Who Seeks a Fecal Microbiota Transplant for Recurrent C. *difficile* Infection?: Patient Profile of the PUNCH™ CD Study

Robert Orenstein, DO, Infectious Diseases, Mayo Clinic Arizona, Phoenix, AZ, Erik Dubberke, MD, MSPH, Infectious Disease, Washington University School of Medicine, St. Louis, MO, Cheryl Griesbach, RN, Clinical Studies Unit-Phoenix, Mayo Phoenix, Phoenix, AZ and Mary Kay Sobcinski, RN, MHA, Rebiotix Inc, Roseville, MN

Background

Managing recurrent C. difficile infection (CDI) is challenging due to the limited effectiveness of current treatment options. Fecal transplant (FT) appears to be an effective mechanism for managing multiply recurrent CDI. Studies have looked at the acceptability of FT. However, little is known about the patients who seek FT for recurrent CDI. We report the demographics of subjects enrolled in an ongoing Phase 2 study (PUNCH CD) of RBX2660 (microbiota suspension), a next generation standardized and commercially prepared version of FT for the treatment of recurrent CDI.

Methods

The PUNCH CD study enrolled a range of patients with recurrent CDI who were: = 18 years with recurrent CDI who had at least two recurrences after a primary episode and completed at least two rounds of standard-of-care oral antibiotic therapy or had at least two episodes of severe CDI resulting in hospitalization.

Results

A total of 40 subjects were enrolled at 11 US centers with 34 subjects receiving treatment with RBX2660. Study centers were geographically and functionally diversified to include academic health centers located in major metro areas, community-based specialty clinics, and an integrated health system serving a sparsely populated largely rural state. Baseline characteristics of the 34 treated subjects were: mean age: 66.8 years (range 26.7 to 89.6 years); female (67.6%, n=23); Caucasian (94.2%, n=32) with a mean BMI 24.4 (range: 15.0 to 37.0). Comorbid illnesses included: gastrointestinal (61.8%, n=21), cardiovascular (55.9%, n=19), genitourinary (69.2%, n=18), and psychiatric (28.1%, n=9). A total of 76% (n=26) of subjects were not working (mostly retired) while 24% (n=8) reported working full- or part-time with no restrictions.

Conclusion

The population of patients seeking treatment in this study of next-generation FT drug was primarily Caucasian, elderly and female with multiple comorbidities. These demographics highlight populations with recurrent CDI and increase the awareness of opportunities to diversify enrollment in trials for recurrent CDI.

Baseline Demographics

Demographics	**N = 34**
Age (years)	
Mean (SD)	66.8 (15.1)
Median	68.7
Min/Max	26.7/89.6
Gender (n/N [%])	
Male	11/34 (32.4)
Female	23/34 (67.6)
Ethnicity (n/N[%])	
Hispanic or Latino	1/34 (2.9)
Not Hispanic or Latino	33/34 (97.1)
Not Reported	0/34 (0.0)
Unknown	0/34 (0.0)
Race (n/N[%])	
American Indian or Alaska Native	1/34 (2.9)
Asian	0/34 (0.0)
Black or African American	1/34 (2.9)
Native Hawaiian or Other Pacific Islander	0/34 (0.0)
White	33/34 (97.1)
Other	0/34 (0.0)

Program: RBX01 A Demographics.sas
Data Source: RBX01_D_Demographics, RBX01_D_Disposition
Date Run: 24MAR2014 - 16:23

Who Seeks a Fecal Microbiota Transplant for Recurrent *C. difficile* Infection?: Patient Profile of the PUNCH™ CD Study

Robert Orenstein, DO, Infectious Diseases, Mayo Clinic Arizona, Phoenix, AZ, Erik Dubberke, MD, MSPH, Infectious Disease, Washington University School of Medicine, St. Louis, MO, Cheryl Griesbach, RN, Clinical Studies Unit-Phoenix, Mayo Phoenix, Phoenix, AZ and Mary Kay Sobcinski, RN, MHA, Rebiotix Inc, Roseville, MN

Background

Managing recurrent C. difficile infection (CDI) is challenging due to the limited effectiveness of current treatment options. Fecal transplant (FT) appears to be an effective mechanism for managing multiply recurrent CDI. Studies have looked at the acceptability of FT. However, little is known about the patients who seek FT for recurrent CDI. We report the demographics of subjects enrolled in an ongoing Phase 2 study (PUNCH CD) of RBX2660 (microbiota suspension), a next generation standardized and commercially prepared version of FT for the treatment of recurrent CDI.

Methods

The PUNCH CD study enrolled a range of patients with recurrent CDI who were: = 18 years with recurrent CDI who had at least two recurrences after a primary episode and completed at least two rounds of standard-of-care oral antibiotic therapy or had at least two episodes of severe CDI resulting in hospitalization.

Results

A total of 40 subjects were enrolled at 11 US centers with 34 subjects receiving treatment with RBX2660. Study centers were geographically and functionally diversified to include academic health centers located in major metro areas, community-based specialty clinics, and an integrated health system serving a sparsely populated largely rural state. Baseline characteristics of the 34 treated subjects were: mean age: 66.8 years (range 26.7 to 89.6 years); female (67.6%, n=23); Caucasian (94.2%, n=32) with a mean BMI 24.4 (range: 15.0 to 37.0). Comorbid illnesses included: gastrointestinal (61.8%, n=21), cardiovascular (55.9%, n=19), genitourinary (69.2%, n=18), and psychiatric (28.1%, n=9). A total of 76% (n=26) of subjects were not working (mostly retired) while 24% (n=8) reported working full- or part-time with no restrictions.

Conclusion

The population of patients seeking treatment in this study of next-generation FT drug was primarily Caucasian, elderly and female with multiple comorbidities. These demographics highlight populations with recurrent CDI and increase the awareness of opportunities to diversify enrollment in trials for recurrent CDI.

Baseline Demographics

Demographics	**N = 34**
Age (years)	
Mean (SD)	66.8 (15.1)
Median	68.7
Min/Max	26.7/89.6
Gender (n/N [%])	
Male	11/34 (32.4)
Female	23/34 (67.6)
Ethnicity (n/N[%])	
Hispanic or Latino	1/34 (2.9)
Not Hispanic or Latino	33/34 (97.1)
Not Reported	0/34 (0.0)
Unknown	0/34 (0.0)
Race (n/N[%])	
American Indian or Alaska Native	1/34 (2.9)
Asian	0/34 (0.0)
Black or African American	1/34 (2.9)
Native Hawaiian or Other Pacific Islander	0/34 (0.0)
White	33/34 (97.1)
Other	0/34 (0.0)

Program: RBX01 A Demographics.sas
Data Source: RBX01_D_Demographics, RBX01_D_Disposition
Date Run: 24MAR2014 - 16:23