Durable Prevention of Recurrent *C. difficile* Infection with RBX2660: Results of the PUNCH CD 2 Trial

Mayur Ramesh, MD;[a] Sahil Khanna, MBBS, MS;[b] Julie Messer, BS;[c] Matt Adams, MS[c]

[a]Henry Ford Hospital, Detroit, MI; [b]Division of Gastroenterology & Hepatology, Mayo Clinic, Rochester, MN; [c]Rebiotix Inc., Roseville, MN

Background

- Antibiotic exposure is a major risk factor for the development of *Clostridium difficile* infection (CDI).
- Antibiotic treatment of CDI with standard of care antibiotics contributes to persistent disruption of the intestinal microbiota and predisposes to recurrent infection. The risk of recurrence increases following a second episode.
- Microbiota-based drugs have shown promise in durable prevention of CDI recurrence.
- RBX2660, a microbiota-based drug manufactured from live human-derived microbes, is under study for the prevention of recurrent CDI.
- We report on the durability of RBX2660 in a post-hoc analysis of PUNCH CD 2, a Phase 2b randomized, double-blinded, placebo-controlled trial.

Methods

- Patients with recurrent CDI enrolled in the three-arm PUNCH CD 2 trial were randomized 1:1:1 to receive either: 2 doses of RBX2660, (Group A); 2 doses of placebo (Group B); or 1 dose of RBX2660 and 1 dose of placebo (Group C) via enema with doses 7 days apart (Figure 1).
- Failures in any study group were eligible to receive open-label treatment with up to 2 doses of RBX2660.
- Success was measured as the absence of CDI symptoms at 8 weeks post treatment.
- Failure was defined as: the presence of diarrhea, with or without other CDI symptoms, at less than 8 weeks after last treatment; a positive stool test for C. difficile; the need for retreatment for CDI and no other cause for diarrhea was identified (all four criteria were required).
- Safety follow-ups were scheduled in clinic at 1, 4 and 8 weeks and via telephone at 2, 3 and 5-7 weeks and at 3, 6, 12, and 24 months. Durability assessments occurred concurrently and are on-going until 24 months.

Major Inclusion Criteria

- Age ≥ 18 years old
- A positive stool test for *Clostridium difficile* within 60 days prior to enrollment
- At least two recurrences of CDI after a primary episode

 Or
- At least two episodes of severe CDI resulting in hospitalization

Major Exclusion Criteria

- History of inflammatory bowel disease (ulcerative colitis, Crohn's disease or microscopic colitis); irritable bowel syndrome; chronic diarrhea; celiac disease
- Colostomy
- Evidence of active colitis
- Known exposure to antibiotics within 6 months after study enrollment
- Compromised immune system
- White blood cell count <1000 cells/µL

Figure 1. PUNCH CD 2 Trial Design

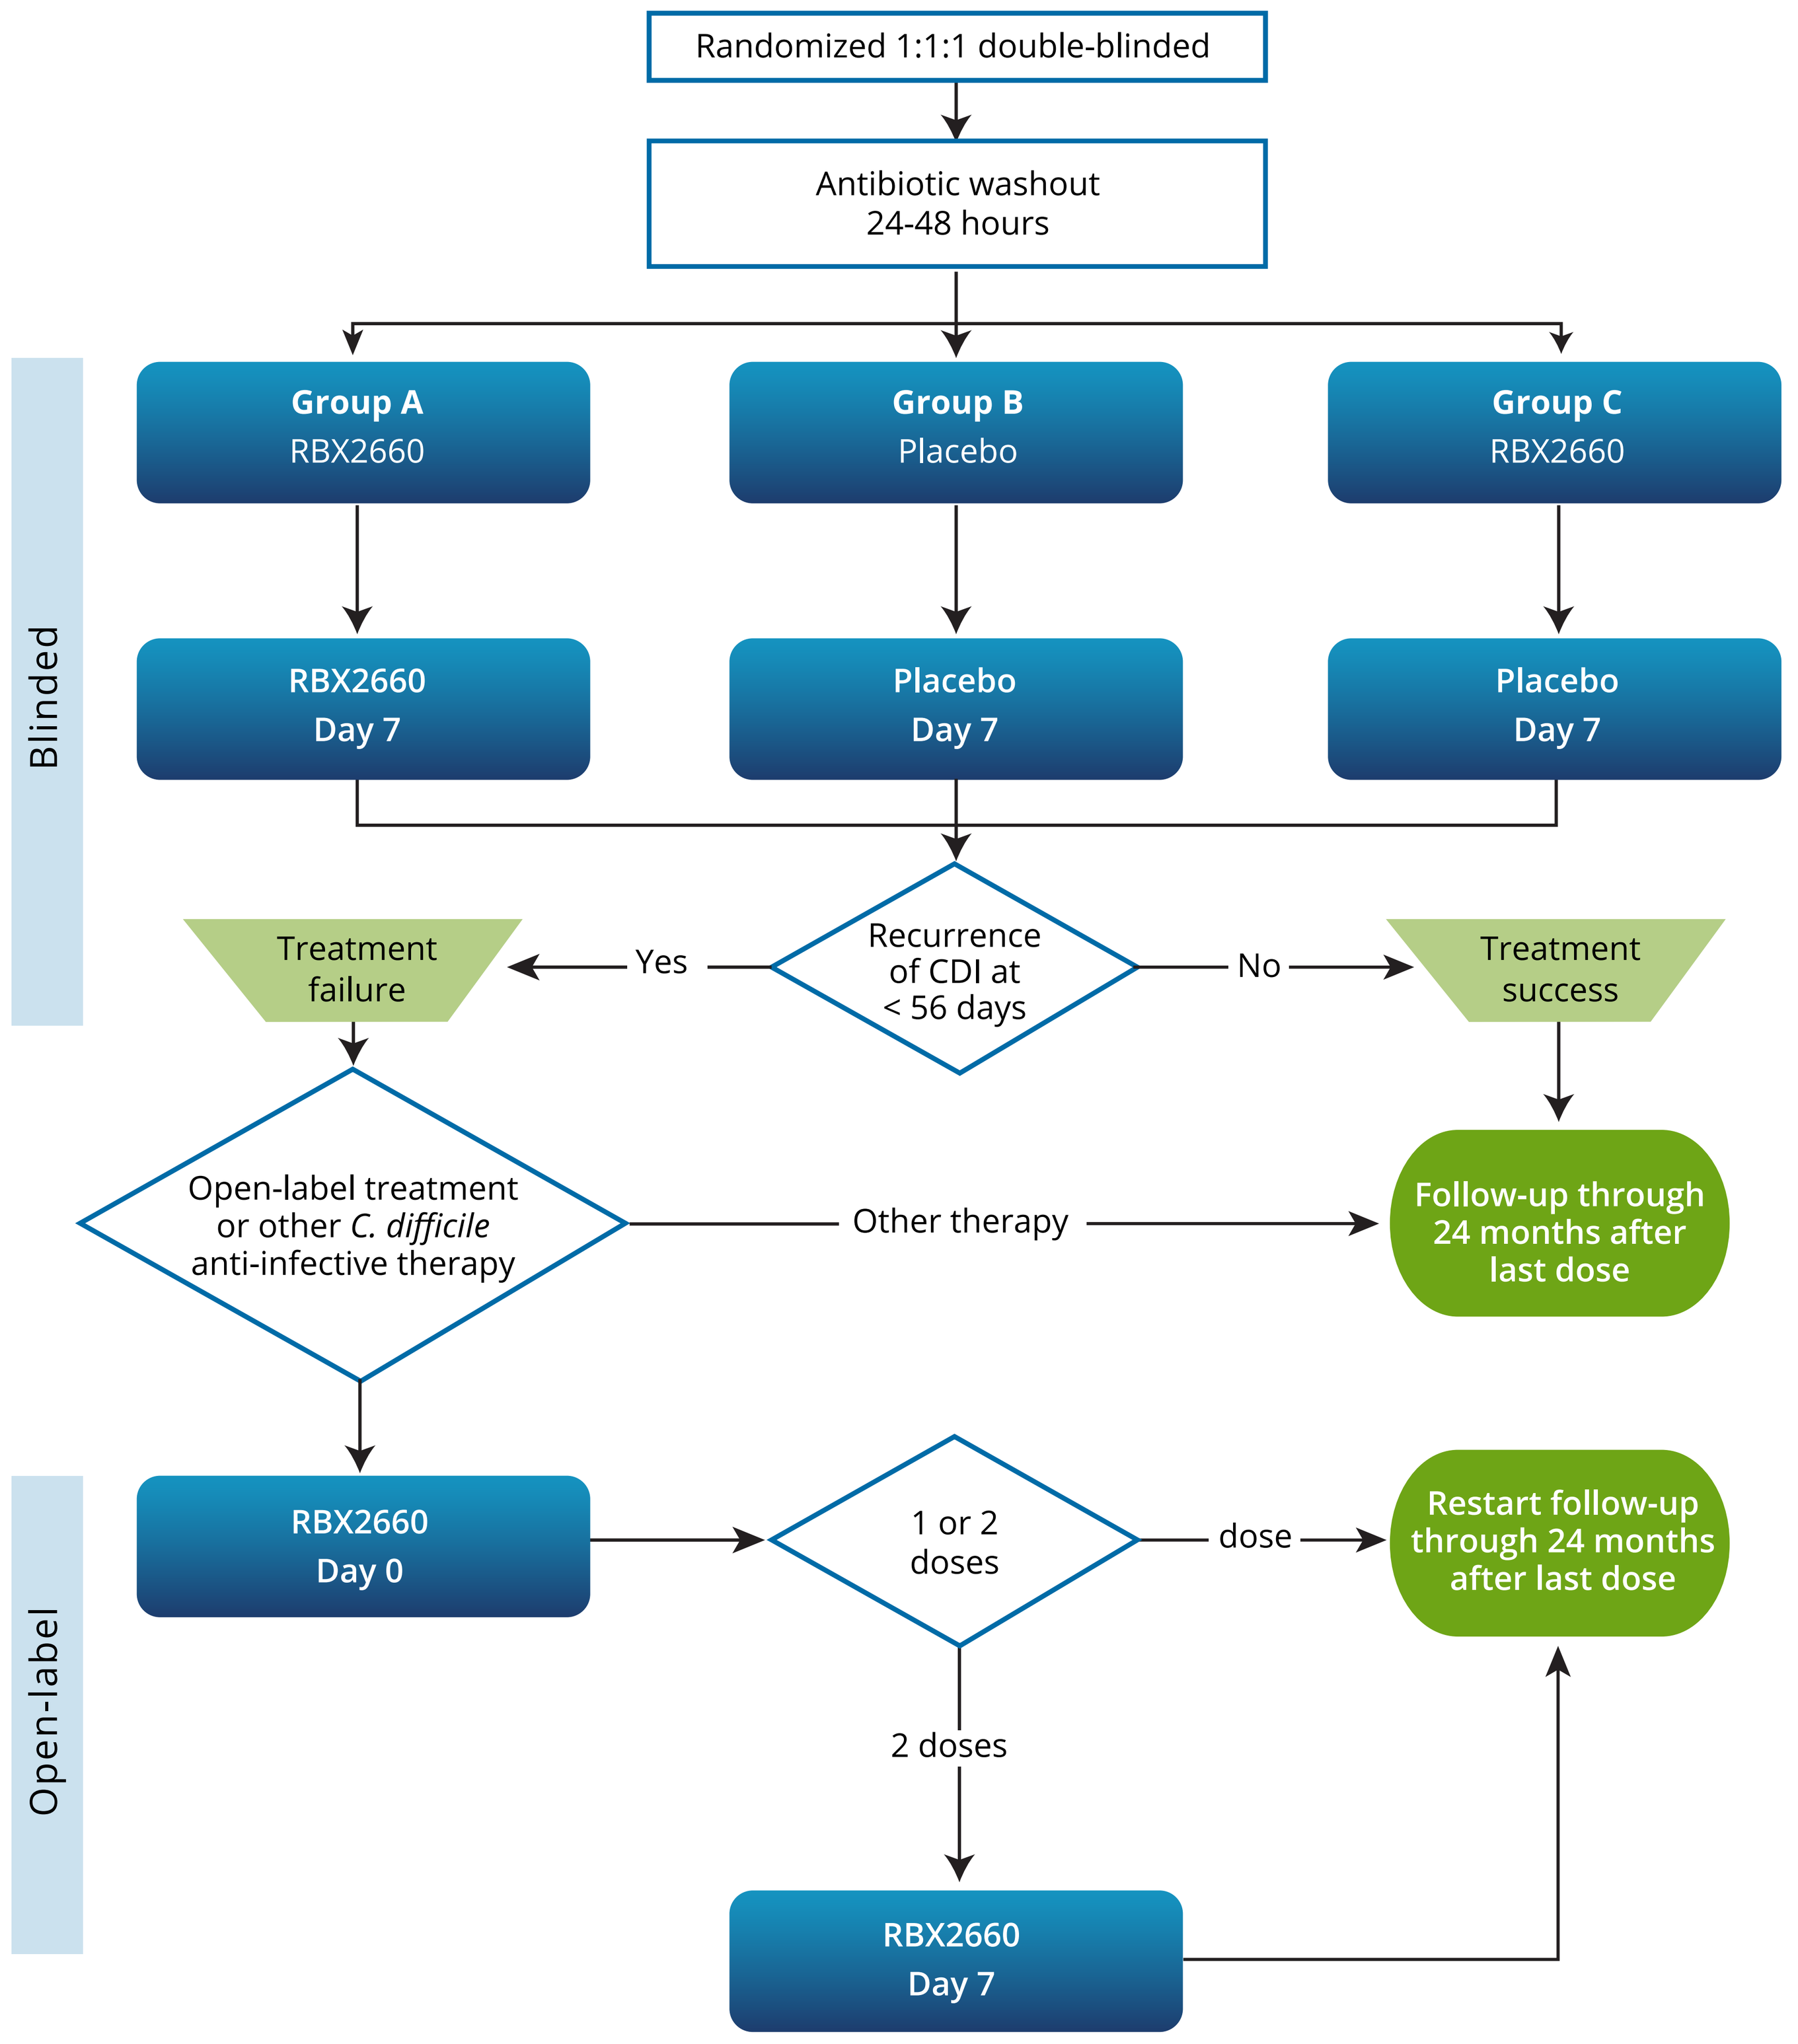
Randomized 1:1:1 double-blinded
Antibiotic washout 24-48 hours
Blinded
Group A RBX2660
Group B Placebo
Group C RBX2660
RBX2660 Day 7
Placebo Day 7
Placebo Day 7
Recurrence of CDI at < 56 days
Yes
No
Treatment failure
Treatment success
Open-label treatment or other C. difficile anti-infective therapy
Other therapy
Follow-up through 24 months after last dose
Open-label
RBX2660 Day 0
1 or 2 doses
dose
Restart follow-up through 24 months after last dose
2 doses
RBX2660 Day 7

Results

- A total of 107 patients (median age 63, range: 18-92 years; 59.8% female) at 21 centers in the U.S. and Canada received at least 1 dose of RBX2660 (Table 1).
 - Group A (n=41) and Group C (n=42)
 - Placebo patients who went on to receive open-label RBX2660 (n=24)
- The overall success rate of patients who received at least one dose of RBX2660 in the blinded and open-label phases of the trial was 88.8% (95/107) (Table 1).
- Of the successful patients, 4.2% (4/95) developed a new episode of CDI confirmed by a positive test > 8 weeks after the last RBX2660 treatment (Table 2).
- The long-term CDI-free rate (median follow-up: 8.3 months; range 1.6 to 14.9 months) was 95.8% (91/95) (Table 3).

TABLE 1. PATIENTS WHO RECEIVED AT LEAST 1 DOSE OF RBX2660 (N=107)

	Treatment Success n (%)	Treatment Failure n (%)
Groups A and C n= 83	74 (89.2)	9 (10.8)
Group B n=24	21 (87.5)	3 (12.5)
Overall N=107	95 (88.8)	12 (11.2)

TABLE 2. RECURRENCES AFTER SUCCESSFUL TREATMENT WITH RBX2660

Patient	Reason for Recurrence
1	After antibiotics for a dog bite
2	During a hospital stay for small bowel obstruction
3	Unknown
4	Unknown

TABLE 3. DURABILITY OF RBX2660 TREATMENT

	OVERALL	3 MONTHS	6 MONTHS	12 MONTHS
Long-term infection-free rate %, n,N	95.8 91/95	98.9 90/91	95.7 67/70	87.5 14/16

Conclusions

- RBX2660, a microbiota-based drug, provided a durable cure for recurrent CDI in a randomized, double-blinded, placebo-controlled trial with up to 12-months follow-up. Long-term follow-up to 24 months is ongoing.
- Longitudinal assessment of the impact of RBX2660 treatment on the intestinal microbiota of patients may provide additional insight into durable prevention of CDI.

Corresponding Author

Mayur Ramesh, MD
Transplant Infectious Diseases & Immunotherapy
Henry Ford Hospital
2799 West Grand Blvd, 313 CFP
Detroit, MI 48202
E-mail: mramesh1@hfhs.org

The PUNCH CD 2 trial was sponsored by Rebiotix Inc., Roseville, MN.