Does the Donor Matter? Results from PUNCH CD 2, a Randomized Controlled Trial of a Microbiota-based Drug for Recurrent *Clostridium difficile* Infection

Arnab Ray, MD;[a] Courtney Jones, BS,[b] Bill Shannon, PhD, MBA,[c] Sharina Carter, BS[c]

[a]Ochsner Clinic Foundation, New Orleans, LA; [b]Rebiotix Inc., Roseville, MN; [c]BioRankings, St. Louis, MO

Corresponding Author
Courtney Jones
2660 Patton Road
Roseville, MN 551113
Tel: 651-705-8770
E-mail: cjones@rebiotix.com

Background

Questions have been raised about the applicability of a universal donor in regard to microbiota-based therapies for recurrent *Clostridium difficile* infection (CDI).

Conventional procedure has been to use a second donor in case of treatment failure.

Analysis of PUNCH CD, an open-label Phase 2 study of RBX2660, a microbiota-based drug targeted at recurrent CDI, found that the specific donor used to manufacture drug product did not affect outcomes.[1]

Donor vs. patient factors in therapy outcomes were further studied in PUNCH CD 2, a randomized, double-blinded, placebo-controlled multicenter study of RBX2660.

We report on results from the blinded phase of the PUNCH CD 2 study.

Methods

Patients in the blinded phase of the PUNCH CD 2 study were randomized into one of 3 arms: 2 doses of RBX2660 (Group A); 2 doses of placebo (Group B); or 1 dose of RBX2660 and 1 dose of placebo (Group C) via enema with doses 7 days apart.

Failures could receive up to 2 doses of open-label active treatment, 7 days apart.

Patients who received multiple doses of RBX2660 could receive drug manufactured from the same or different donors. Donors could also be used in a different order.

Success was defined as the absence of *Clostridium difficile*-associated diarrhea at 8 weeks following completion of the last treatment.

A mathematical model was used to determine whether the specific donor affected the results.

RBX2660

Each dose of RBX2660 consisted of 50 gm of human stool/150 mL solution in a single-dose ready-to-use enema bag.

RBX2660 was manufactured using standardized, quality-controlled processes.

Sourcing and Traceability

A total of 20 donors were used to prepare the RBX2660 drug product used used in the study.

Donations were collected on site at Rebiotix Inc., Roseville, MN.

Donations were not pooled across donors.

Units of RBX2660 were identified by a unique batch number and traceable to a specific donor and recipient.

Patients who received two doses of RBX2660 randomly received product from the next available batch.

Mathematical Model

A mixed model was used to predict treatment success taking into account donors and dose number.

Repeated measures data was included to account for patients who received 2 doses of RBX2660. In this analysis, a patient was classified as either a success or failure for each dose.

$P < .05$ indicates statistical significance; all analyses were done using the R statistical package.

Results

A total of 83 patients (mean age 62 years, 59% female) received at least one dose of RBX2660.

Success was not impacted by the donor or the dose order (Table 1, Figure 1).

TABLE 1. OUTCOMES FOR DONORS AND PATIENTS

	Estimate	P Value
Fixed Effect	Coefficient	
Intercept	11.042	4.07e-12
Random Effect	Variance	
Donor	0	> 0.99

- The donor was not significant (P > 0.99).
- The variance = 0, indicated that no difference in outcome by donor was expected and that the treatment rates for success and failure were the same for each donor.

Figure 1. Proportion of Successes and Failures with RBX2660 by Donor

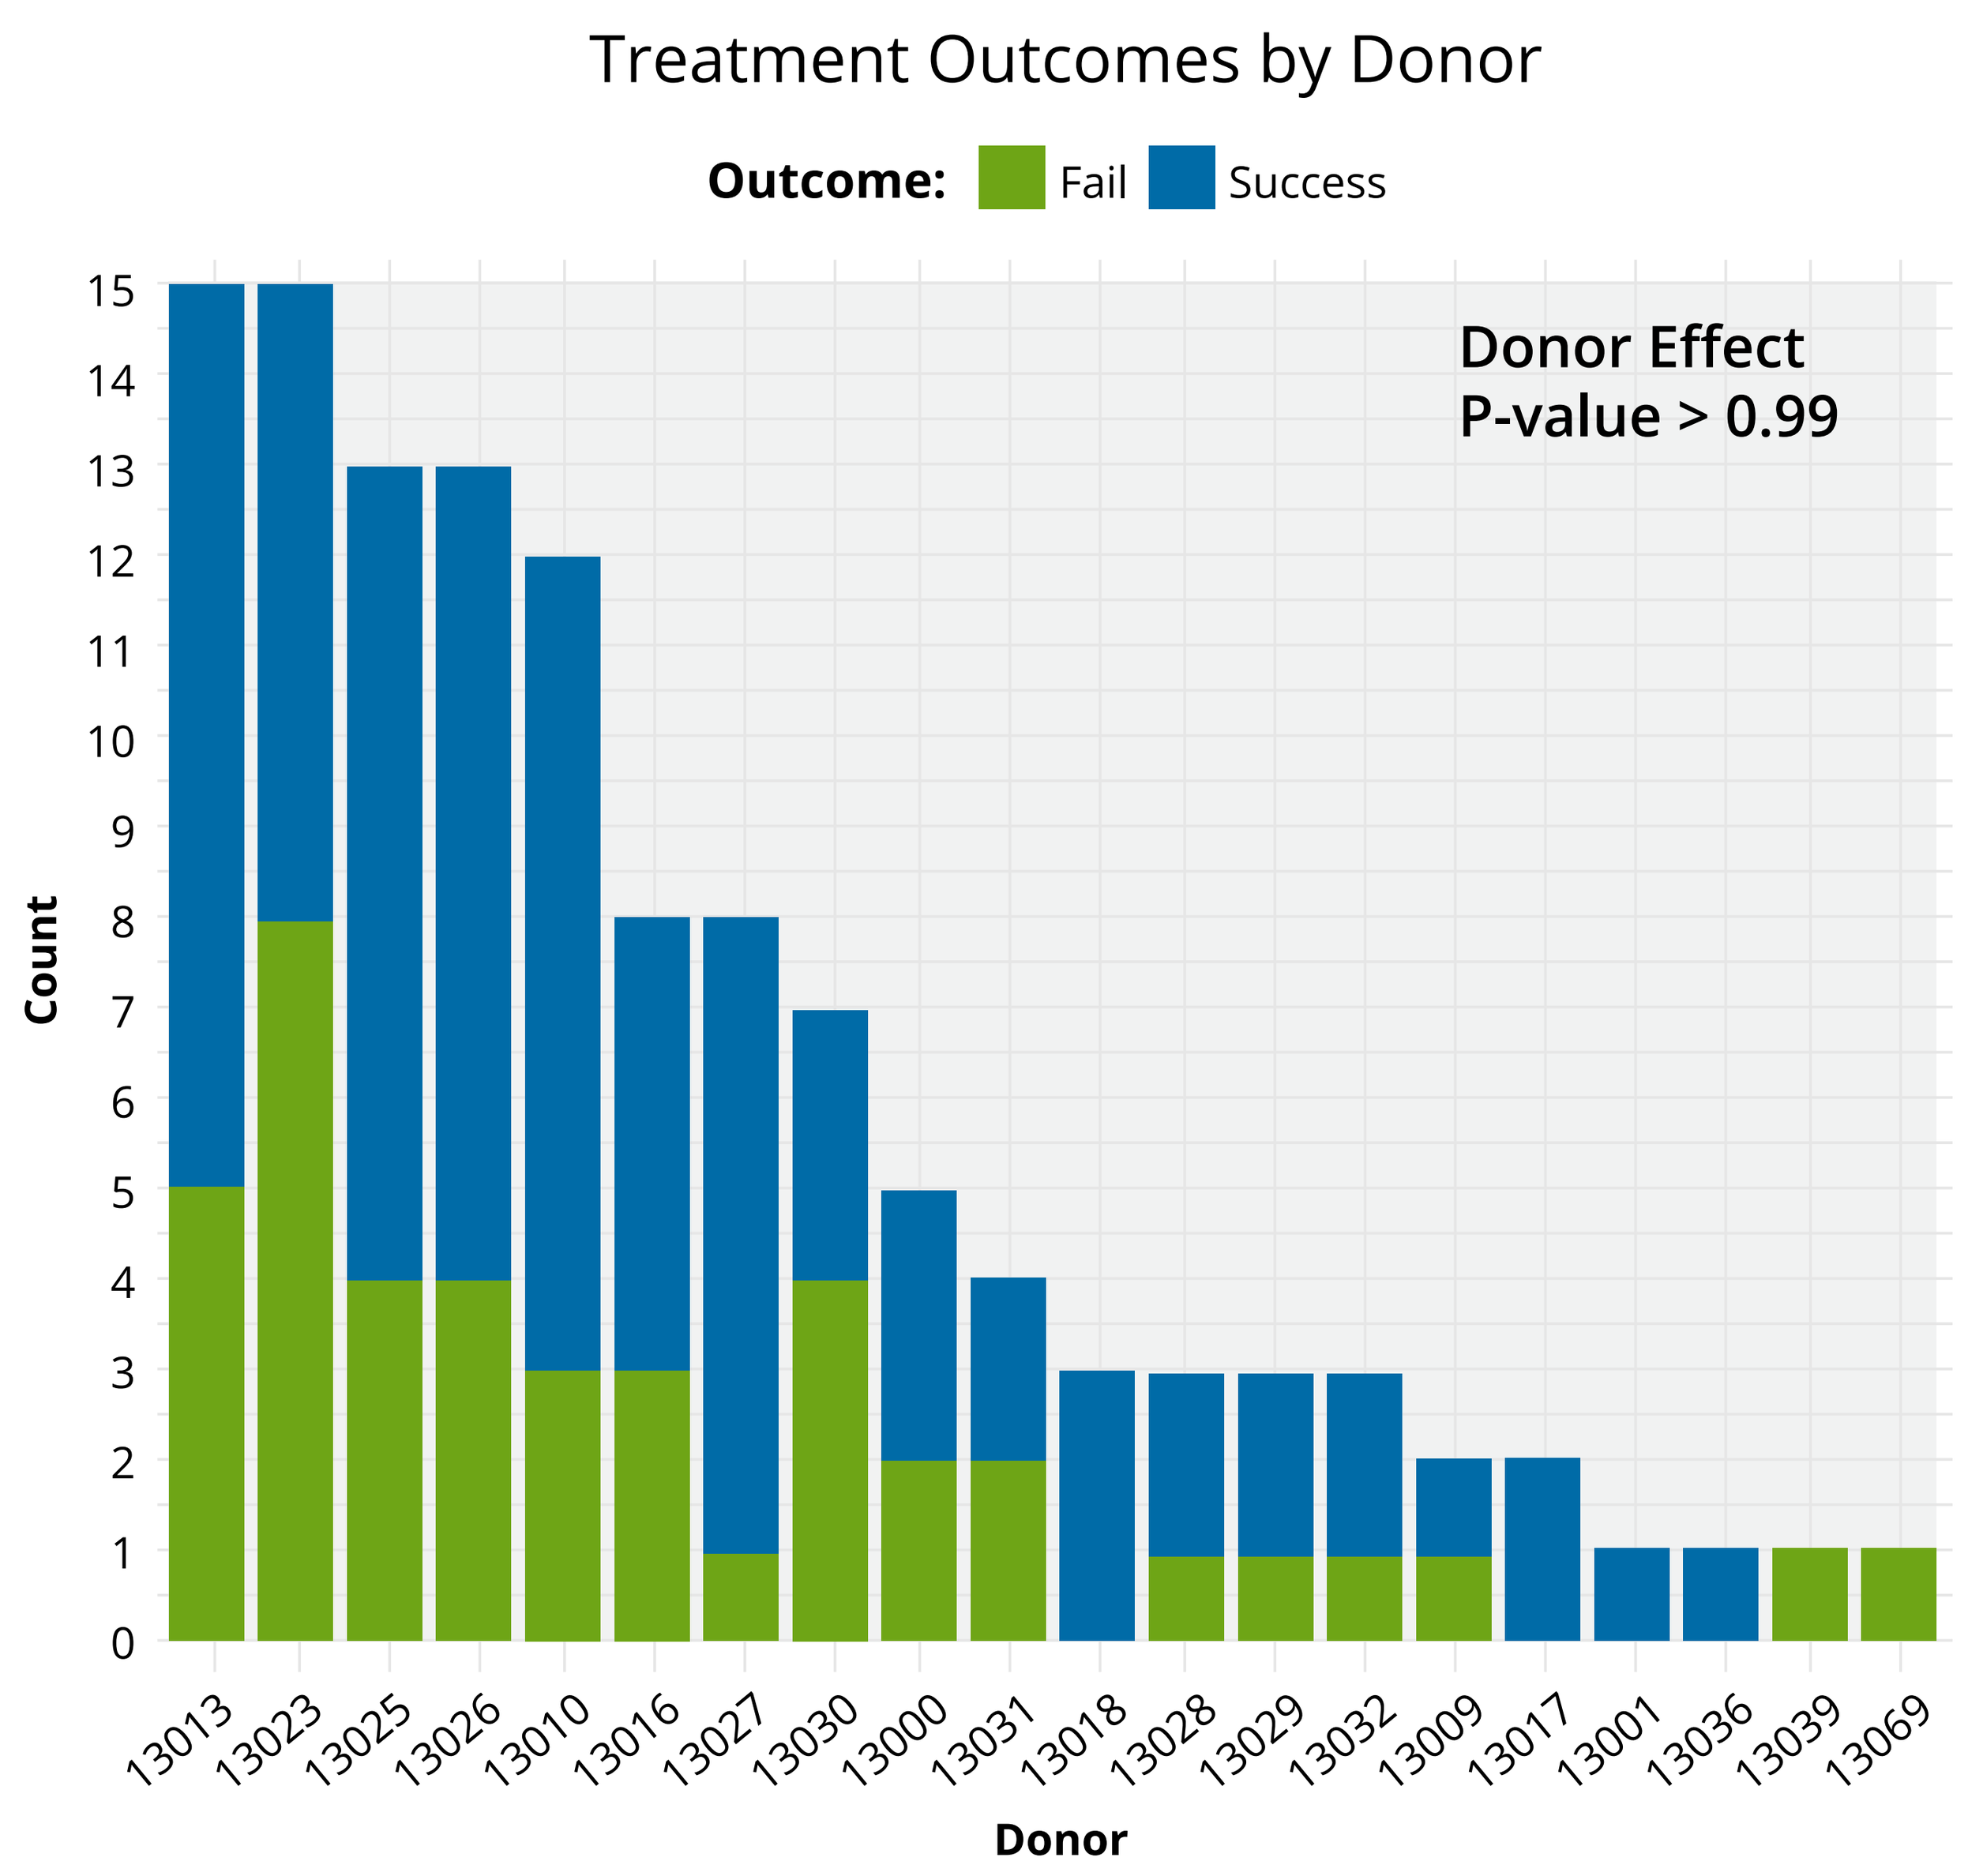
Treatment Outcomes by Donor
Outcome: Fail Success
Donor Effect
P-value > 0.99
Count
0
1
2
3
4
5
6
7
8
9
10
11
12
13
14
15
13013
13023
13025
13026
13010
13016
13027
13030
13000
13031
13018
13028
13029
13032
13009
13017
13001
13036
13039
13069
Donor

- One outcome (patient) may be associated with up to two different donors. Therefore, patients that received two doses of RBX2660 are represented twice.
- The number of patients per donor varied but the proportion of successes and failures were similar across the donors.

Conclusions

This analysis of a randomized double-blinded, placebo-controlled multicenter study, demonstrated that the specific donor does not affect the outcomes achieved with administration of RBX2660 for recurrent CDI.

The results are consistent with a previous analysis of an open-label study of RBX2660 and suggest that donor-to-patient matching is not necessary the treatment of recurrent CDI.

References

1. Ray A, Jones C. Does the donor matter? Donor vs patient effects in the outcome of a next- generation microbiota-based drug trial for recurrent *Clostridium difficile* infection. *Future Microbiol*. 2016;11:611-6.

This analysis was funded by Rebiotix Inc., Roseville, MN.