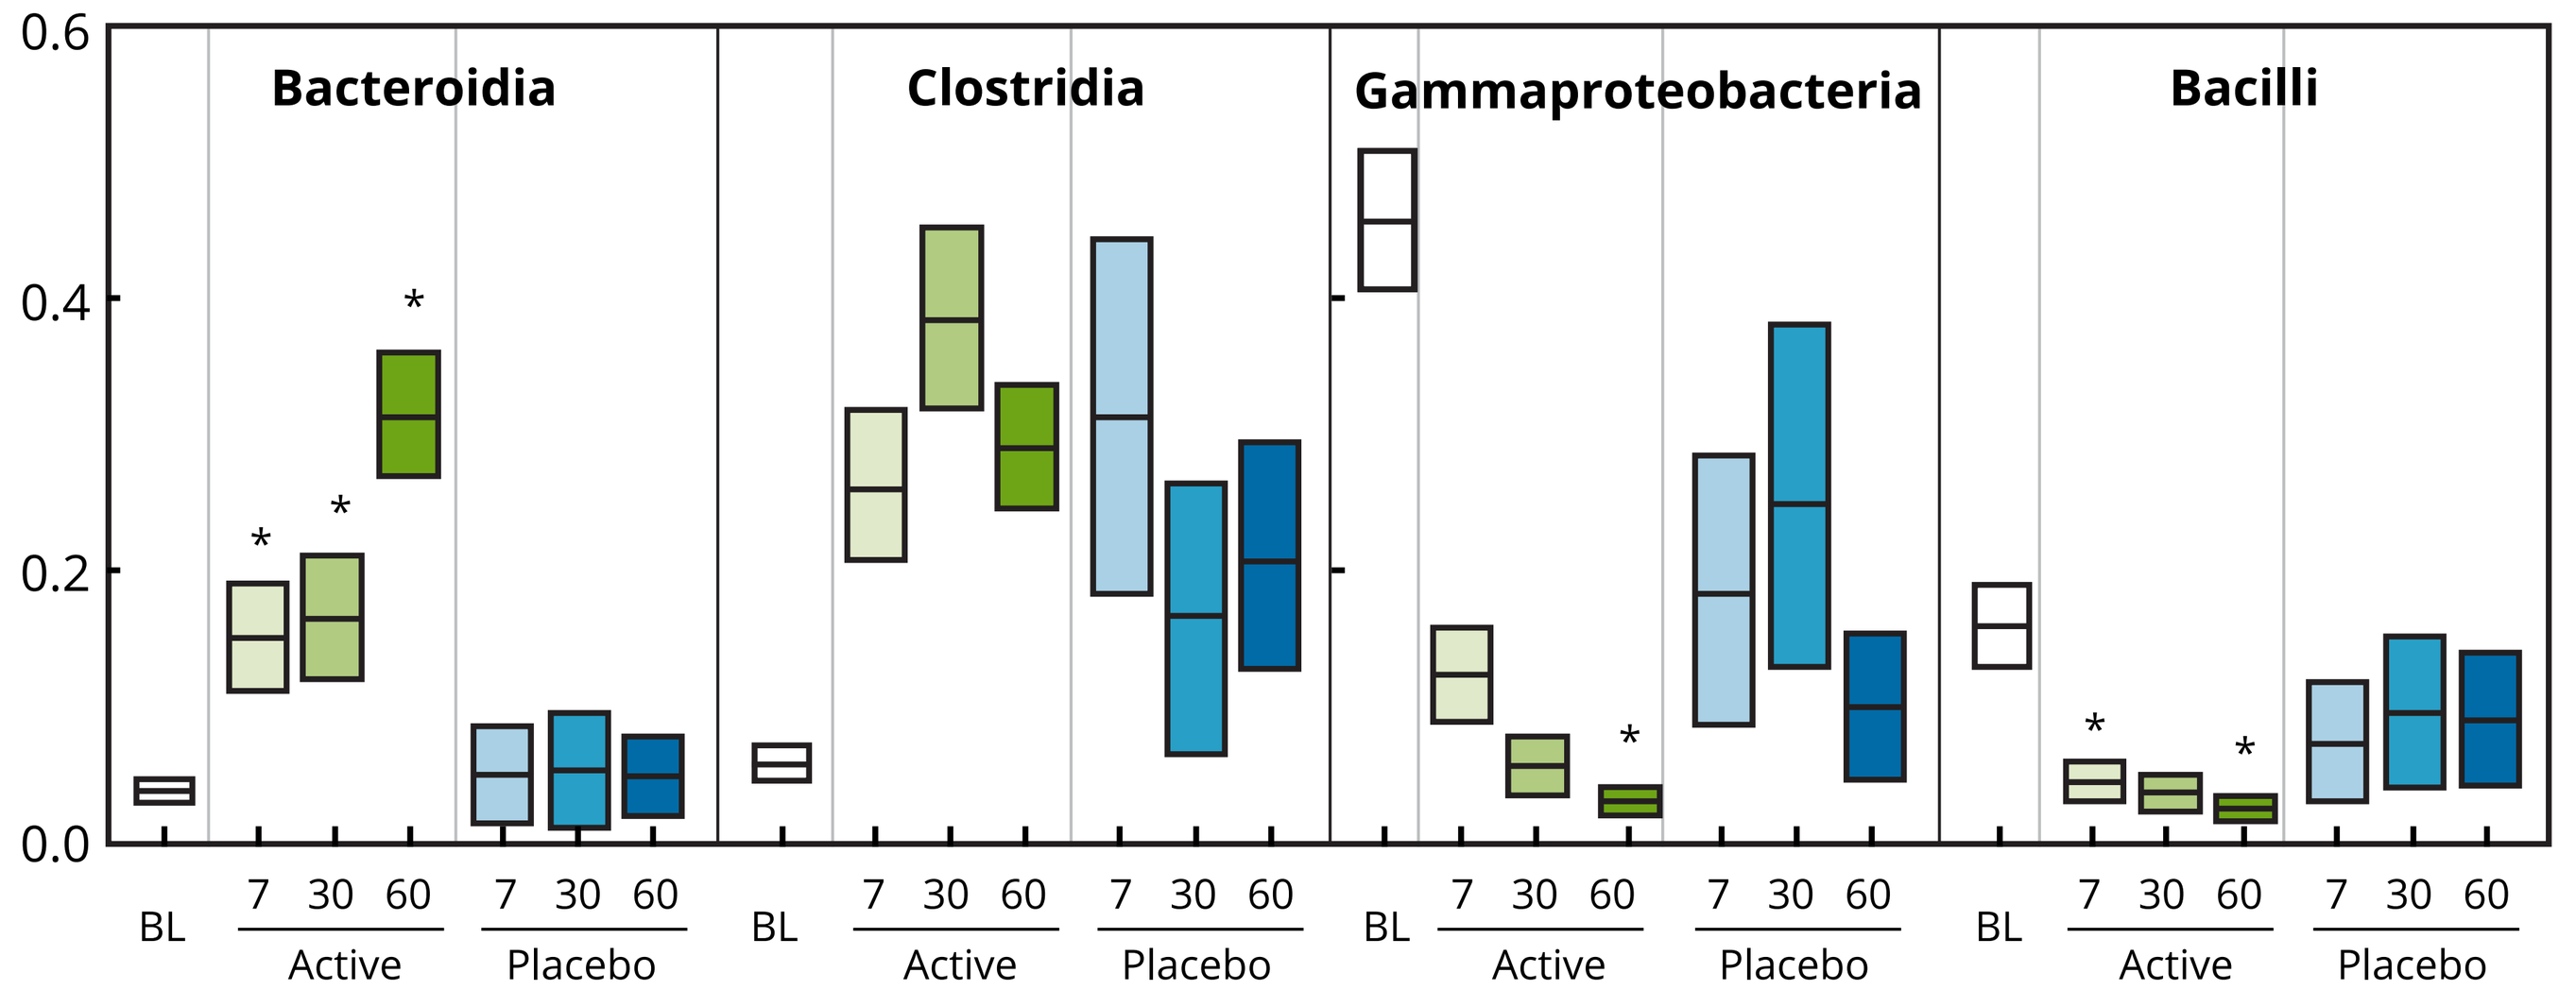

Bacteroidia
Clostridia
Gammaproteobacteria
Bacilli
0.6
0.4
0.2
0.0
BL
7 30 60
Active
7 30 60
Placebo

Dirichlet Comparisons were made at the taxa level using Dirichlet Multinomial parameter (pi) representing the group's mean relative taxa abundance, at each time point. The differences between active and placebo were significant by Wilcoxon test (p < 0.05) for Bacteroidia at 7, 30, and 60d; Gammaproteobacteria at 30d; and Bacilli at 7 and 60d. There were no significant differences between active and placebo for these taxonomic classes at baseline.

Figure 5: Active responders progressed closer to RBX2660 product profile, a representative of a healthy microbiome, with a longitudinal trend. In contrast, Placebo responders do not progress closer to the product profile and do not appear to show a longitudinal trend

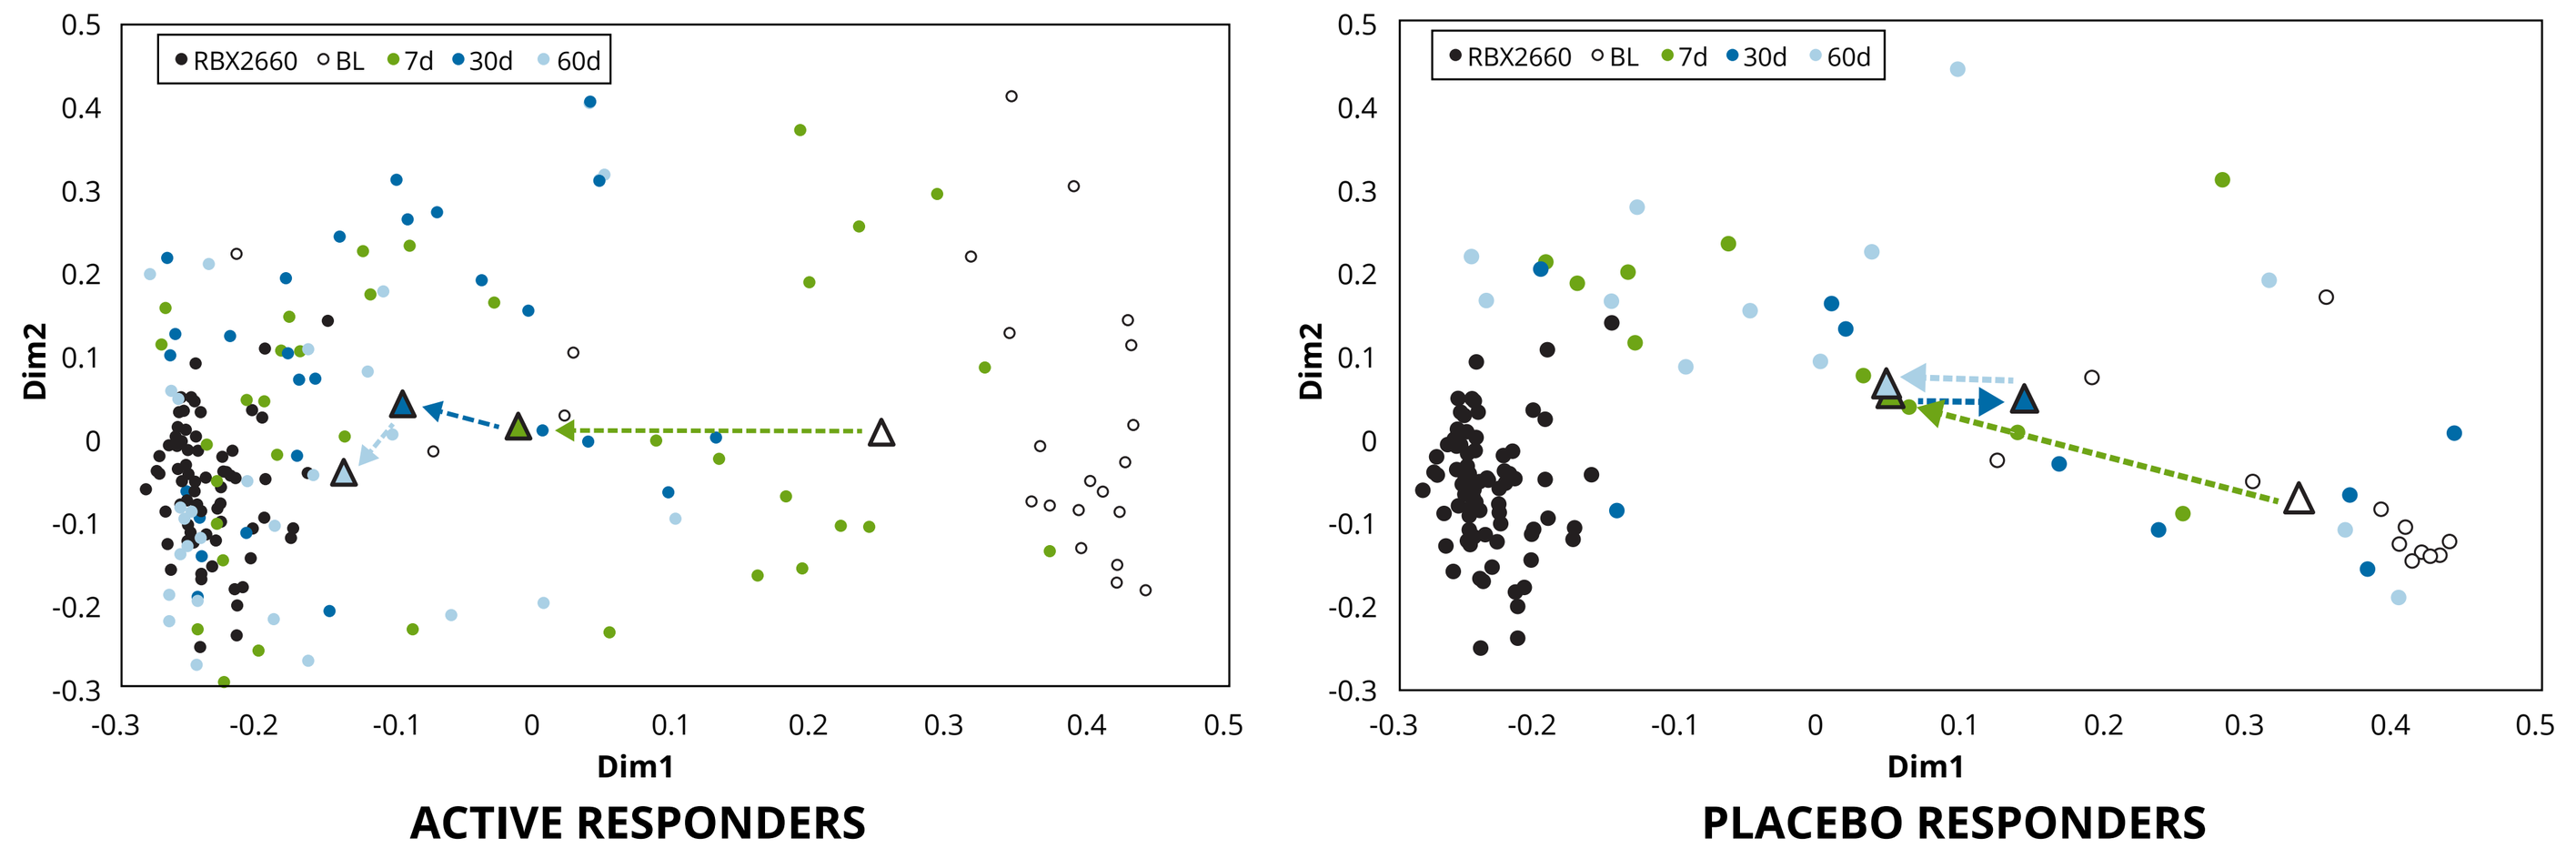

RBX2660
BL
7d
30d
60d
Dim2
Dim1
ACTIVE RESPONDERS
PLACEBO RESPONDERS

Conclusion

- Among patients who were recurrence-free at 8 weeks, those who received RBX2660 had significantly greater microbiome changes than those who received placebo. Most notably Bacteroides were significantly higher after RBX2660 treatment
- These changes are consistent with the hypothesis that RBX2660 can restore a healthier microbiome in rCDI patients. Longer-term studies are needed to compare the durability of microbiome changes and recurrence-free rates

Disclaimer: This analysis was funded by Rebiotix Inc., Roseville, MN.